A Phase 1 Study to Evaluate the Pharmacokinetics of a Single Dose of Apatinib Mesylate (YN968D1) Tablets in Healthy Male Caucasian, Japanese, and Chinese Subjects

Luana Pesco-Koplowitz [1], Barry Koplowitz [1], Cheol Hee Park [2], Arlo N. McGinn [2]

[1] DUCK FLATS Pharma, Elbridge, NY, USA; [2] LSK BioPartners, Inc. (dba LSK BioPharma), Salt Lake City, UT

Background

- Apatinib (YN968D1) is an orally administered, selective tyrosine kinase inhibitor of vascular endothelial growth factor receptor-2 (VEGFR-2)/kinase insert domain receptor (KDR) that has been studied in various solid tumors.
- Apatinib was approved in China in 2014 for the treatment of advanced/metastatic gastric cancer (GC), based on the Chinese phase 3 GC trial.[1]
- Outside of China, apatinib is currently undergoing a multinational phase 3 study in previously treated advanced GC patients in North America, Europe, and Asia Pacific (Clinicaltrials.gov identifier: NCT03042611).[2]
- The objective of this study is to evaluate the pharmacokinetics (PK) of apatinib in healthy Caucasian, Chinese, and Japanese subjects following a single dose administration to determine if ethnicity causes any differences in the drug's PKs.

Methods

General Methodology

This was a single-dose, open label, parallel design study of one dose of 250 mg of apatinib mesylate tablets (YN968D1) (201 mg freebase) in healthy male volunteers of Caucasian, Chinese, or Japanese descent. Plasma samples for PK analysis were collected up to 72 hours postdose. All subjects were genotyped for CYP3A4 and CYP2C19 prior to study enrollment. Subjects that were CYP3A4 poor metabolizers were to be excluded from the study. The Lower Limit of Quantitation (LLOQ) for apatinib in plasma was 0.500 ng/mL.

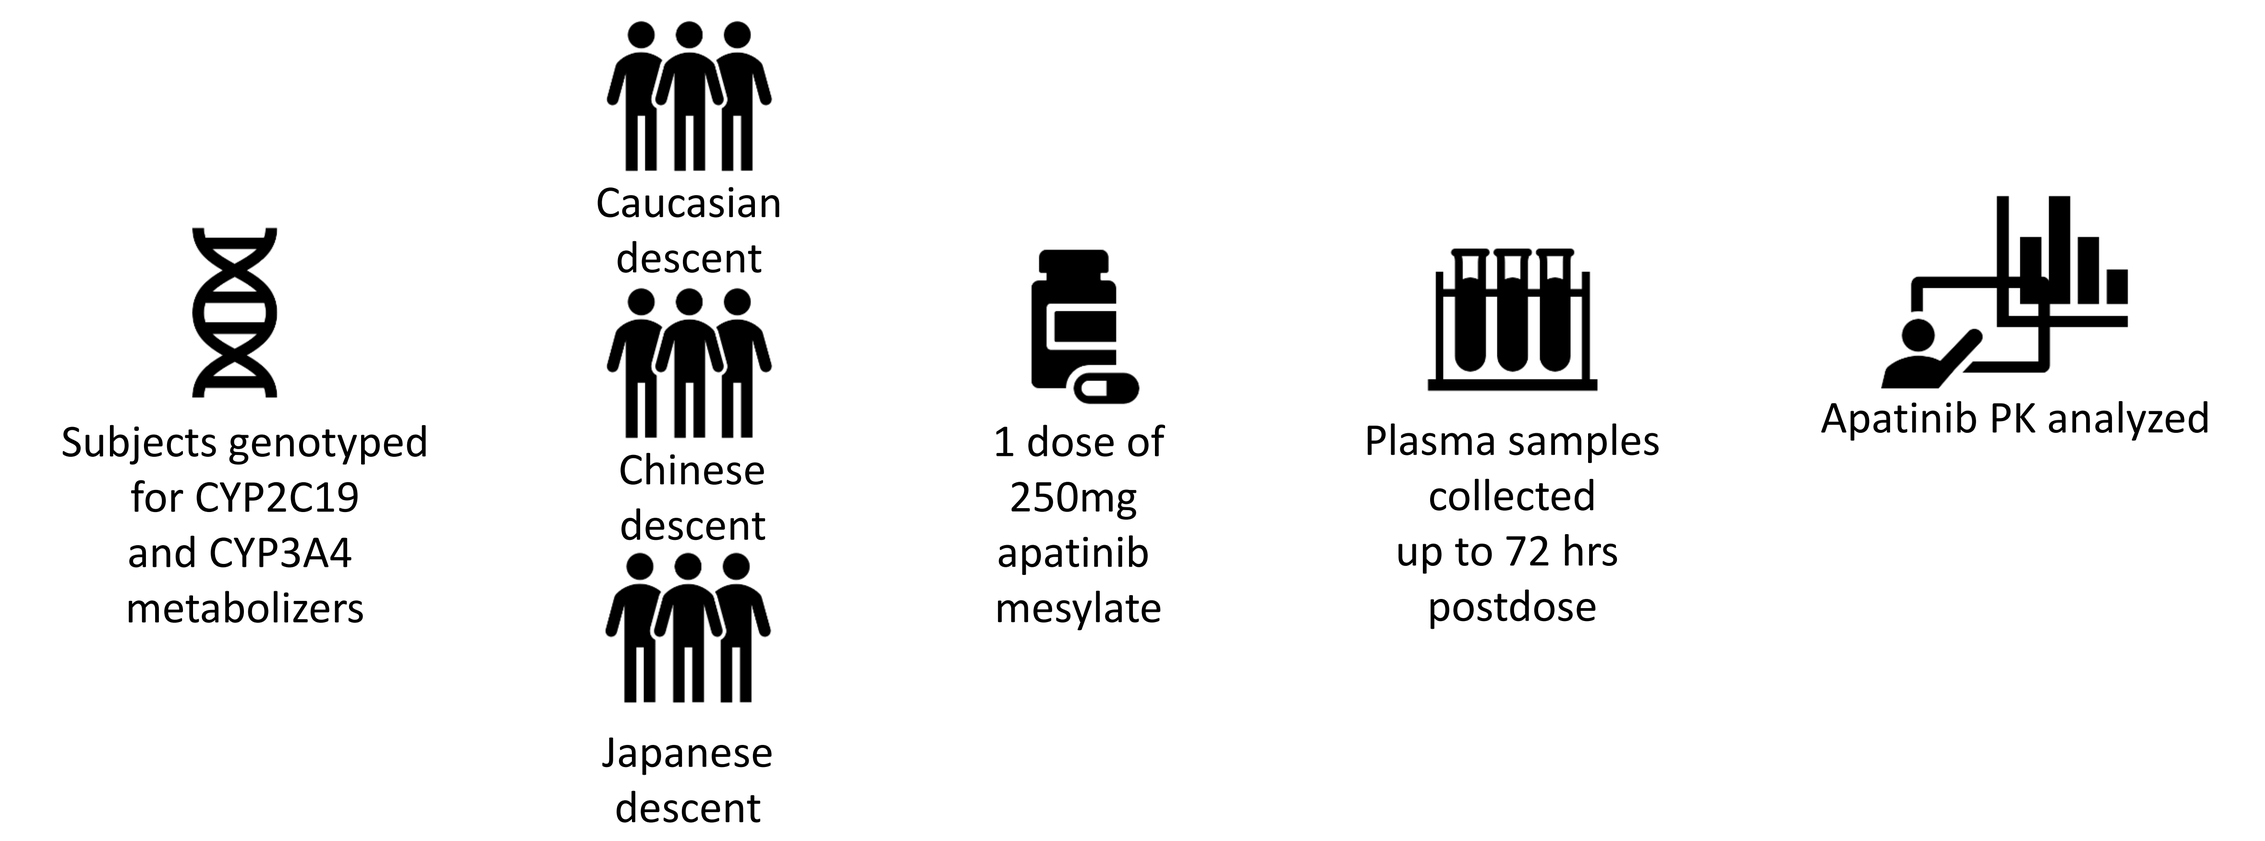
Subjects genotyped for CYP2C19 and CYP3A4 metabolizers
Caucasian descent
Chinese descent
Japanese descent
1 dose of 250mg apatinib mesylate
Plasma samples collected up to 72 hrs postdose
Apatinib PK analyzed

Database and PK Methodology

The database was created in accordance with FDA Guidelines 21 CFR Part 11 and was WinNonlin® compliant. WinNonlin 6.4 was used for analysis. There were no adjustments for covariates and there was no imputation (substitution) for missing data.

The following PK parameters for apatinib were estimated using noncompartmental analysis:

C_{max}	The maximum measured plasma concentration, obtained directly from the data without interpolation.
t_{max}	The time delay between drug administration and the first measurable (above LLOQ concentration).
t_{lag}	Area under the plasma concentration-time curve from time 0 to the last measurable concentration (where time is postdose), calculated by the linear trapezoidal method.
$AUC_{0\text{-}t}$	Percentage of the area extrapolated beyond the last quantifiable plasma concentration.
$AUC_{0\text{-}\infty}$	Terminal first-order elimination rate constant.
AUC_{extrap}	Percentage of the area extrapolated beyond the last quantifiable plasma concentration.
λ_z	Terminal first-order elimination rate constant.
$t_{1/2}$	The apparent first-order terminal elimination half-life, calculated as $0.693/\lambda_z$.
Cl/F	Apparent plasma clearance after oral administration.
V_z/F	Apparent volume of distribution after oral administration, uncorrected for bioavailability (F).

An exploratory compartmental analysis was conducted. The PK profile was adequately fitted to a 2-compartment model and the following parameters were estimated using compartmental analysis:

V_c/F	The volume of the central compartment after oral administration.
V_p/F	The volume of the peripheral compartment after oral administration.
k_a	The first-order absorption rate constant into central compartment.
k_{10}	The first-order elimination rate constant from the central compartment.
k_{12}	The first-order transfer rate constant from the central compartment to the peripheral compartment.
k_{21}	The first-order transfer rate constant from the peripheral compartment to the central compartment.

Statistical Methodology

Individual subject plasma concentrations, actual sampling times, and PK parameters obtained from the noncompartmental analysis for apatinib were summarized using the following descriptive statistics: number of observations, arithmetic mean, standard deviation (SD), coefficient of variation (CV%), minimum, median, and maximum. Geometric mean and geometric coefficient of variation (GCV%) were calculated for continuous PK variables. Descriptive statistics for t_{max} and t_{lag} only included number of observations, median, minimum, and maximum values.

Results

Table 1: Summary of Subject Demographics

	Caucasian (N=18)	Chinese (N=18)	Japanese (N=18)
Age	33.8 ± 7.1 yrs	34.2 ± 9.1 yrs	34.9 ± 8.7 yrs
Body Mass Index	23.4 ± 2.3 kg/m²	22.8 ± 2.7 kg/m²	22.4 ± 2.1 kg/m²

Table 2: Summary of Subject CYP Genotypes

Genotype	Caucasian (N=18)	Chinese (N=18)	Japanese (N=18)
CYP2C19		Number of Subjects	
Poor Metabolizers	0	3	3
Intermediate Metabolizers	8	11	7
Extensive Metabolizers	8	4	8
Ultrarapid Metabolizers	2	0	0
CYP3A4		Number of Subjects	
Intermediate Metabolizers	0	1	1
Extensive Metabolizers	18	17	17

Figure 1: Mean Plasma Concentrations vs. Time Profiles of Apatinib for Caucasian, Chinese, and Japanese Subjects: Linear and Log Scale

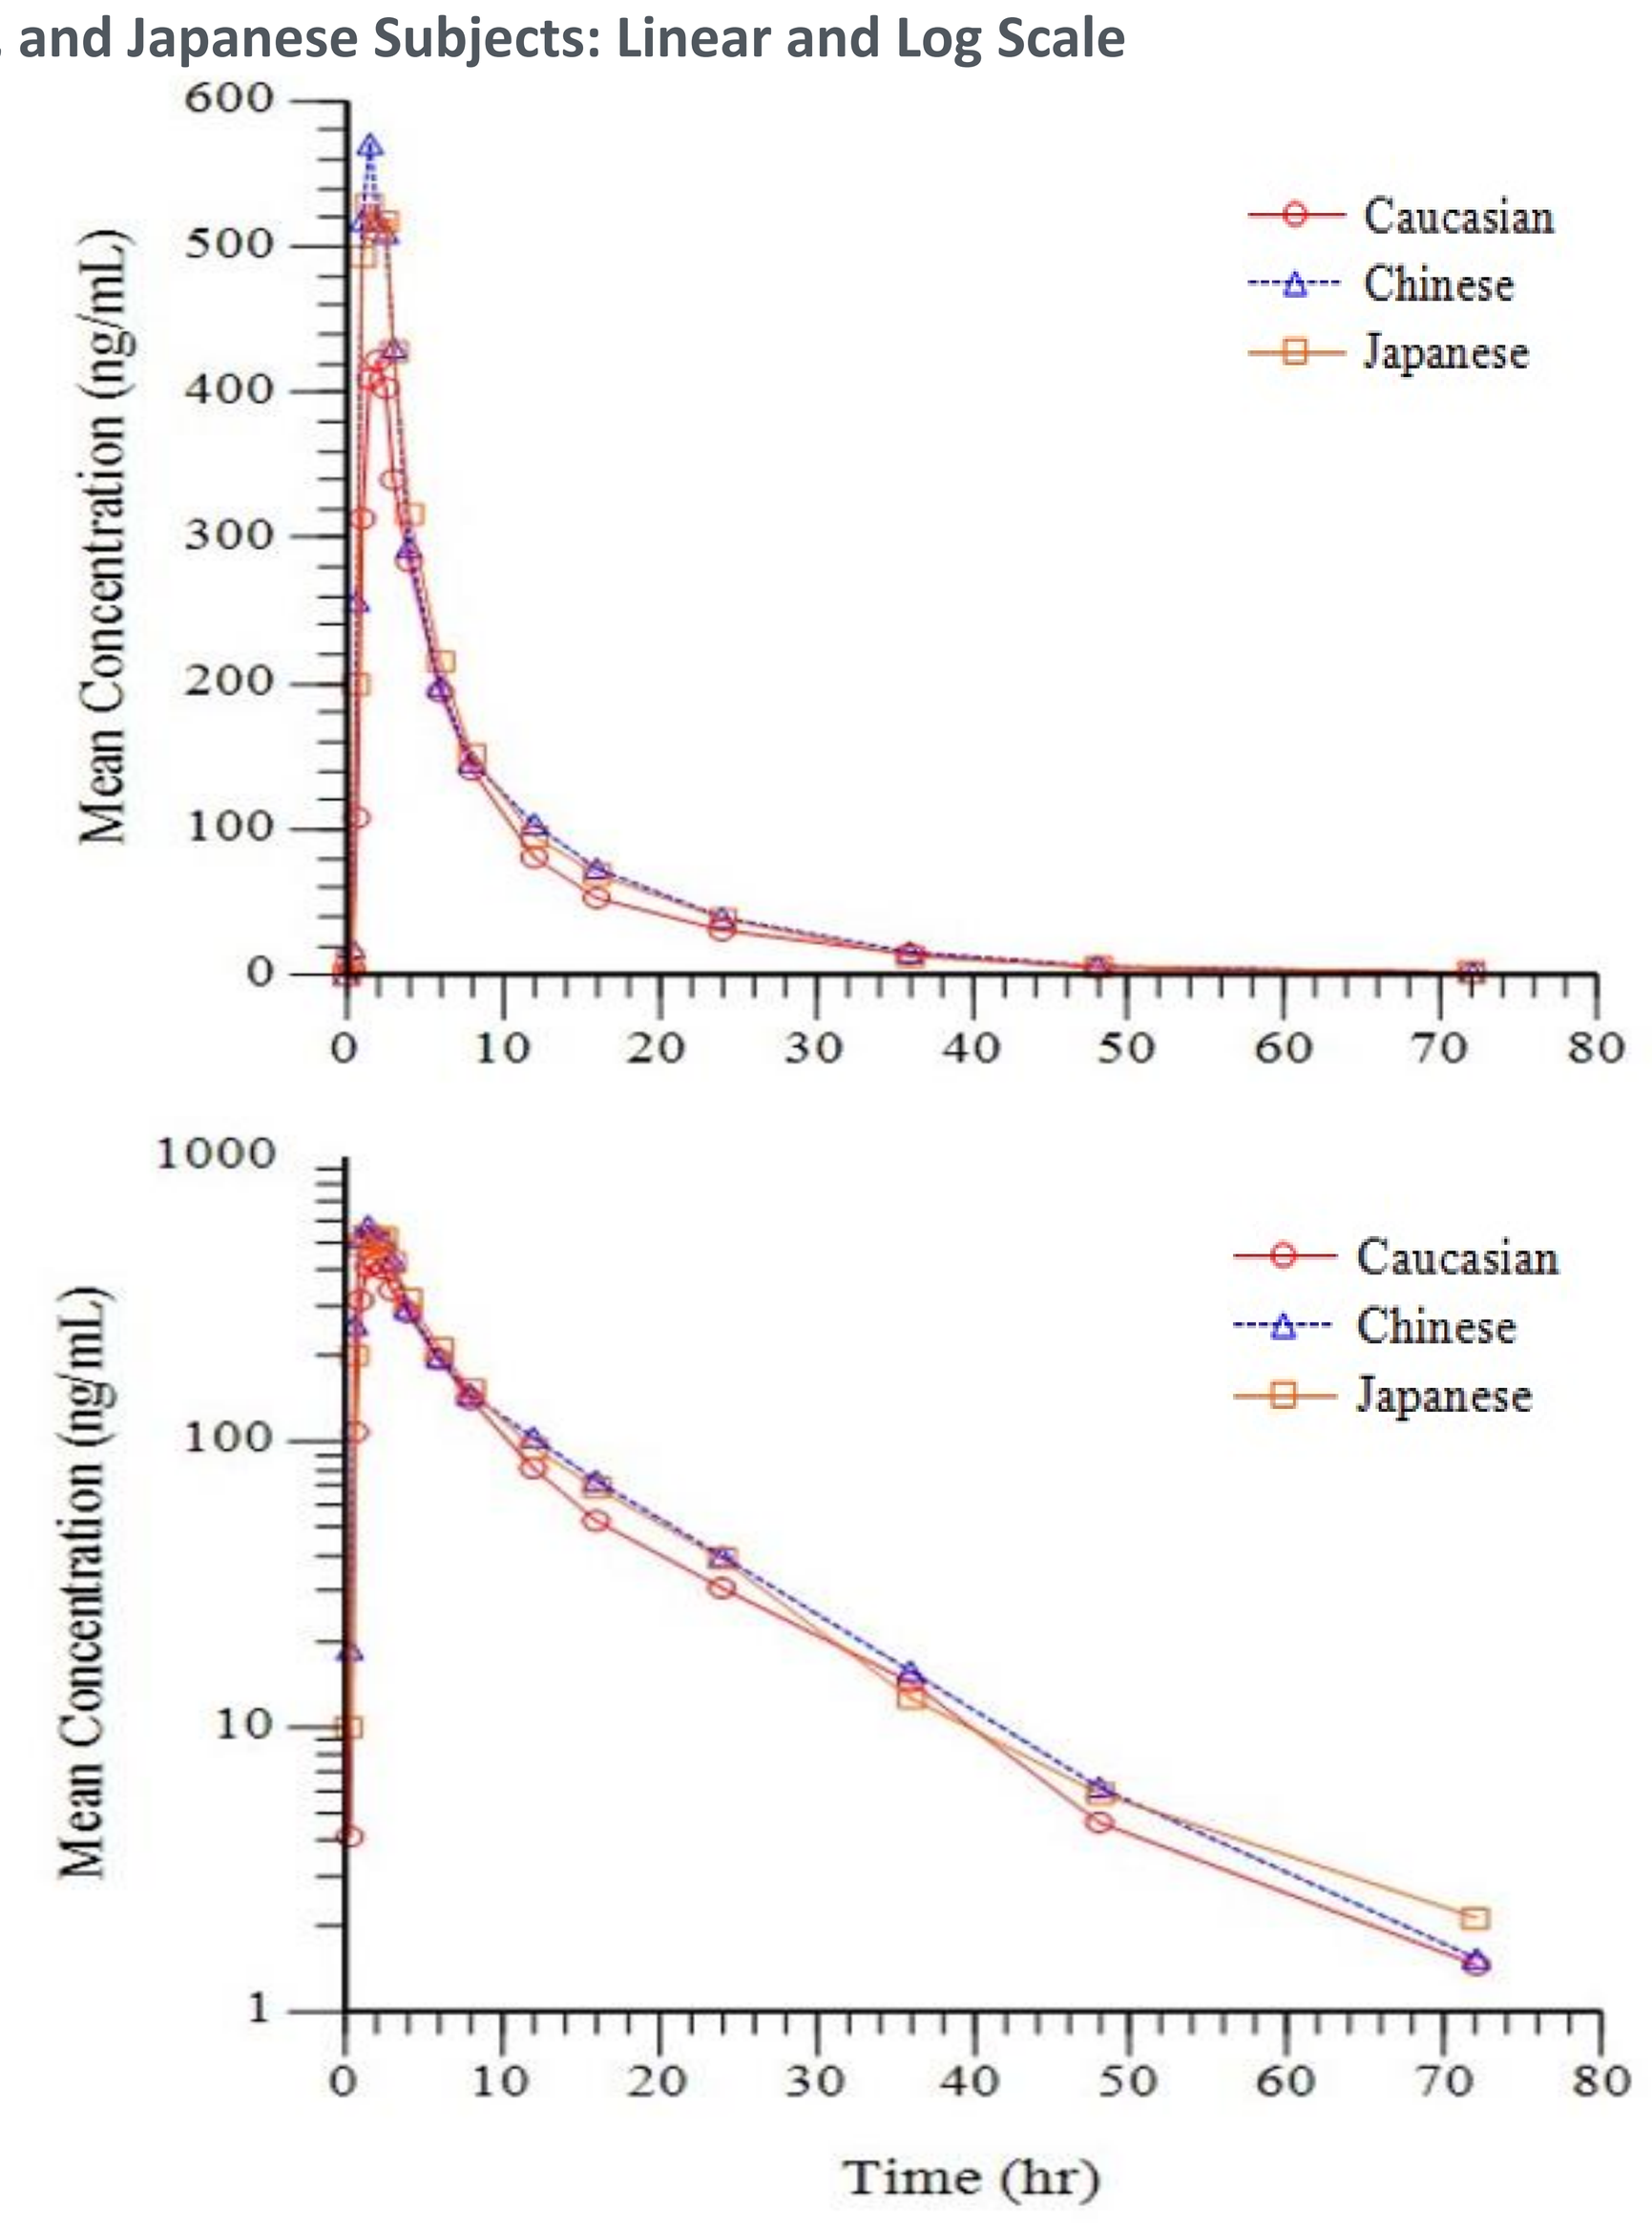
Mean Concentration (ng/mL)
600
500
400
300
200
100
0
0 10 20 30 40 50 60 70 80
Caucasian
Chinese
Japanese
1000
100
10
1
Time (hr)

Table 3: Summary Statistics of YN968D1 PK Parameters in Caucasian, Chinese, and Japanese Subjects

	Mean ± SD	Min	Median	Max	CV%	Geometric Mean	CV% Geometric Mean
			Caucasian (N=18)				
C_{max} (ng/mL)	478 ± 255	95.4	413	1090	53	411	67
t_{max} (hr)	-	1	2	6	-	-	-
t_{lag} (hr)	-	0	0	0.33	-	-	-
$AUC_{0\text{-}t}$ (hr·ng/mL)	3475 ± 1527	1316	2925	6486	44	3160	48
$AUC_{0\text{-}\infty}$ (hr·ng/mL)	3496 ± 1529	1000	2946	6503	44	3182	48
AUC_{extrap} (%)	0.68 ± 0.41	0.079	0.57	1.4	60	0.54	88
λz (1/hr)	0.0943 ± 0.0296	0.0589	0.0915	0.176	31	0.0905	30
Half-Life (hr)	7.96 ± 2.21	3.95	7.57	11.8	28	7.66	30
Cl/F (mL/min)	1448 ± 703	641	1414	3157	49	1310	48
Vz/F (L)	995 ± 588	263	899	2959	59	868	58
			Chinese (N=18)				
C_{max} (ng/mL)	701 ± 425	143	635	1690	61	570	81
t_{max} (hr)	-	-	0.72	1.5	2.5	-	-
t_{lag} (hr)	-	-	0	0	0.33	-	-
$AUC_{0\text{-}t}$ (hr·ng/mL)	4206 ± 2518	1677	3411	10685	60	3673	55
$AUC_{0\text{-}\infty}$ (hr·ng/mL)	4246 ± 2532	2000	3462	10707	60	3711	55
AUC_{extrap} (%)	1 ± 0.83	0.14	0.82	3	81	0.72	114
λz (1/hr)	0.0978 ± 0.0296	0.0682	0.0935	0.191	30	0.0944	27
Half-Life (hr)	7.56 ± 1.78	3.63	7.42	10.2	24	7.34	27
Cl/F (mL/min)	1256 ± 569	389	1210	2443	45	1123	55
Vz/F (L)	786 ± 351	289	749	1601	45	713	49
			Japanese (N=18)				
C_{max} (ng/mL)	647 ± 299	130	595	1400	46	575	58
t_{max} (hr)	-	1	1.5	3	-	-	-
t_{lag} (hr)	-	0	0	0.33	-	-	-
$AUC_{0\text{-}t}$ (hr·ng/mL)	4123 ± 1631	1441	3779	6944	40	3804	45
$AUC_{0\text{-}\infty}$ (hr·ng/mL)	4169 ± 1662	1000	3860	7098	40	3842	45
AUC_{extrap} (%)	0.99 ± 1.3	0.13	0.79	5.5	129	0.57	146
λz (1/hr)	0.105 ± 0.0385	0.045	0.0957	0.189	37	0.0986	38
Half-Life (hr)	7.5 ± 2.91	3.66	7.25	15.4	39	7.03	38
Cl/F (mL/min)	1189 ± 568	587	1080	2866	48	1084	45
Vz/F (L)	727 ± 344	332	633	1613	47	660	47

Abbreviations: - = not calculable, CV = coefficient of variation, Max = maximum, Min = minimum.

Figure 2. Box Plot of C_{max}, t_{max}, $AUC_{0\text{-}t}$, $AUC_{0\text{-}\infty}$ vs. Caucasian, Chinese, and Japanese Subjects

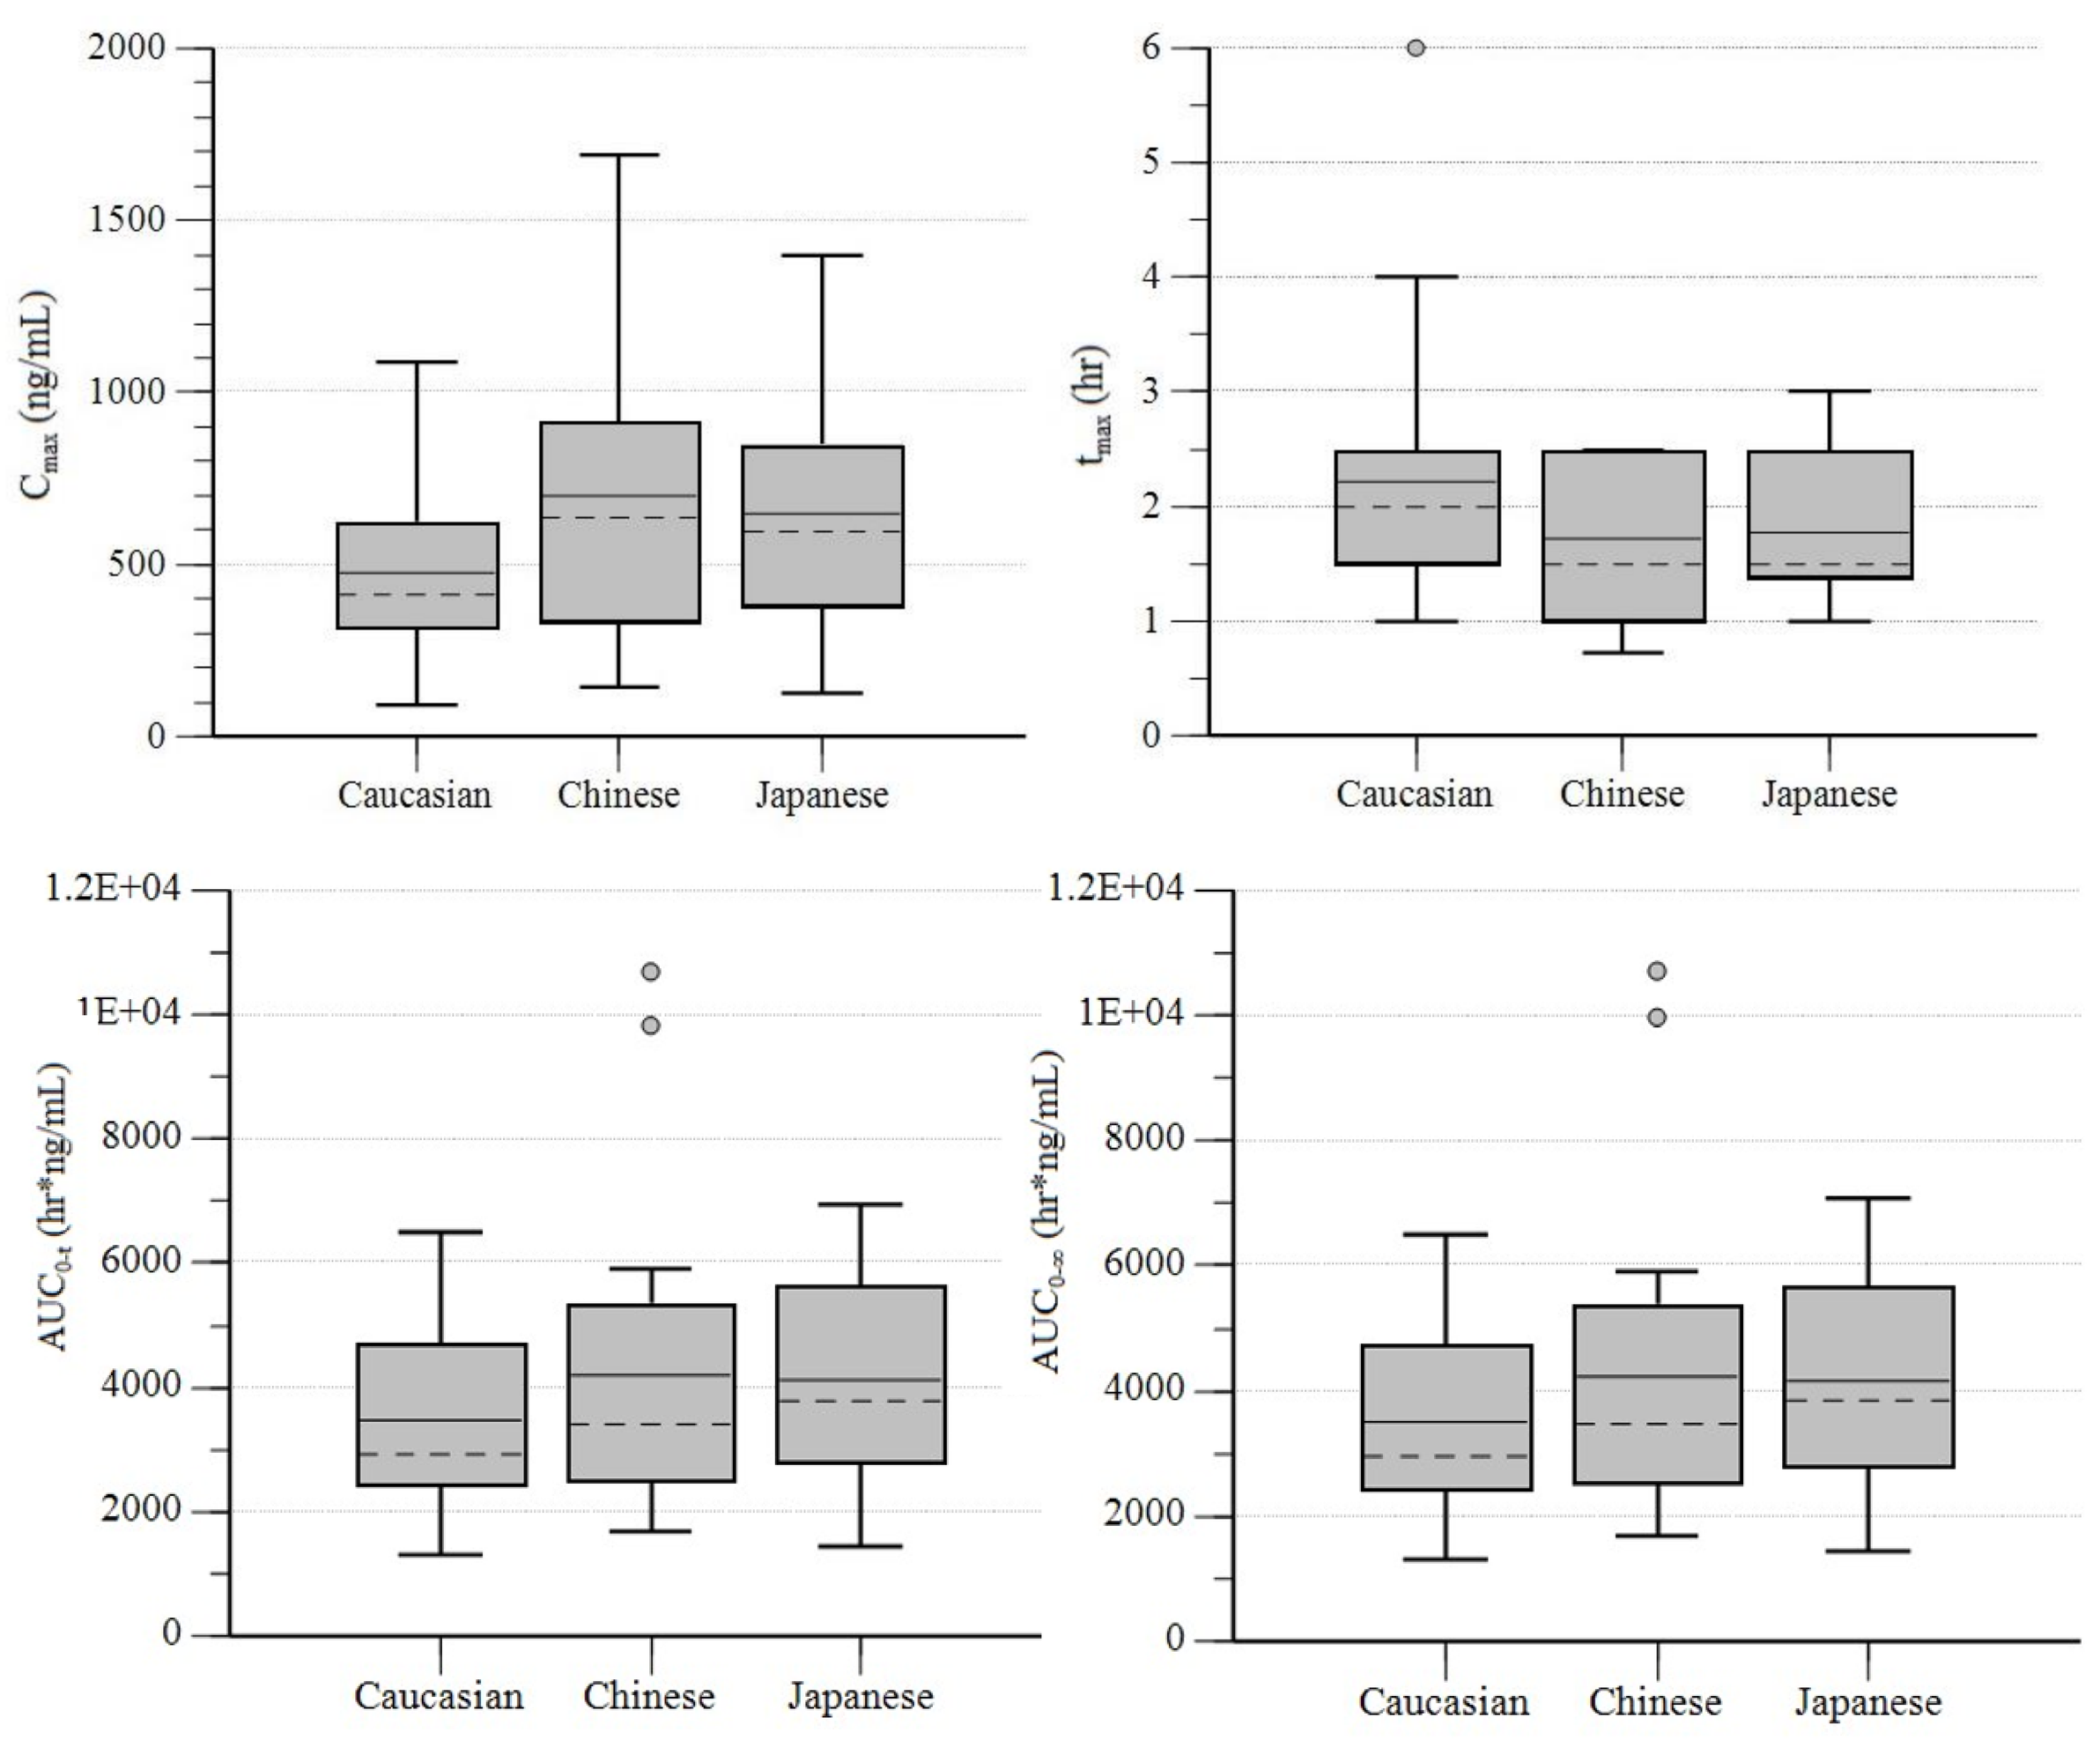
Cmax (ng/mL)
2000
1500
1000
500
0
Caucasian
Chinese
Japanese
tmax (hr)
6
5
4
3
2
1
0
AUC0-t (hr*ng/mL)
AUC0-∞ (hr*ng/mL)
1.2E+04
1E+04
8000
6000
4000
2000
0

Note: The dashed line (---) is the median; the solid line (-) is the arithmetic mean. The ends of the "box" (⊥) are the 25th and 75th percentiles, which are computed by the same method as in Descriptive Stats. These are also referred to as the first and third quartiles. The whiskers show the lowest data value still within 1.5 IQR of the lower quartile, and the highest value still within 1.5 IQR of the upper quartile, where IQR is the interquartile range (the difference between the third and first quartiles, the middle 50%). Data values that do not fall between the whiskers are plotted as outliers (●), markers outside of the whiskers.

Table 6: Statistical Comparison of Mean for C_{max} and AUC

	Number DF	Denom DF	P Value
C_{max}	2	51	0.12
$AUC_{0\text{-}\infty}$	2	51	0.46
$AUC_{0\text{-}t}$	2	51	0.47

Abbreviations: Number = numerator, DF = degrees of freedom, Denom = denominator.

Table 4. Comparison of C_{max}, $AUC_{0\text{-}t}$, $AUC_{0\text{-}\infty}$ among Caucasian, Chinese, and Japanese Subjects

	Mean ± SD	Min	Median	Max	CV%	Geometric Mean	CV% Geometric Mean
			C_{max} (ng/mL)				
Caucasian (N=18)	478 ± 255	95.4	413	1090	53	411	67
Chinese (N=18)	701 ± 425	143	635	1690	61	570	81
Japanese (N=18)	647 ± 299	130	595	1400	46	575	58
			$AUC_{0\text{-}t}$ (hr·ng/mL)				
Caucasian (N=18)	3475 ± 1527	1316	2925	6486	44	3160	48
Chinese (N=18)	4206 ± 2518	1677	3411	10685	60	3673	55
Japanese (N=18)	4123 ± 1631	1441	3779	6944	40	3804	45
			$AUC_{0\text{-}\infty}$ (hr·ng/mL)				
Caucasian (N=18)	3496 ± 1529	1000	2946	6503	44	3182	48
Chinese (N=18)	4246 ± 2532	2000	3462	10707	60	3711	55
Japanese (N=18)	4169 ± 1662	1000	3860	7098	40	3842	45

Abbreviations: CV = coefficient of variation, Max = maximum, Min = minimum.

Table 5: Summary Statistics of YN968D1 PK Parameters using Compartment Analysis in Caucasian, Chinese, and Japanese Subjects

	Mean ± SD	Min	Median	Max	CV%	Geometric Mean	CV% Geometric Mean
			Caucasian (N=15)				
K_a (1/hr)	2.02 ± 1.56	0.682	1.57	6.99	78	1.66	67
K_{10} (1/hr)	0.222 ± 0.0768	0.135	0.196	0.348	35	0.211	35
K_{12} (1/hr)	0.217 ± 0.193	0.0164	0.169	0.655	89	0.151	120
K_{21} (1/hr)	0.175 ± 0.101	0.0781	0.133	0.454	58	0.155	53
V_c/F (L)	444 ± 290	124	342	1300	65	376	64
V_p/F (L)	478 ± 430	106	381	1850	90	368	82
			Chinese (N=17)				
K_a (1/hr)	3.07 ± 5.17	0.642	1.25	21.6	169	1.69	116
K_{10} (1/hr)	0.312 ± 0.145	0.162	0.266	0.784	47	0.288	40
K_{12} (1/hr)	0.418 ± 0.255	0.00771	0.377	0.891	61	0.303	155
K_{21} (1/hr)	0.223 ± 0.0793	0.0496	0.233	0.339	36	0.205	50
V_c/F (L)	266 ± 164	99.4	210	633	61	227	62
V_p/F (L)	450 ± 410	61.9	325	1750	91	337	92
			Japanese (N=16)				
K_a (1/hr)	2.44 ± 2.37	0.532	1.25	8.39	97	1.7	101
K_{10} (1/hr)	0.277 ± 0.121	0.118	0.29	0.569	44	0.252	49
K_{12} (1/hr)	0.315 ± 0.236	0.0344	0.242	0.747	75	0.217	132
K_{21} (1/hr)	0.21 ± 0.0816	0.11	0.187	0.409	39	0.197	37
V_c/F (L)	328 ± 319	127	255	1480	97	266	62
V_p/F (L)	357 ± 209	56.4	316	732	58	292	82

Abbreviations: CV = coefficient of variation, k_{10} = first-order elimination rate constant from the central compartment, k_{12} = first-order transfer rate constant from the central compartment to the peripheral compartment, k_{21} = first-order transfer rate constant from the peripheral compartment to the central compartment, k_a = first-order absorption rate constant into central compartment, Max = maximum, Min = minimum, Vc/F = volume of the central compartment after oral administration, Vp/F = volume of the peripheral compartment after oral administration.

Conclusion

The results indicated that the mean C_{max} and $AUC_{0\text{-}\infty}$ values in Chinese and Japanese subjects were slightly greater compared to Caucasian subjects. The mean clearance values also showed a similar trend. However, none of these differences were statistically significant. In addition, the mean half-life ($t_{1/2}$) values were also very similar and averaged 7.5 hours to 8 hours amongst the 3 subject groups. The body weights of the Chinese and Japanese subjects were lower compared to the Caucasian subjects, which might explain the slight differences in exposure.

References

1. Li J, et al. *J Clin Oncol* 2016;34(13):1448-1454.
2. Kang YK, et al. *Ann Oncol* 2017;35(suppl_15):TPS4138.

Acknowledgements

We extend our thanks to the participants, the investigators, and their site staff members who made this trial possible. This study is supported by LSK BioPartners, Inc. (dba LSK BioPharma).

Contact Information

This poster was presented at the 2017 American College of Clinical Pharmacology (Sept 17-19, 2017); San Diego, CA.

Corresponding Author:
arlo.mcginn@lskbiopharma.com