A Phase 1 Study of Rivoceranib Combined with Nivolumab in Patients with Unresectable or Metastatic Cancer

Presented at ESMO 2024

FPN# 1013P

Neal S. Chawla,[1] CH Park,[2] Erlinda M. Gordon,[1] Sant Chawla,[1] Xianzhang Meng,[2] Bill Strickland,[2] Lynda Finis,[2] Chris Galloway[2]

[1]Sarcoma Oncology Research Center, Santa Monica, CA, USA; [2]Elevar Therapeutics, Fort Lee, NJ, USA

Copies of this poster obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- Rivoceranib (also known as apatinib, rivoceranib mesylate, and YN968D1) is an orally administered small-molecule receptor tyrosine kinase inhibitor (TKI), which selectively inhibits vascular endothelial growth factor receptor (VEGFR)-2 to suppress endothelial cell migration and proliferation, and by reducing the tumor microvascular density, induces an antiangiogenic and antineoplastic effect.
- Rivoceranib is approved for the treatment of gastric cancer, a second-line treatment for patients with advanced hepatocellular carcinoma (HCC), and in combination with camrelizumab for the first-line treatment of patients with unresectable or metastatic HCC in China.
- Phase 1/2 studies of combination treatment strategies of various PD-1/PD-L1 inhibitors with other VEGF/VEGFR TKIs have shown promising efficacy data with an objective response rate (ORR) ranging from 44% to 71%, depending on the studied regimens and patient populations.[1-4]
- This Phase 1 study (NCT03396211) evaluated the safety, tolerability, preliminary efficacy, and selected immunologic effects of the combination of rivoceranib and nivolumab in patients with advanced or metastatic solid tumors.

METHODS

Study Design

- Prospective, open-label, single-center, Phase 1 study evaluating the safety, tolerability, preliminary efficacy, and selected immunologic effects of rivoceranib in patients with advanced or metastatic cancer who were receiving nivolumab therapy concurrently. The study contained 2 parts and each part provided preliminary efficacy data for the treatment combination. The overall design scheme is shown in **Figure 1**.

Figure 1: Study Design

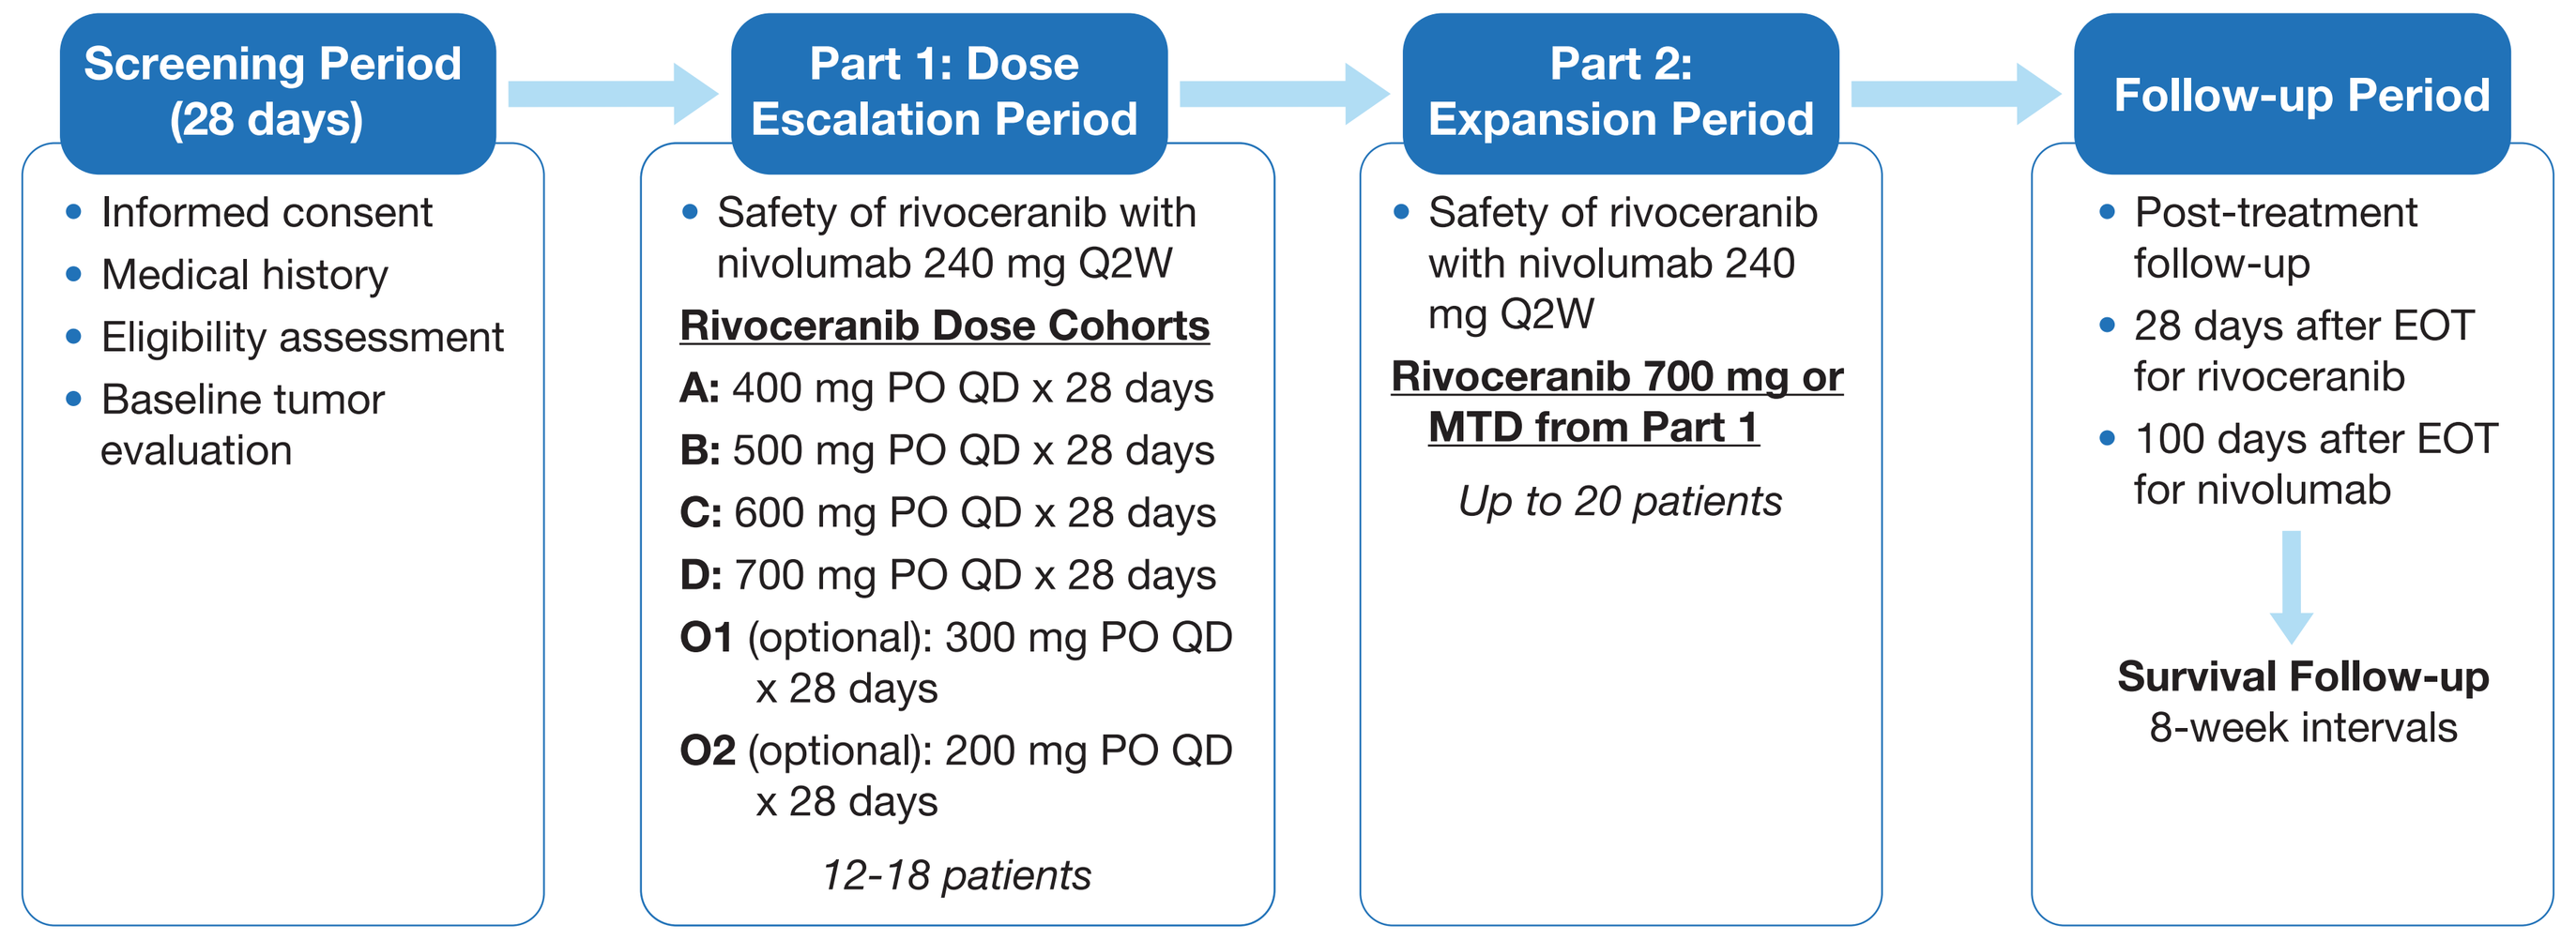
Screening Period (28 days)
Informed consent
Medical history
Eligibility assessment
Baseline tumor evaluation
Part 1: Dose Escalation Period
Safety of rivoceranib with nivolumab 240 mg Q2W
Rivoceranib Dose Cohorts
A: 400 mg PO QD x 28 days
B: 500 mg PO QD x 28 days
C: 600 mg PO QD x 28 days
D: 700 mg PO QD x 28 days
O1 (optional): 300 mg PO QD x 28 days
O2 (optional): 200 mg PO QD x 28 days
12-18 patients
Part 2: Expansion Period
Safety of rivoceranib with nivolumab 240 mg Q2W
Rivoceranib 700 mg or MTD from Part 1
Up to 20 patients
Follow-up Period
Post-treatment follow-up
28 days after EOT for rivoceranib
100 days after EOT for nivolumab
Survival Follow-up
8-week intervals

Parts 1 and 2: Tumor assessment at 6-8 weeks per standard of care. Safety assessments every 2 weeks.

EOT, end of treatment; MTD, maximum tolerated dose; PO, orally; Q2W, once every 2 weeks; QD, once daily.

Key Inclusion Criteria

- Male or female ≥18 years of age
- Documented primary diagnosis of histologic- or cytologic-confirmed solid tumor cancer inclusive of gastric adenocarcinoma, renal cell carcinoma, melanoma, non-small cell lung cancer, breast cancer, angiosarcoma, leiomyosarcoma, synovial sarcoma, and alveolar soft part sarcoma, or other solid tumor for which anti-VEGFR-2 targeted therapy could have been applicable
- Locally advanced unresectable or metastatic disease
- Nivolumab treatment naïve and able to begin nivolumab treatment concurrently with initiation of rivoceranib or had received at least 3 doses of nivolumab treatment and was continuing nivolumab therapy
- ≥ 1 measurable lesions per Response Evaluation Criteria in Solid Tumors version 1.1 (RECIST v1.1)

Objectives

PRIMARY

- Safety/tolerability of rivoceranib + nivolumab in patients with unresectable or metastatic cancer
- Objective response rate, best overall response, time-to-response, and duration of response per RECIST v1.1 and/or immune RECIST (iRECIST)
- Disease control rate and duration of disease control per RECIST v1.1 and/or iRECIST

SECONDARY

- Overall survival
- Event-free survival
- Progression-free survival

EXPLORATORY

- Tumor mutational burden and mutations at baseline and at the time of progression
- Change in serum cytokines pursuant to treatment response
- Change in peripheral blood mononuclear cell subsets and myeloid-derived suppressor cell populations as determined by flow cytometry

RESULTS

Patient Disposition

- A total of 30 patients were enrolled in the study.
 - In Part 1, 3 patients in Cohort A received rivoceranib 400 mg QD plus nivolumab 240 mg Q2W; 7 patients in Cohort B received rivoceranib 300 mg QD and nivolumab 240 mg Q2W.
 - In Part 2, 20 patients received rivoceranib 300 mg QD and nivolumab 240 mg Q2W.
- All of the 30 enrolled patients discontinued from the study: 11 patients (37%) due to confirmed disease progression; 5 patients (17%) due to intolerable toxicity; 4 patients (13%) due to clinically indicated disease progression; 3 patients (10%) each due to Investigator discretion, dose interruption of > 28 days, and death in long-term follow-up; and 1 patient (3%) due to regulatory request.
- Demographics and baseline characteristics are summarized in **Table 1**.

Table 1: Demographics and Baseline Characteristics

Parameter	Part 1 Cohort A 400 mg (N=3)	Part 1 Cohort B 300 mg (N=7)	Part 2 Cohort 300 mg (N=20)	Overall (N=30)
Median age, years	44	50	59	53
Male sex, n (%)	2 (66.7)	2 (28.6)	11 (55.0)	15 (50.0)
Race				
American Indian or Alaska Native	0	0	0	0
Asian	1 (33.3)	1 (14.3)	0	2 (6.7)
Black or African American	0	0	1 (5.0)	1 (3.3)
Native Hawaiian or Other Pacific Islander	0	0	0	0
White	2 (66.7)	6 (85.7)	19 (95.0)	27 (90.0)
Other	0	0	0	0
Ethnicity				
Hispanic or Latino	2 (66.7)	2 (28.6)	10 (50.0)	14 (46.7)
Not Hispanic or Latino	1 (33.3)	5 (71.4)	10 (50.0)	16 (53.3)
ECOG				
0	0	0	0	0
1	3 (100.0)	7 (100.0)	20 (100.0)	30 (100.0)

Dose Limiting Toxicities

- Two patients in Part 1 Cohort A had a dose-limiting toxicity (DLT) of Grade 3 hypertension. Due to the observed DLTs at rivoceranib 400 mg, no further dose escalation was performed for rivoceranib as planned. Part 1 Cohort B evaluated rivoceranib 300 mg, where 1 patient had a DLT of Grade 3 hypertension.
- Therefore, the maximum tolerated dose was rivoceranib 300 mg/day combined with nivolumab 240 mg IV Q2W; this dose combination was set as the dose for the expansion cohort in Part 2 of the study.
- All DLTs were considered probably related to rivoceranib but unrelated to nivolumab; all DLTs recovered after medication treatment.

Safety

- In Part 1 Cohort A, common (reported in ≥ 2 patients) Grade 3 treatment-emergent adverse events (TEAEs) were hypertension (n=2, 66.7%), generalized rash, ALT increased, and neutropenia (n=1 each, 33.3%). No Grade ≥ 4 TEAEs were observed for this cohort.
- In Part 1 Cohort B, common (reported in ≥ 2 patients) Grade 3 TEAEs were anemia (n=3, 42.9%) asthenia and hyponatremia (n=2, 28.6%). One patient experienced Grade 5 TEAEs of acute respiratory failure and pulseless electrical activity; both events were fatal and assessed as treatment-unrelated.
- In Part 2 Cohort, common (reported in ≥ 2 patients) Grade ≥ 3 TEAEs were anemia (n= 6, 30.0%), hypophosphatemia (n=4, 20.0%), fatigue, nausea, abdominal pain, hypokalemia, dyspnea, thrombocytopenia, and neutropenia (n=3 each, 15.0%), hypoalbuminemia, acute respiratory failure, ALT increased, peritonitis bacterial, and hypotension (n=2 each, 10.0%). Two patients experienced Grade ≥ 4 TEAEs including Grade 4 neutropenia, thrombocytopenia, anemia, and acute respiratory failure, and Grade 5 acute myeloid leukemia and respiratory distress. Both Grade 5 events were fatal and assessed as treatment-unrelated.

Table 2: Summary of Treatment-Emergent Adverse Events by Cohort and Overall

Parameter, n (%)	Part 1 Cohort A 400 mg (N=3)	Part 1 Cohort B 300 mg (N=7)	Part 2 Cohort 300 mg (N=20)	Overall (N=30)
Maximum TEAE severity grade				
1	0	0	1 (5.0)	1 (3.3)
2	1 (33.3)	2 (28.6)	3 (15.0)	6 (20.0)
3	2 (66.7)	4 (57.1)	14 (70.0)	20 (66.7)
4	0	0	0	0
5	0	1 (14.3)	2 (10.0)	3 (10.0)
≥3	2 (66.7)	5 (71.4)	16 (80.0)	23 (76.7)
TEAE related to rivoceranib	3 (100.0)	7 (100.0)	19 (95.0)	29 (96.7)
TEAE related to nivolumab	2 (66.7)	3 (42.9)	16 (80.0)	21 (70.0)

Efficacy

- Of the 30 patients enrolled, 5 patients experienced a partial response with an objective response rate of 16.7% (95% CI: 5.64–34.72) across treatment cohorts (**Table 3**).

Table 3: Summary of Objective Response Rate and Best Overall Response by Cohort

Parameter	Part 1 Cohort A 400 mg (N=3)	Part 1 Cohort B 300 mg (N=7)	Part 2 Cohort 300 mg (N=20)	Overall (N=30)
ORR, n (%) [95% CI]	0	1 (14.3) [0.4, 57.9]	4 (20.0) [5.7, 43.7]	5 (16.7) [5.6, 34.7]
Best overall response, n (%)				
Complete response	0	0	0	0
Partial response	0	1 (14.3)	4 (20.0)	5 (16.7)
Stable disease	3 (100)	3 (42.9)	13 (65.0)	19 (63.3)
Progressive disease	0	1 (14.3)	2 (10.0)	3 (10.0)
Not evaluable	0	0	0	0
Missing	0	2 (28.6)	1 (5.0)	3 (10.0)

ORR, objective response rate.

- Among the 5 patients who experienced a partial response, the median time between the first documented response and enrollment was 5.32 months, and the median time between the first documented response and the date of first disease progression or death from any cause was 1.97 months. The overall disease control rate was 80% (**Table 4**).

Table 4: Summary of Time-to-Response, Duration of Response, and Disease Control Rate

Parameter	Part 1 Cohort A 400 mg (N=3)	Part 1 Cohort B 300 mg (N=7)	Part 2 Cohort 300 mg (N=20)	Overall (N=30)
Median TTR, months	NA	5.55	4.50	5.32
Median DoR, months	NA	1.77	2.81	1.97
DCR, n (%)	3 (100.0)	4 (57.1)	17 (85.0)	24 (80.0)

DCR, disease control rate; DoR, duration of response; NA, not applicable; TTR, time to response.

- Patients in Part 1 Cohort B and Part 2 Cohort demonstrated relatively larger reductions in sum of target lesions over time compared with patients in Part 1 Cohort A (**Figure 2**).

Figure 2: Waterfall Plot of Tumor Burden Changes from Baseline (Full Analysis Set: N=30)

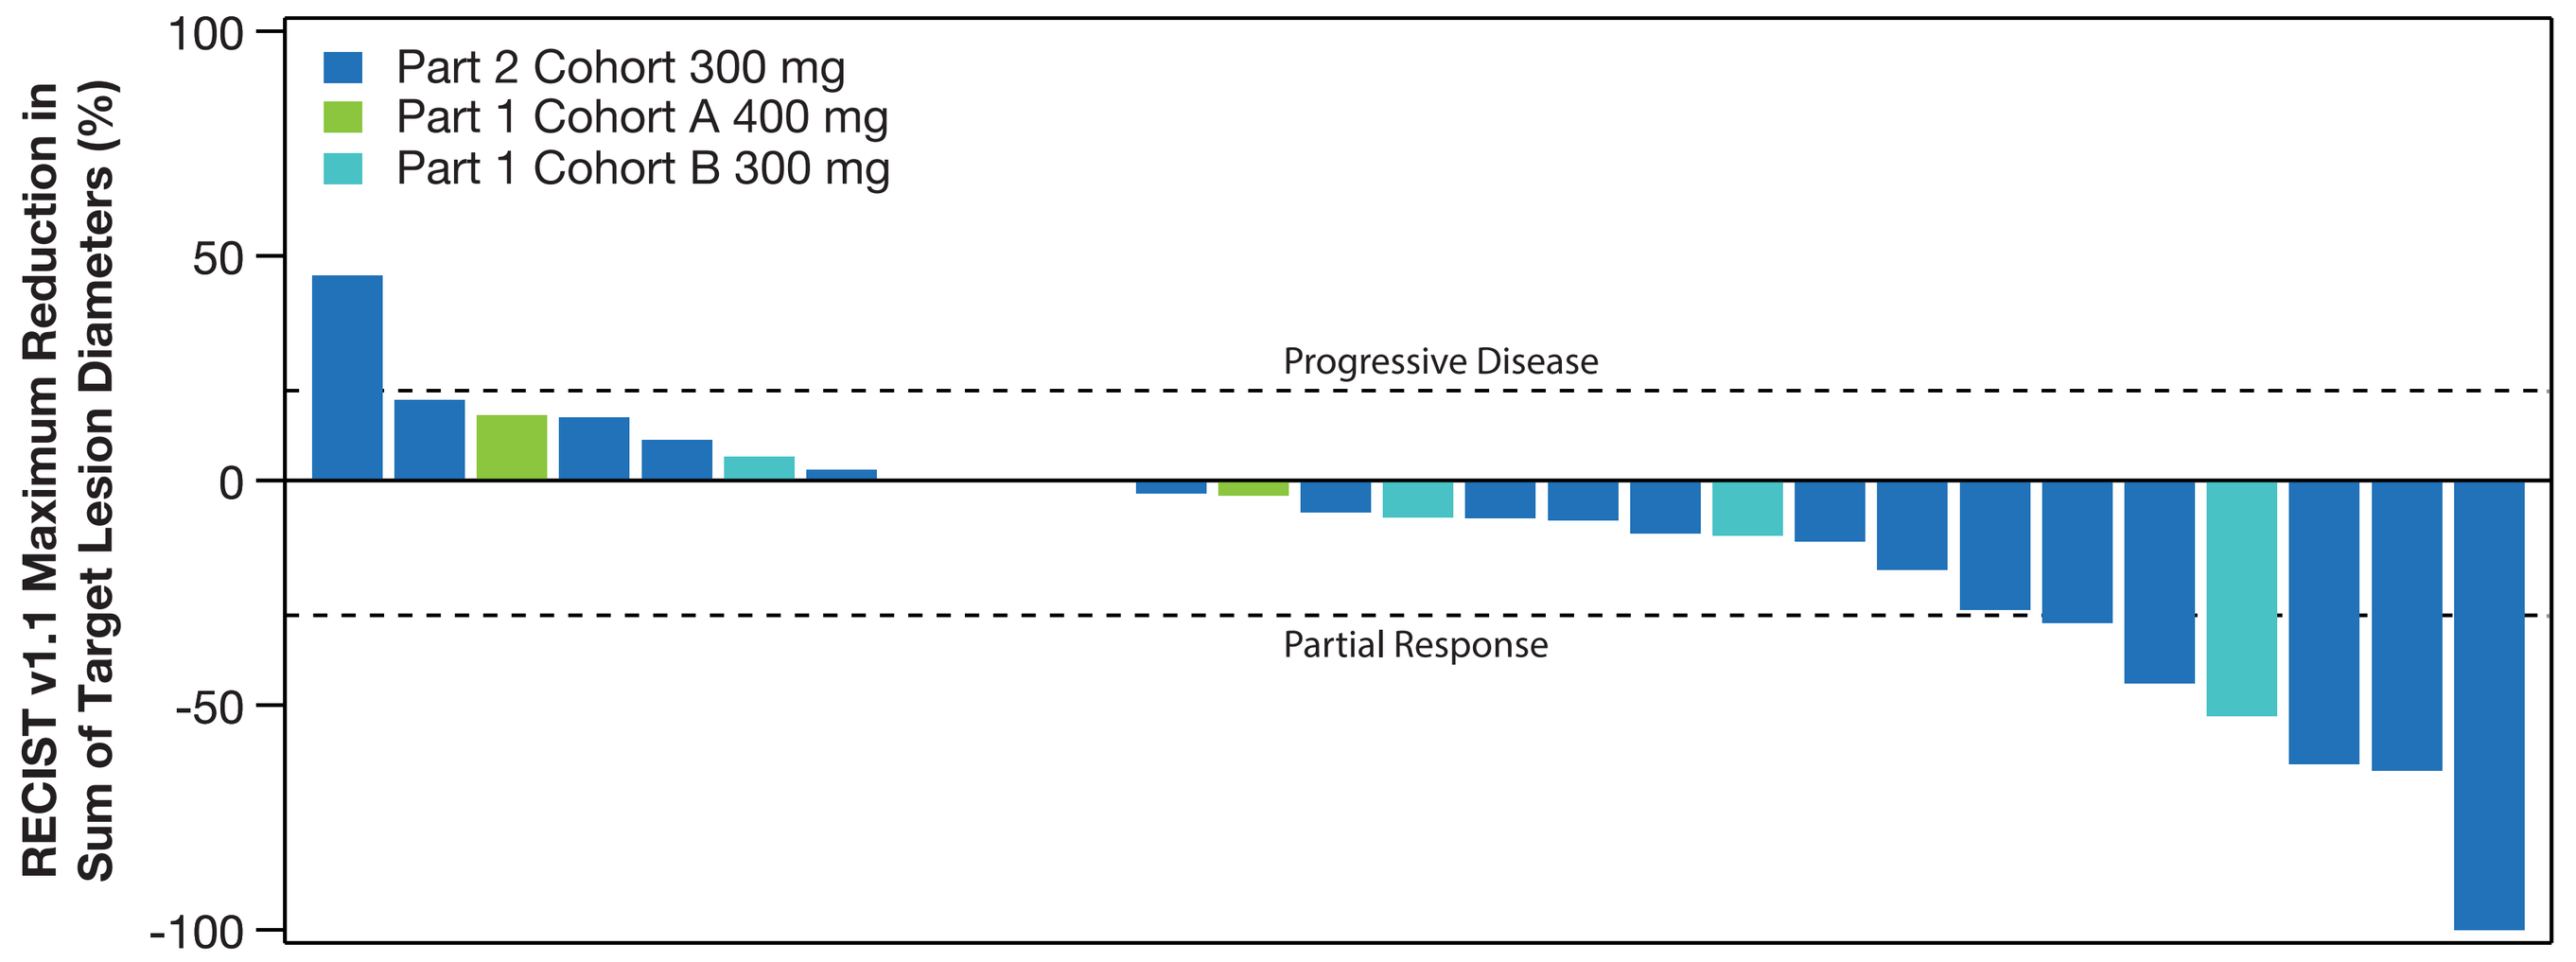
RECIST v1.1 Maximum Reduction in Sum of Target Lesion Diameters (%)
100
50
0
-50
-100
Part 2 Cohort 300 mg
Part 1 Cohort A 400 mg
Part 1 Cohort B 300 mg
Progressive Disease
Partial Response

Efficacy *(continued)*

- The median overall survival was 7.13 months (95% CI: 3.68–9.30) across treatment cohorts. The median progression-free survival was 5.49 months (95% CI: 3.68–9.30) across treatment cohorts.

Figure 3: Kaplan-Meier Survival Curve for Oveall Survival (Full Analysis Set: N=30)

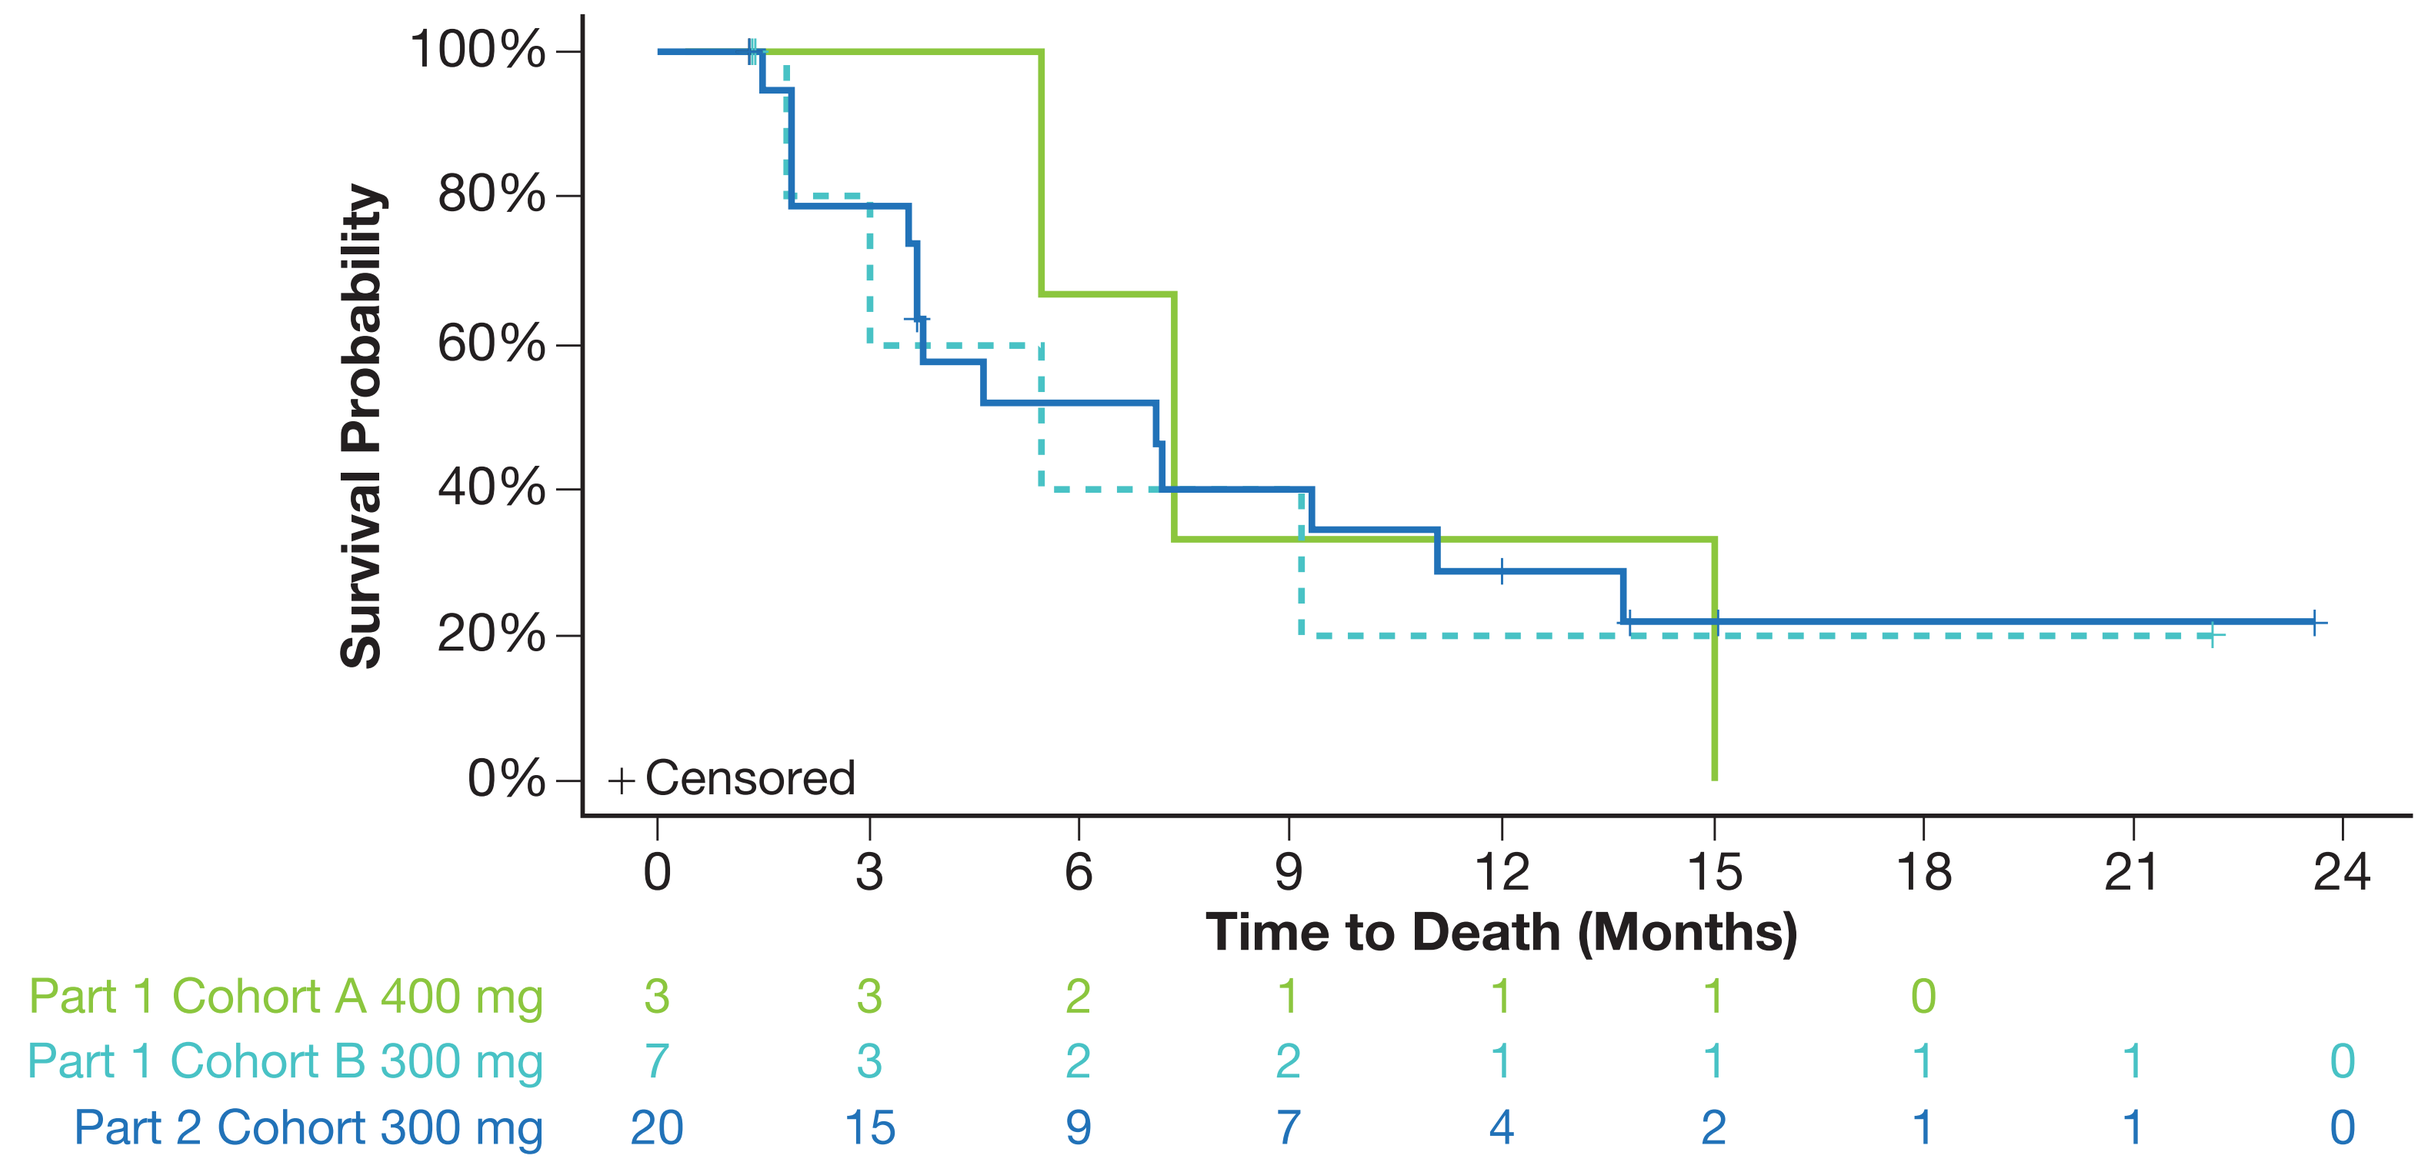
Survival Probability
100%
80%
60%
40%
20%
0%
+ Censored
0 3 6 9 12 15 18 21 24
Time to Death (Months)
Part 1 Cohort A 400 mg 3 3 2 1 1 1 0
Part 1 Cohort B 300 mg 7 3 2 2 1 1 1 1 0
Part 2 Cohort 300 mg 20 15 9 7 4 2 1 1 0

	Patients	Events	Censored	Median Survival	95% CI
Part 1 Cohort A 400 mg	3	3	0	7.39	(5.49, 15.05)
Part 1 Cohort B 300 mg	7	4	3	5.49	(1.84, NA)
Part 2 Cohort 300 mg	20	4	6	7.13	(3.58, 11.07)

Median survival is based on the Kaplan-Meier estimates.

Figure 4: Kaplan-Meier Survival Curve for Progression-Free Survival (Full Analysis Set: N=30)

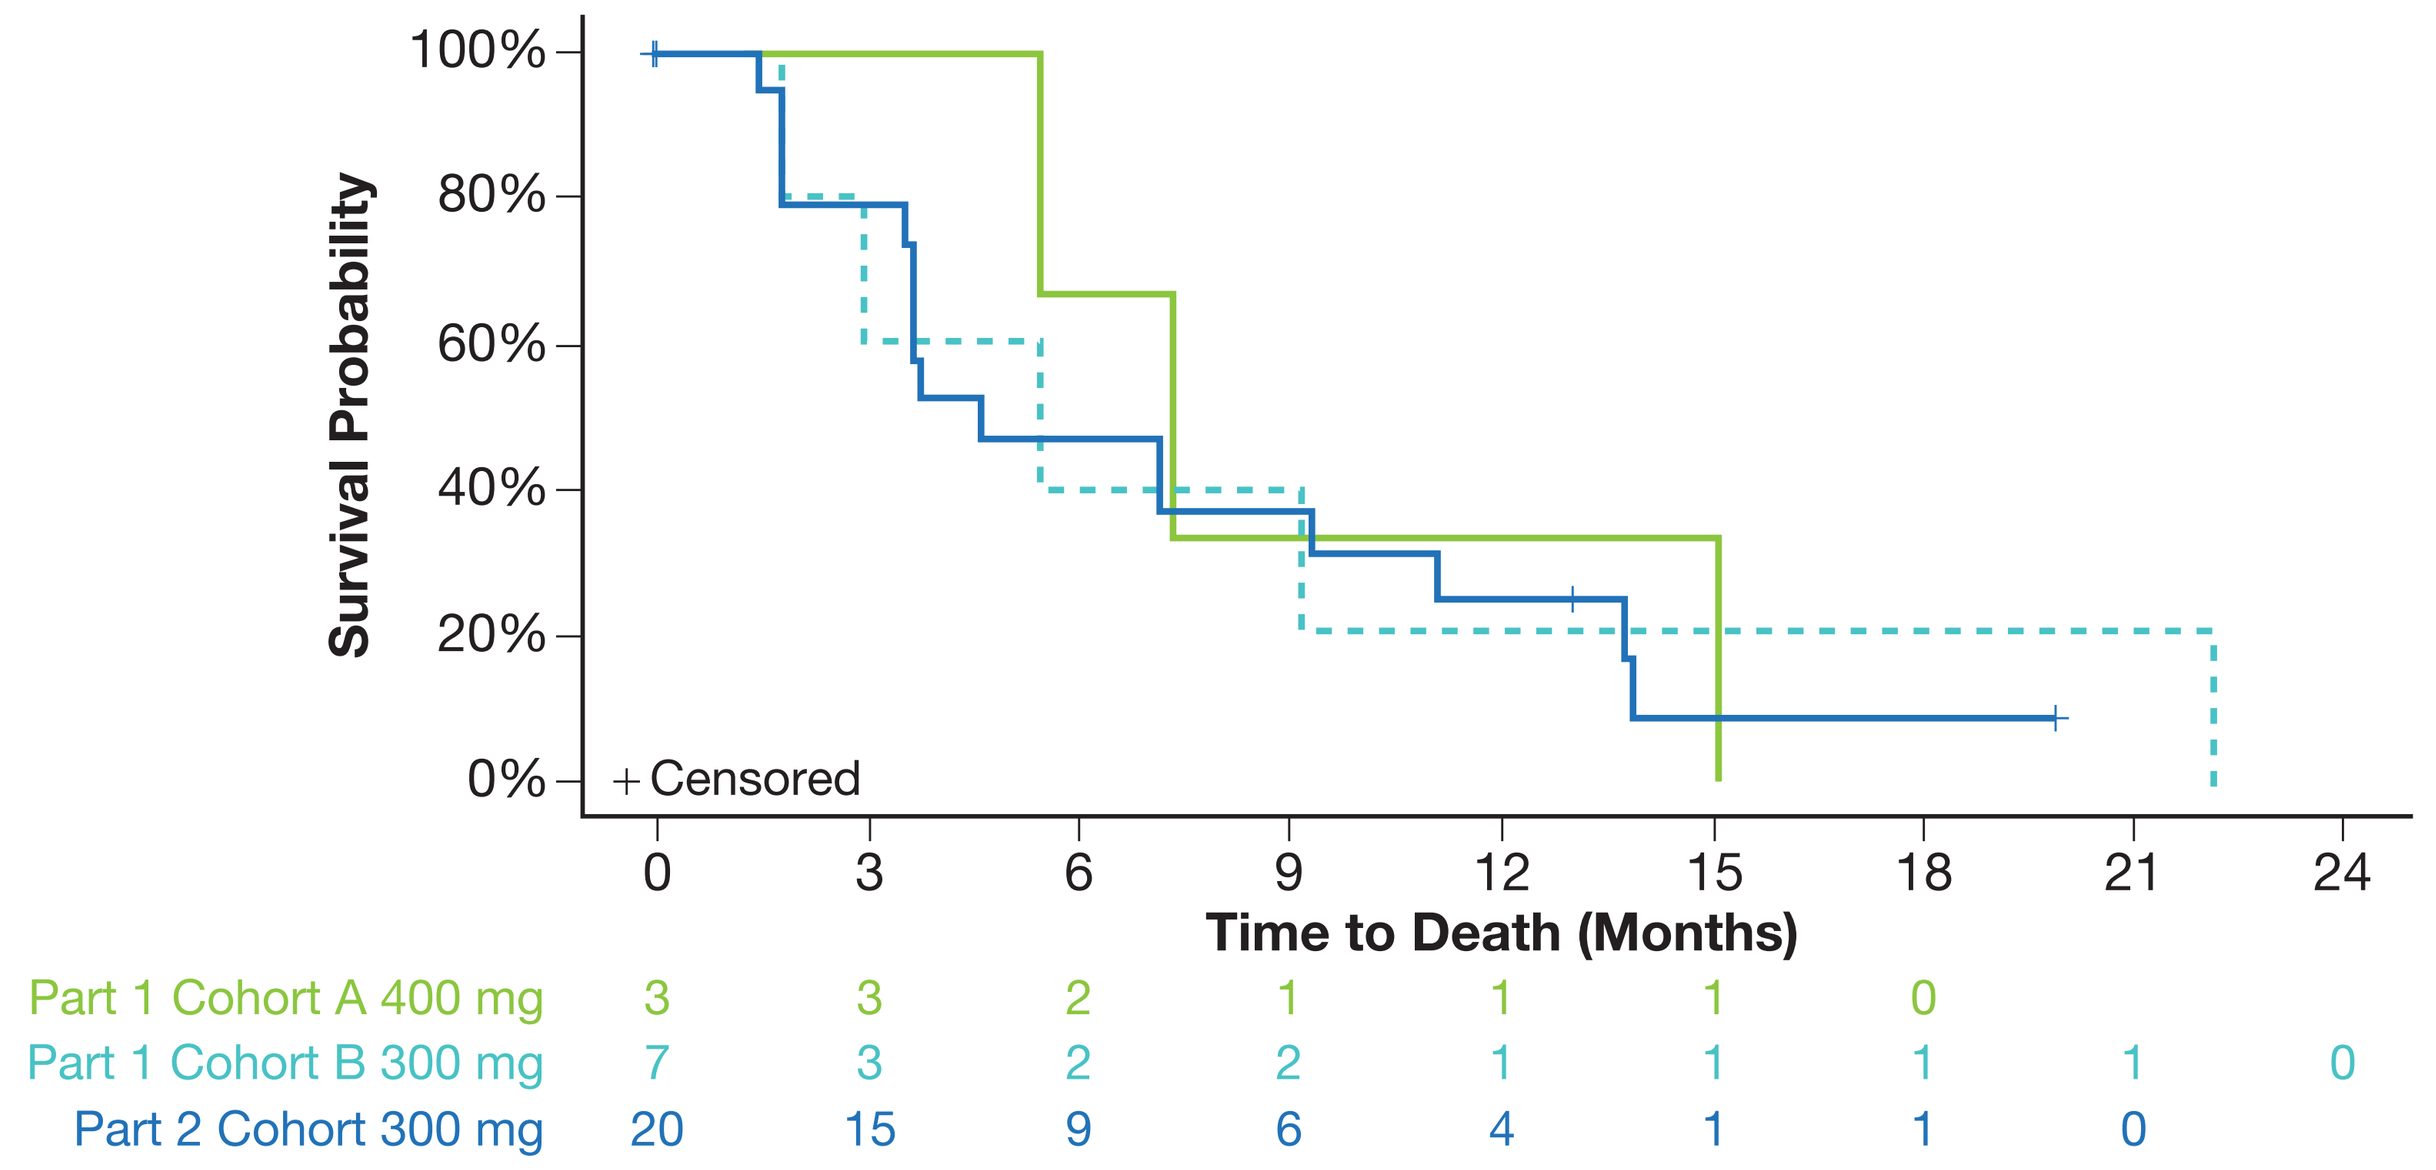
Survival Probability
100%
80%
60%
40%
20%
0%
+ Censored
0 3 6 9 12 15 18 21 24
Time to Death (Months)
Part 1 Cohort A 400 mg 3 3 2 1 1 1 0
Part 1 Cohort B 300 mg 7 3 2 2 1 1 1 1 0
Part 2 Cohort 300 mg 20 15 9 6 4 1 1 0

	Patients	Events	Censored	Median Survival	95% CI
Part 1 Cohort A 400 mg	3	3	0	7.39	(5.49, 15.05)
Part 1 Cohort B 300 mg	7	5	2	5.49	(1.84, 22.11)
Part 2 Cohort 300 mg	20	16	4	4.67	(3.58, 11.07)

Median survival is based on the Kaplan-Meier estimates.

CONCLUSIONS

- Rivoceranib 300 mg PO QD combined with nivolumab 240 mg IV Q2W demonstrated a manageable safety profile and promising antitumor efficacy in patients with advanced or metastatic solid tumors.
- All patients showed clinical benefit from the combination treatment with rivoceranib and nivolumab as measured by reduction in tumor burden and duration over time.
- Further studies are needed to confirm the safety and efficacy of this combination therapy.

REFERENCES

1. Apolo AB, et al. *J Clin Oncol*. 2014;32(4_suppl):307.
2. McDermott DF, et al. *J Clin Oncol*. 2017;35(6_suppl):431.
3. Rini BI, et al. *J Clin Oncol*. 2017;35(15_suppl):TPS4597.
4. Choueiri TK, et al. *J Clin Oncol*. 2017;35(15_suppl):4504.

AUTHOR INFO

DOI for presenting author, Neal Chawla: no declarations of interest.

For questions or additional information, please contact medicalinformation@elevartherapeutics.com.

ACKNOWLEDGMENTS

- This study was funded by Elevar Therapeutics. The Phillips Group Oncology Communications Inc. provided professional assistance with poster preparation. Financial support for writing and editorial services was provided by Elevar Therapeutics.