Analysis of Antidrug Antibodies (ADA) to Camrelizumab in CARES-310: The Pivotal Phase 3 Study of Camrelizumab + Rivoceranib in Unresectable Hepatocellular Carcinoma (uHCC)

Ahmed Omar Kaseb,[1] James Bugni,[2] Mansi Karkhanis,[2] Xianzhang Meng,[2] Tracey Wei,[2] Mingyan Zhou,[3] Kai Shen,[3] ChunMei Yang,[3] MeiJing Yu,[3] Seong Jang,[2] Kristin Ryan,[2] Laura Alexander[2]

[1]Department of Gastrointestinal Medical Oncology, MD Anderson Cancer Center, Houston, Texas, USA; [2]Elevar Therapeutics, Fort Lee, NJ, USA; [3]Hengrui USA, Princeton, NJ, USA

Presented at ESMO 2024

FPN# 985P

Copies of this poster obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- High antidrug antibody (ADA) levels can reduce systemic drug exposure of biologic therapies, whereas ADAs that are neutralizing (NAb) can attenuate pharmacodynamic effects.
- Study CARES-310 (NCT03764293) compared the combination of the anti-programmed death-1 (PD-1) antibody camrelizumab plus the vascular endothelial growth factor receptor (VEGFR)-targeted tyrosine kinase inhibitor (TKI) rivoceranib with sorafenib for the treatment of unresectable HCC.[1]
- At the protocol-specified final analysis (median follow-up: 22.1 months), camrelizumab plus rivoceranib continued to show significantly improved overall survival (OS) and progression-free survival (PFS) compared with sorafenib (median OS 23.8 months vs 15.2 months; HR 0.64 [95% CI, 0.52-0.79; P<0.0001]; mPFS 5.6 months vs 3.7 months; HR 0.54 [95% CI, 0.44-0.67; P<0.0001]).[2]
- The most common (≥10%) grade ≥3 treatment-related adverse events at the protocol-specified final analysis for camrelizumab plus rivoceranib were hypertension (38.2%), increased aspartate aminotransferase (AST) (17.3%), increased alanine aminotransferase (ALT) (14.0%), palmar-plantar erythrodysesthesia (PPE) syndrome (12.1%), and platelet count decreased (11.8%) vs PPE syndrome (15.6%) and hypertension (14.9%) for sorafenib.[2]
- Herein, we report levels of ADAs and NAbs to camrelizumab, and their implications for pharmacokinetics (PK), efficacy, and safety in Study CARES-310.

METHODS

CARES-310 Study Design and Endpoints[1]

Key Eligibility Criteria

- BCLC Stage B (unsuitable for radical surgery and/or locoregional treatment) or C
- No prior systemic therapy
- ECOG PS 0 or 1
- Child-Pugh A
- At least one measurable lesion per RECIST v1.1

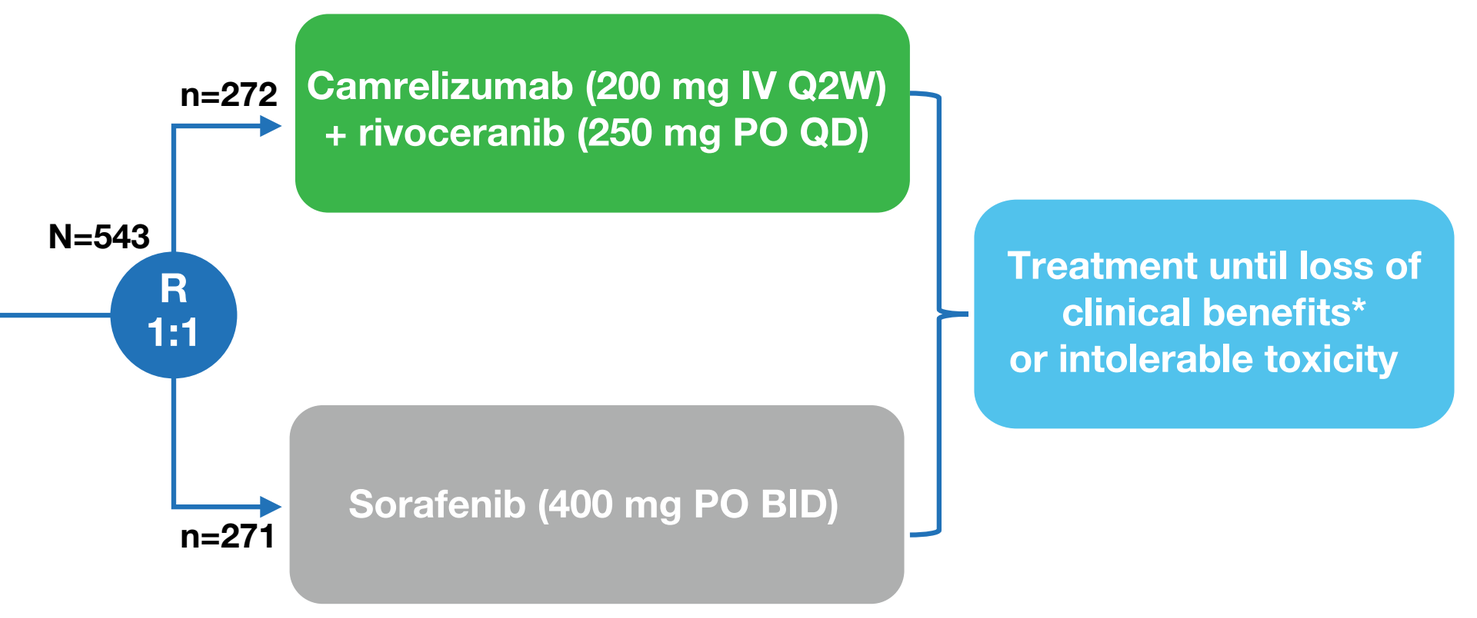
N=543
R
1:1
n=272
Camrelizumab (200 mg IV Q2W) + rivoceranib (250 mg PO QD)
n=271
Sorafenib (400 mg PO BID)
Treatment until loss of clinical benefits* or intolerable toxicity

Stratification Factors

- MVI and/or EHS (yes vs no)
- Geographical region (Asia vs non-Asia)
- Baseline serum AFP (<400 vs ≥ 400 ng/mL)

Primary Endpoints

- PFS‡
- OS

Key Secondary Endpoint

- ORR‡

*Treatment beyond progression allowed if there was evidence of clinical benefits per investigator. ‡By BIRC per RECIST v1.1.

AFP, alpha-fetoprotein; BCLC, Barcelona Clinic Liver Cancer; BID, twice daily; BIRC, blinded independent review committee; ECOG PS, Eastern Cooperative Oncology Group performance status; EHS, extrahepatic spread; IV, intravenous; MVI, macrovascular invasion; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PO, by mouth; Q2W, every 2 weeks; QD, once daily; RECIST v1.1, Response Evaluation Criteria for Solid Tumors version 1.1.

Antibody Analysis

- Electrochemiluminescent immunoassays were used to detect ADAs and NAbs to camrelizumab in serum samples at sensitivities of 1.7-10.9 ng/mL and 210 ng/mL, respectively.
 - The assay can detect 10 ng/mL positive control ADA in the presence of 200 µg/mL of camrelizumab. Nab antibody level of 200 ng/mL was detectable in the presence of 50.0 µg/mL of camrelizumab; Nab antibody level of 500 ng/mL was detectable in the presence of 250 µg/mL of camrelizumab.
- Efficacy and safety were assessed by ADA+ and ADA- status.
- The baseline and steady state clearance (CL) of camrelizumab were predicted by a previously developed population PK model and were compared in ADA+, with positive patients stratified by median titers.
- Onset and Duration and Pharmacokinetics results are from the CARES-310 interim analysis, Mar 21, 2023 (ADA+, n=54; ADA-, n=199). All other results are from the CARES-310 final analysis, Feb 2, 2024 (ADA+, n=55, ADA-, n=199).

REFERENCES

1. Qin S, et al. *Lancet*. 2023;402(10408):1133-1146.
2. Vogel A, et al. *J Clin Oncol*. 2024;42(suppl_16):abs 4110.

ACKNOWLEDGMENTS

- This study was funded by Jiangsu Hengrui Pharmaceuticals Co., Ltd. and Elevar Therapeutics. The Phillips Group Oncology Communications Inc. provided professional assistance with poster preparation. Financial support for writing and editorial services was provided by Elevar Therapeutics.

AUTHOR INFO

DOI for presenting author, Ahmed Kaseb: Research funding: Elevar, BMS, Merck, Roche, AdaptImmune, Exelixis, AstraZeneca, and Tvardi; Honoraria: Elevar, BMS, Merck, Roche, Eisai, Exelixis, and AstraZeneca

For questions or additional information, please contact medicalinformation@elevartherapeutics.com.

RESULTS

Antibody Analysis Results

- The ADA and Nabs were measured from 254 (out of 272) patients (93.3%) who were treated with camrelizumab + rivoceranib.
- The incidence of ADAs and NAbs were 21.7% (n=55) and 22.0% (n=56), respectively (**Table 1**).
- Median ADA onset was 8.1 weeks (interquartile range 4.4-12.0 weeks) with a median duration of 4.6 weeks (95% CI, 3.9-12.1) (**Figure 1** and **Figure 2**).
- Median NAb onset was 8.3 weeks (IQR 4.4-12.1 weeks) with a median duration of 4.1 weeks (95% CI, 0.1-8.0).

Table 1: Summary of Patients by ADA and NAb Categories

Category, n (%)	Camrelizumab + Rivoceranib (N=254)
ADA-positive[a]	**55 (21.7)**
Treatment-induced positive[b]	53 (20.9)
Transient ADA positive[c]	9 (3.5)
Persistent ADA positive[d]	38 (15.0)
Other ADA positive	6 (2.4)
Treatment-enhanced positive[e]	2 (0.8)
ADA-negative	**199 (78.3)**
Non-treatment emergent ADA positive[f]	17 (6.7)
ADA never positive[g]	182 (71.6)
NAb-positive	56 (22.0)
NAb-negative	196 (77.2)
NAb-inconclusive	2 (0.8)

[a]ADA-positive: the number of patients who were treatment-induced positive or treatment-enhanced positive; [b]Treatment-induced ADA positive: patients who had baseline negative and post-baseline positive samples; [c]Transient ADA positive: negative at baseline and had only one ADA positive sample at post-baseline before the last sample or second-to-last sample; [d]Persistent ADA positive: Negative at baseline, there are at least 2 positive samples post-baseline, or negative at baseline, the last sample of post-baseline was positive, or the last sample of post-baseline was negative, and the second last sample was positive before an ADA negative last sample; [e]Treatment-enhanced ADA positive: patients who had both baseline and post-baseline positive samples, and the titer of the post-baseline samples was equal or greater than 4-fold of the baseline titer; [f]Non-treatment emergent ADA positive: a) patients who had baseline positive samples and post-baseline negative samples; or b) patients who had both baseline and post-baseline positive samples, and the titer of the post-baseline samples was less than 4-fold of the baseline titer; [g]ADA never positive: patients who did not have any ADA positive samples.

Onset and Duration of Antidrug Antibodies

Figure 1: Time of First ADA Detection

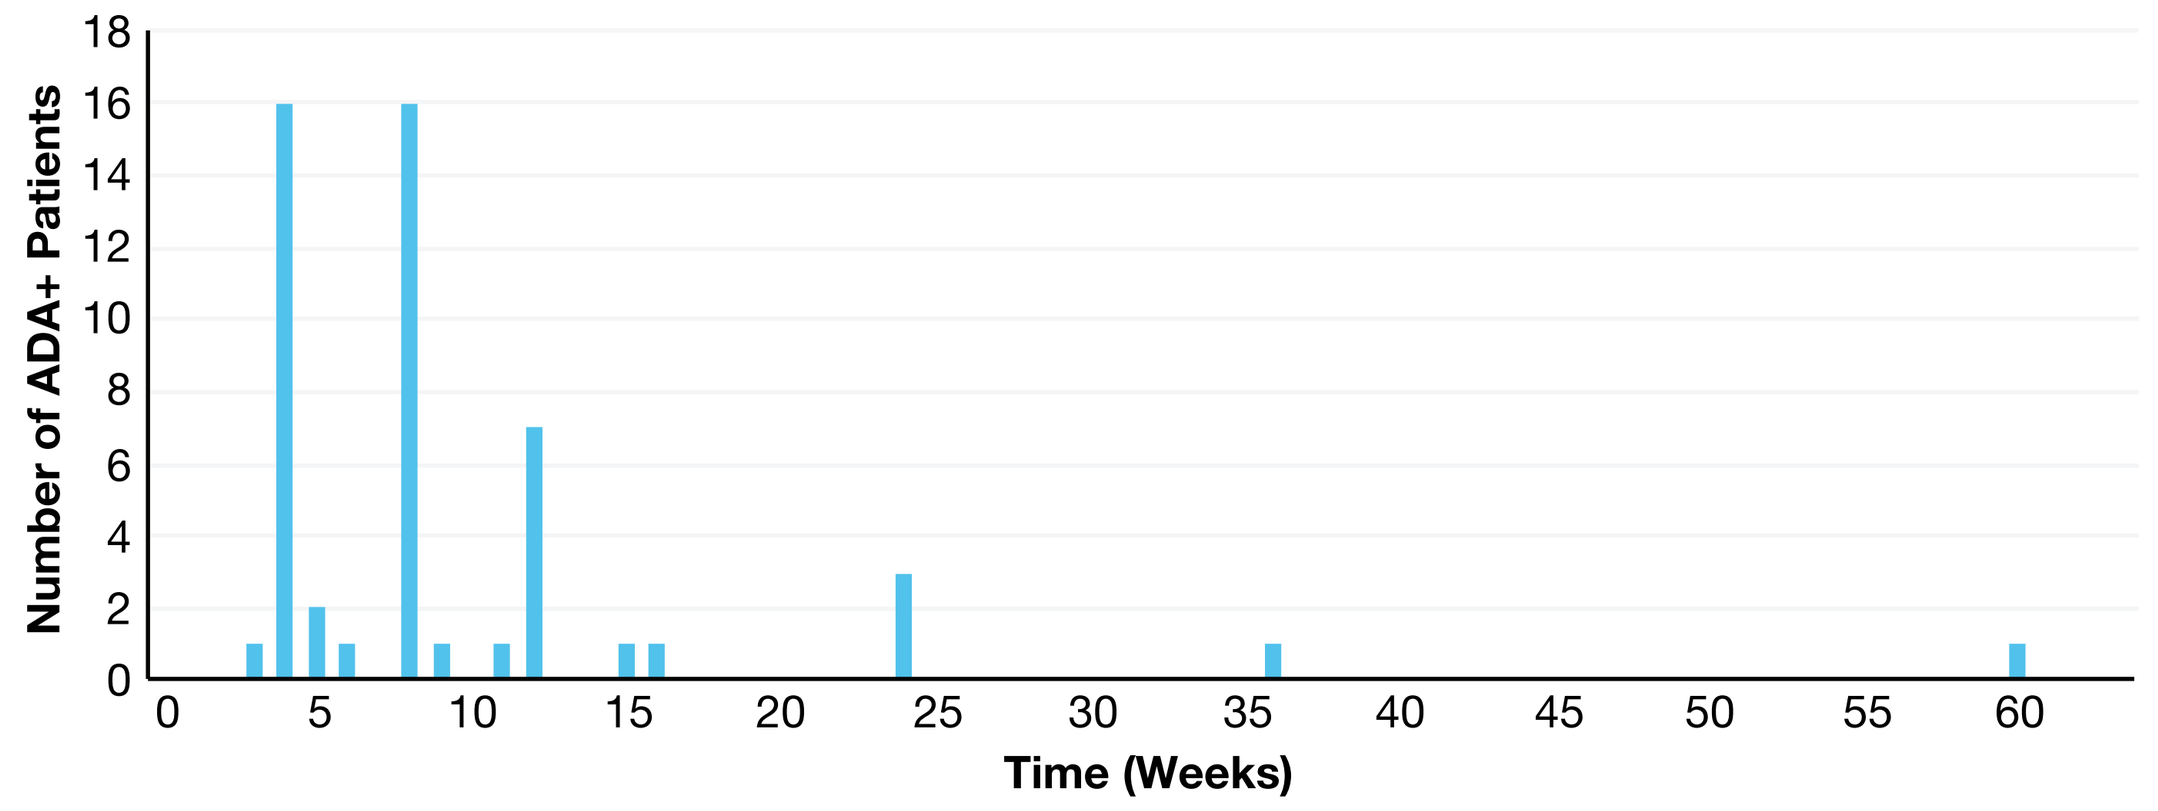
Number of ADA+ Patients
Time (Weeks)

Figure 2: Duration of ADA Detection Interval

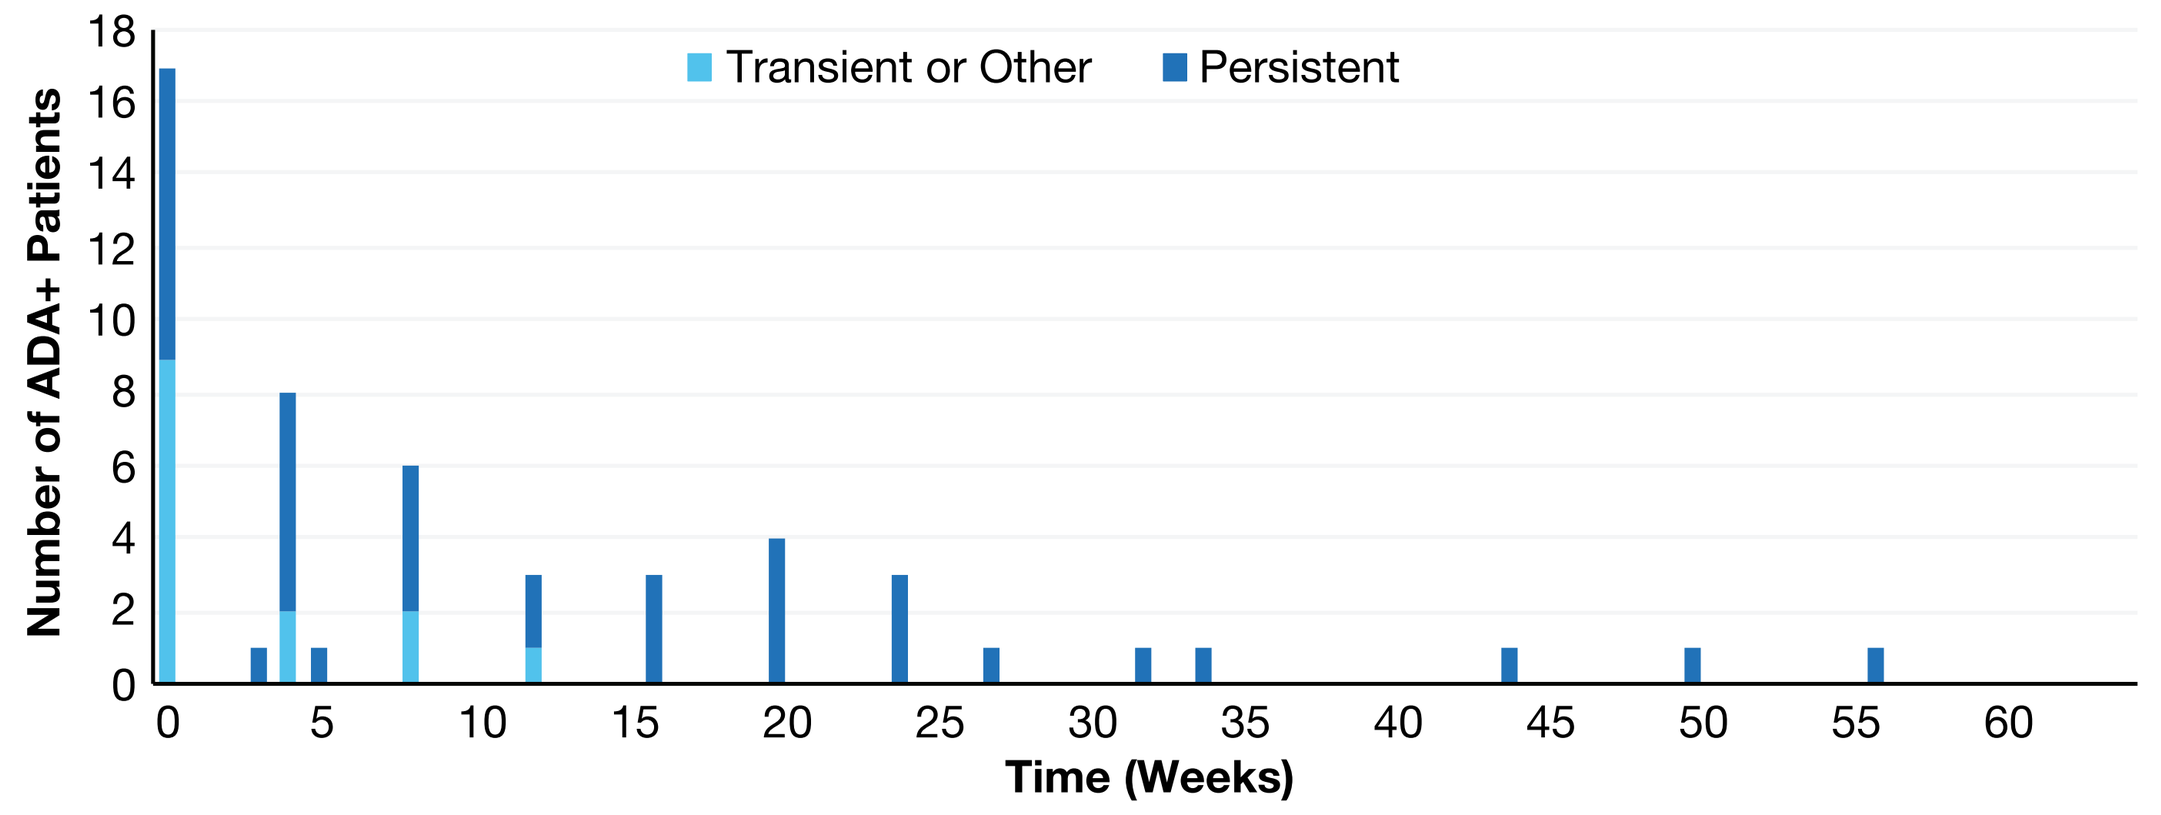
Transient or Other
Persistent
Number of ADA+ Patients
Time (Weeks)

Analysis of Hepatocellular Carcinoma History and Disease Characteristics

- Differences were observed in baseline characteristics between ADA+ and ADA- groups (**Table 2**).
- Imbalances expected to be associated with worse prognosis in ADA+ compared to ADA- subgroup:
 - 16.2% higher proportion of macrovascular invasion (MVI)
 - 10.9% higher proportion of MVI and/or extrahepatic metastases (EHM)
 - 26 mm greater median sum of target lesion diameter
 - 4.4% higher proportion of Child-Pugh A6
- Imbalances expected to be associated with worse prognosis for ADA- compared to ADA+ subgroup:
 - 5.3% higher proportion of baseline alpha fetoprotein (AFP) ≥400 ng/mL in ADA-positive subgroup

Table 2. Baseline Hepatocellular Carcinoma Disease Characteristics

	ADA-positive (n = 55)	ADA-negative (n = 199)
Root cause of HCC, n (%)		
HBV	36 (65.5)	164 (82.4)
HCV	4 (7.3)	13 (6.5)
Non-viral	15 (27.3)	22 (11.1)
BCLC stage at screening, n (%)		
B (middle stage)	6 (10.9)	30 (15.1)
C (advanced stage)	49 (89.1)	169 (84.9)
MVI, n (%)		
Yes	15 (27.3)	22 (11.1)
No	40 (72.7)	177 (88.9)
EHM, n (%)		
Yes	36 (65.5)	126 (63.3)
No	19 (34.5)	73 (36.7)
EHM and/or MVI, n (%)		
Yes	45 (81.8)	141 (70.9)
No	10 (18.2)	58 (29.1)
Child-Pugh score, n (%)		
A5	47 (85.5)	177 (88.9)
A6	8 (14.5)	22 (11.1)
AFP category at screening, n (%)		
<400 ng/mL	38 (69.1)	127 (63.8)
≥400 ng/ml	17 (30.9)	72 (36.2)
Baseline sum of target lesion diameter (mm) per RECIST v1.1 per BIRC[a]		
Mean (SD)	92.0 (64.32)	69.9 (51.74)
Median	78.0	52.0
Min, Max	33.0, 128.0	33.5, 90.5
Time since initial cancer diagnosis (days)		
Mean (SD)	507.5 (831.85)	647.9 (943.17)
Median	118.0	322.0
Min, Max	4, 4029	2, 6141
Time since advanced/metastatic disease diagnosis (days)[b]		
Mean (SD)	173.7 (388.66)	224.5 (471.84)
Median	41.0	47.0
Min, Max	6, 1801	5, 3080

[a]ADA+ (n=49), ADA- (n=196); [b]ADA+ (n=53), ADA- (n=197).

AFP, alpha fetoprotein; BCLC, Barcelona Clinic Liver Cancer; BIRC, blinded independent review committee; EHM, extrahepatic metastases; HBV, Hepatitis B virus; HCV, Hepatitis C Virus; MVI, macrovascular invasion; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1.

Safety

- Treatment-emergent adverse events (TEAEs) leading to treatment discontinuation (34.5% vs 29.1%) and TEAEs leading to dose reduction or interruption (90.9% vs 84.4%) were higher in the ADA+ patients compared with the ADA- patients (**Table 3**).
- The incidence of treatment-related serious adverse events (SAEs) was numerically lower in ADA+ patients than that in ADA- patients (18.2% vs 25.6%)
- Minor differences in AE rates were possibly due to imbalances in baseline disease characteristics between ADA+ and ADA- subgroups.

Table 3: Overview of Adverse Events by ADA Status

Adverse event category, n (%)	ADA-positive (n=55)	ADA-negative (n=199)
TEAEs	55 (100)	198 (99.5)
TEAEs related to any study treatment	54 (98.2)	197 (99.0)
Grade ≥3 AEs	48 (87.3)	176 (88.4)
Grade ≥3 AEs related to camrelizumab or rivoceranib	43 (78.2)	166 (83.4)
Serious AEs	21 (38.2)	86 (43.2)
Serious AEs related to study treatment	10 (18.2)	51 (25.6)
AEs leading to discontinuation of any study treatment	19 (34.5)	58 (29.1)
AEs leading to discontinuation of camrelizumab	13 (23.6)	40 (20.1)
AEs leading to dose reduction/interruption of any study treatment	50 (90.9)	168 (84.4)

AE, adverse event; TEAE, treatment-emergent adverse event.

Table 4. Overview of Camrelizumab-Related Adverse Events by ADA Status

Adverse event category, n (%)	ADA-positive (n=55)	ADA-negative (n=199)
Camrelizumab-related AEs	47 (85.5)	164 (82.4)
Grade ≥3 camrelizumab-related AEs	12 (21.8)	34 (17.1)
Grade 5 camrelizumab-related AEs	0	0
Serious camrelizumab-related AEs	4 (7.3)	16 (8.0)
Camrelizumab-related AEs leading to discontinuation of any study treatment	6 (10.9)	9 (4.5)
Camrelizumab-related AEs leading to dose reduction/interruption of any study treatment	14 (25.5)	49 (24.6)

AE, adverse event.

Efficacy

- No differences were observed between ADA+ and ADA- subgroups in OS (mOS: 25.1 mo vs 95% 25.4 mo, respectively) (**Figure 3**) and PFS (mPFS: 5.6 mo vs 6.1 mo, respectively, **Figure 4**).
- The objective response rate (ORR) in the ADA+ subgroup was 20.0% (11/55) compared to 31.2% (62/199) in the ADA- subgroup. Imbalances in baseline demographics and disease characteristics between ADA subgroups confounded a conclusion on the potential impact of ADAs on ORR.

Figure 3: Kaplan-Meier Curves of OS by ADA Status

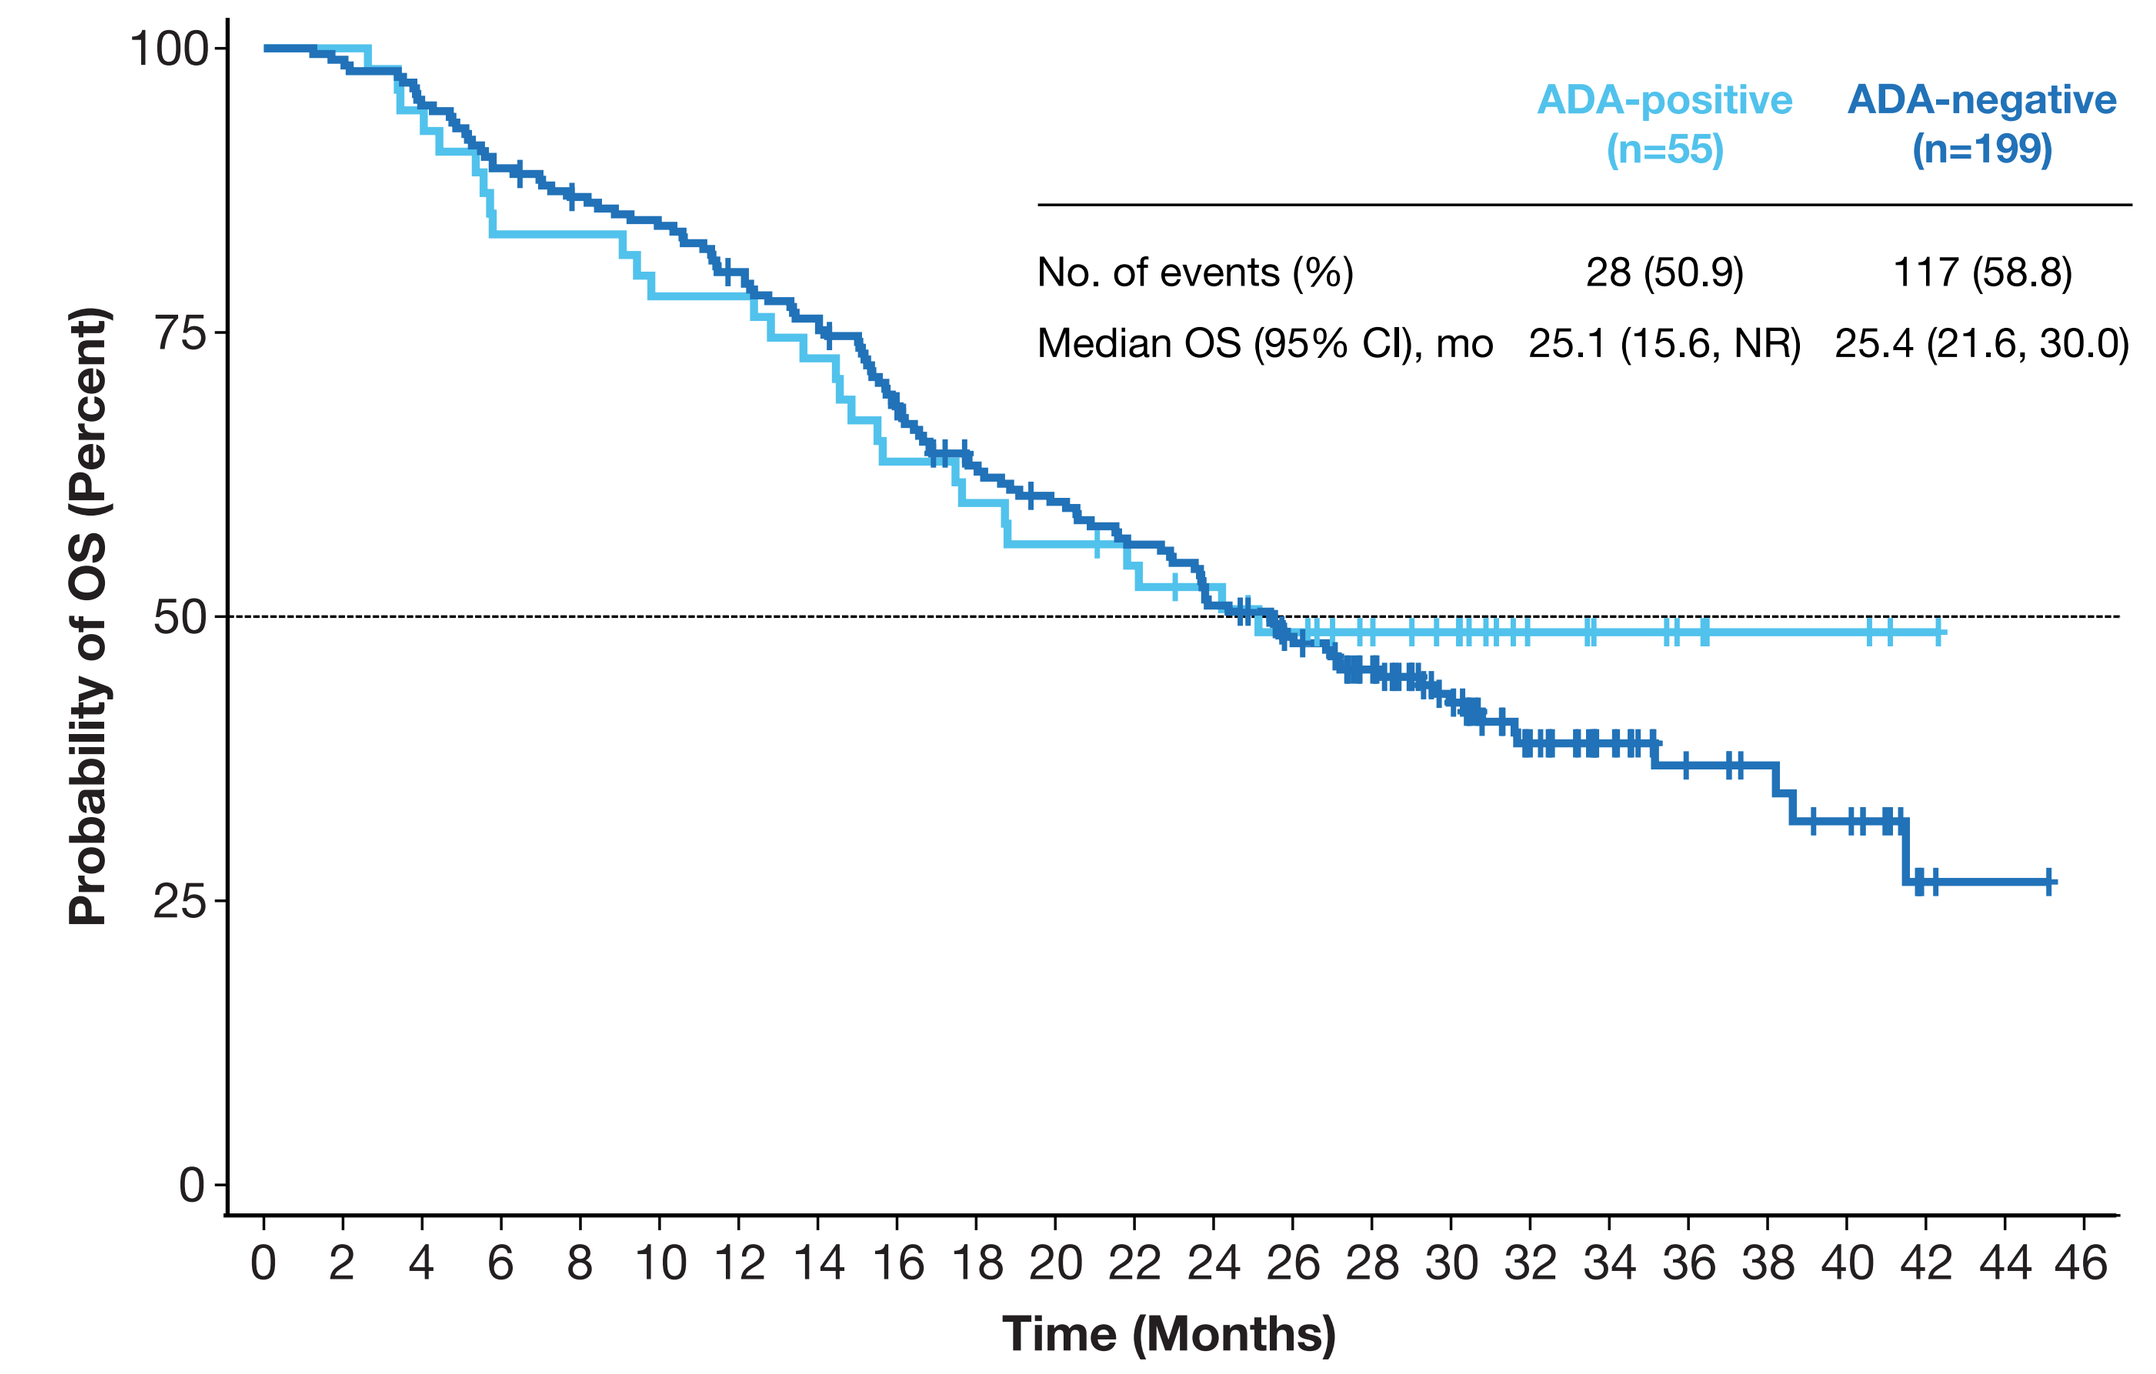
ADA-positive (n=55)
ADA-negative (n=199)
No. of events (%) 28 (50.9) 117 (58.8)
Median OS (95% CI), mo 25.1 (15.6, NR) 25.4 (21.6, 30.0)
Probability of OS (Percent)
Time (Months)

Number of Patients at Risk

ADA-positive 55 55 52 46 46 43 43 40 35 33 31 29 27 24 20 17 9 7 5 3 3 1 0

ADA-negative 199 197 189 178 171 166 157 149 133 119 112 105 95 85 73 55 39 27 18 15 12 2 1 0

NR, not reached.

Figure 4: Kaplan-Meier Curves of PFS by ADA Status

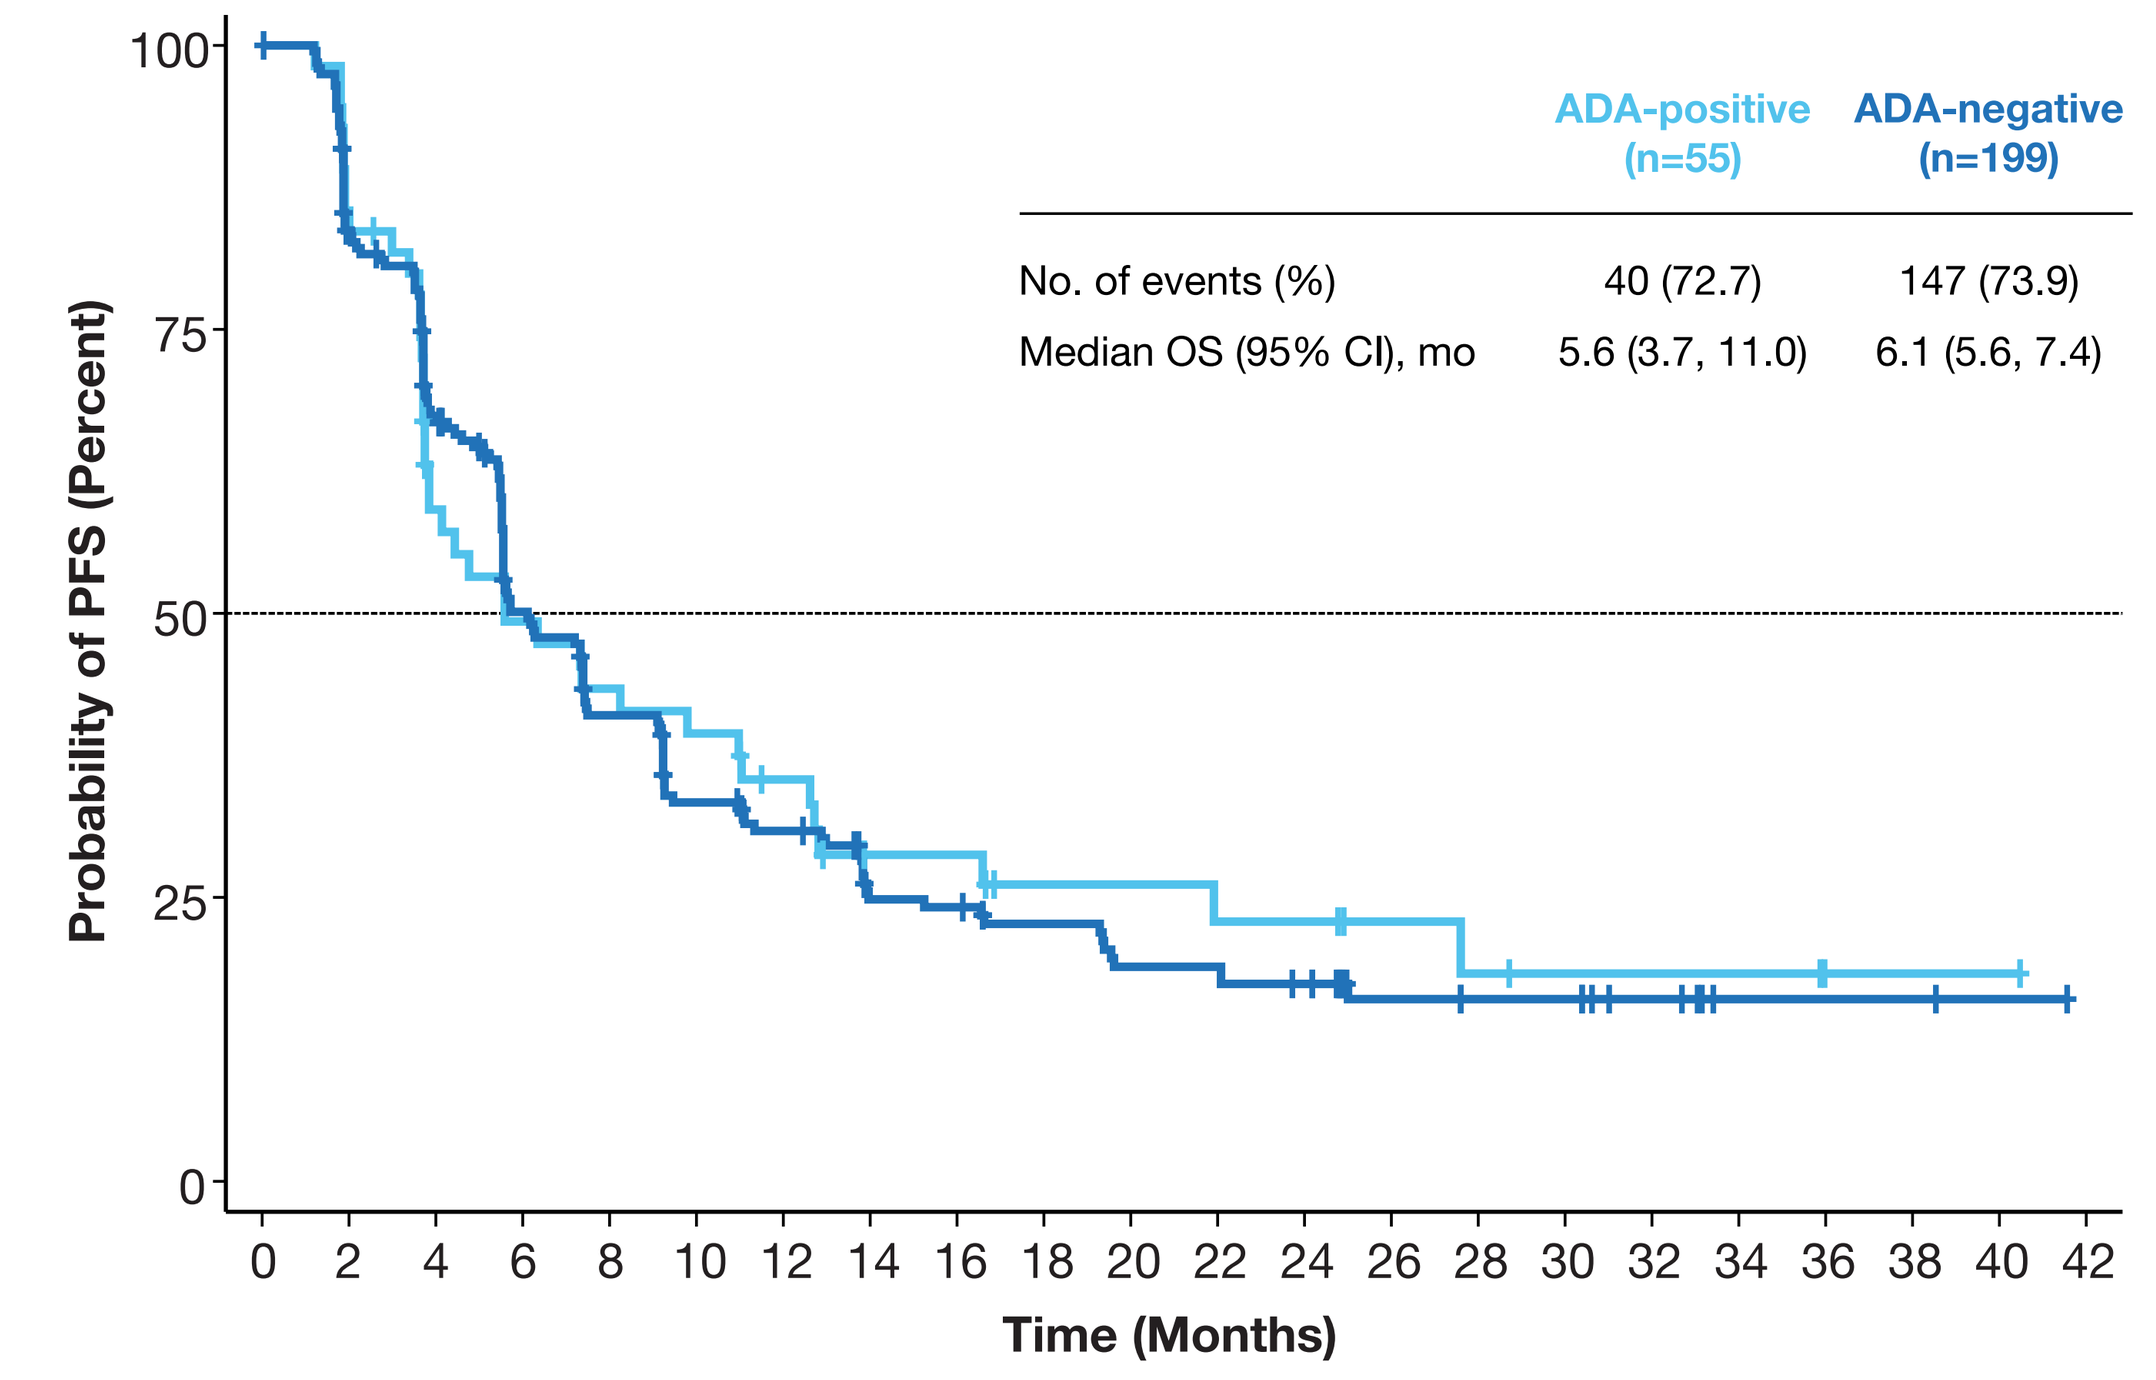
ADA-positive (n=55)
ADA-negative (n=199)
No. of events (%) 40 (72.7) 147 (73.9)
Median OS (95% CI), mo 5.6 (3.7, 11.0) 6.1 (5.6, 7.4)
Probability of PFS (Percent)
Time (Months)

Number of Patients at Risk

ADA-positive 55 47 30 25 22 20 16 11 11 8 8 7 7 5 4 3 3 3 1 1 1 0

ADA-negative 199 161 125 89 71 55 49 36 35 30 25 25 22 12 10 10 6 2 2 2 1 0

Pharmacokinetics

- The steady-state clearances (L/day) of camrelizumab in the ADA+ (mean 0.267, range 0.155 to 0.551) and NAb+ (mean 0.266, range 0.153 to 0.551) subgroups were numerically higher than, but mostly overlapped, the ADA- (mean 0.202, range 0.103 to 0.452) and NAb- (mean 0.202, range 0.103 to 0.452) subgroups (**Figure 5A**).
- A similar trend was observed for baseline clearance where neither ADA nor NAb have developed; the baseline clearances for the ADA+ (mean, 0.301, range 0.161 to 0.572) and the NAb+ (mean 0.300, range 0.161 to 0.572) subgroups were numerically greater than the ADA- (mean 0.243, 0.133 to 0.535) and NAb- (mean 0.243, 0.133 to 0.535) subgroups, respectively (**Figure 5B**).
- ADA titer had a minor effect on clearance: steady-state clearance (L/day) in the > Median peak titer group (mean 0.286, range 0.159 to 0.551) was numerically higher than, but mostly overlapped, clearance in the ≤ Median peak titer group (mean 0.249, range 0.155 to 0.460) (**Figure 6A**)
- Again, a similar trend was observed for baseline clearance where ADA had not developed: baseline clearance in the ≥ Median titer group (mean 0.327, range 0.183 to 0.572) was numerically higher than in the < Median titer group (mean 0.275, range 0.161 to 0.478) (**Figure 6B**)

Figure 5: Clearance of Camrelizumab by ADA and NAb Status

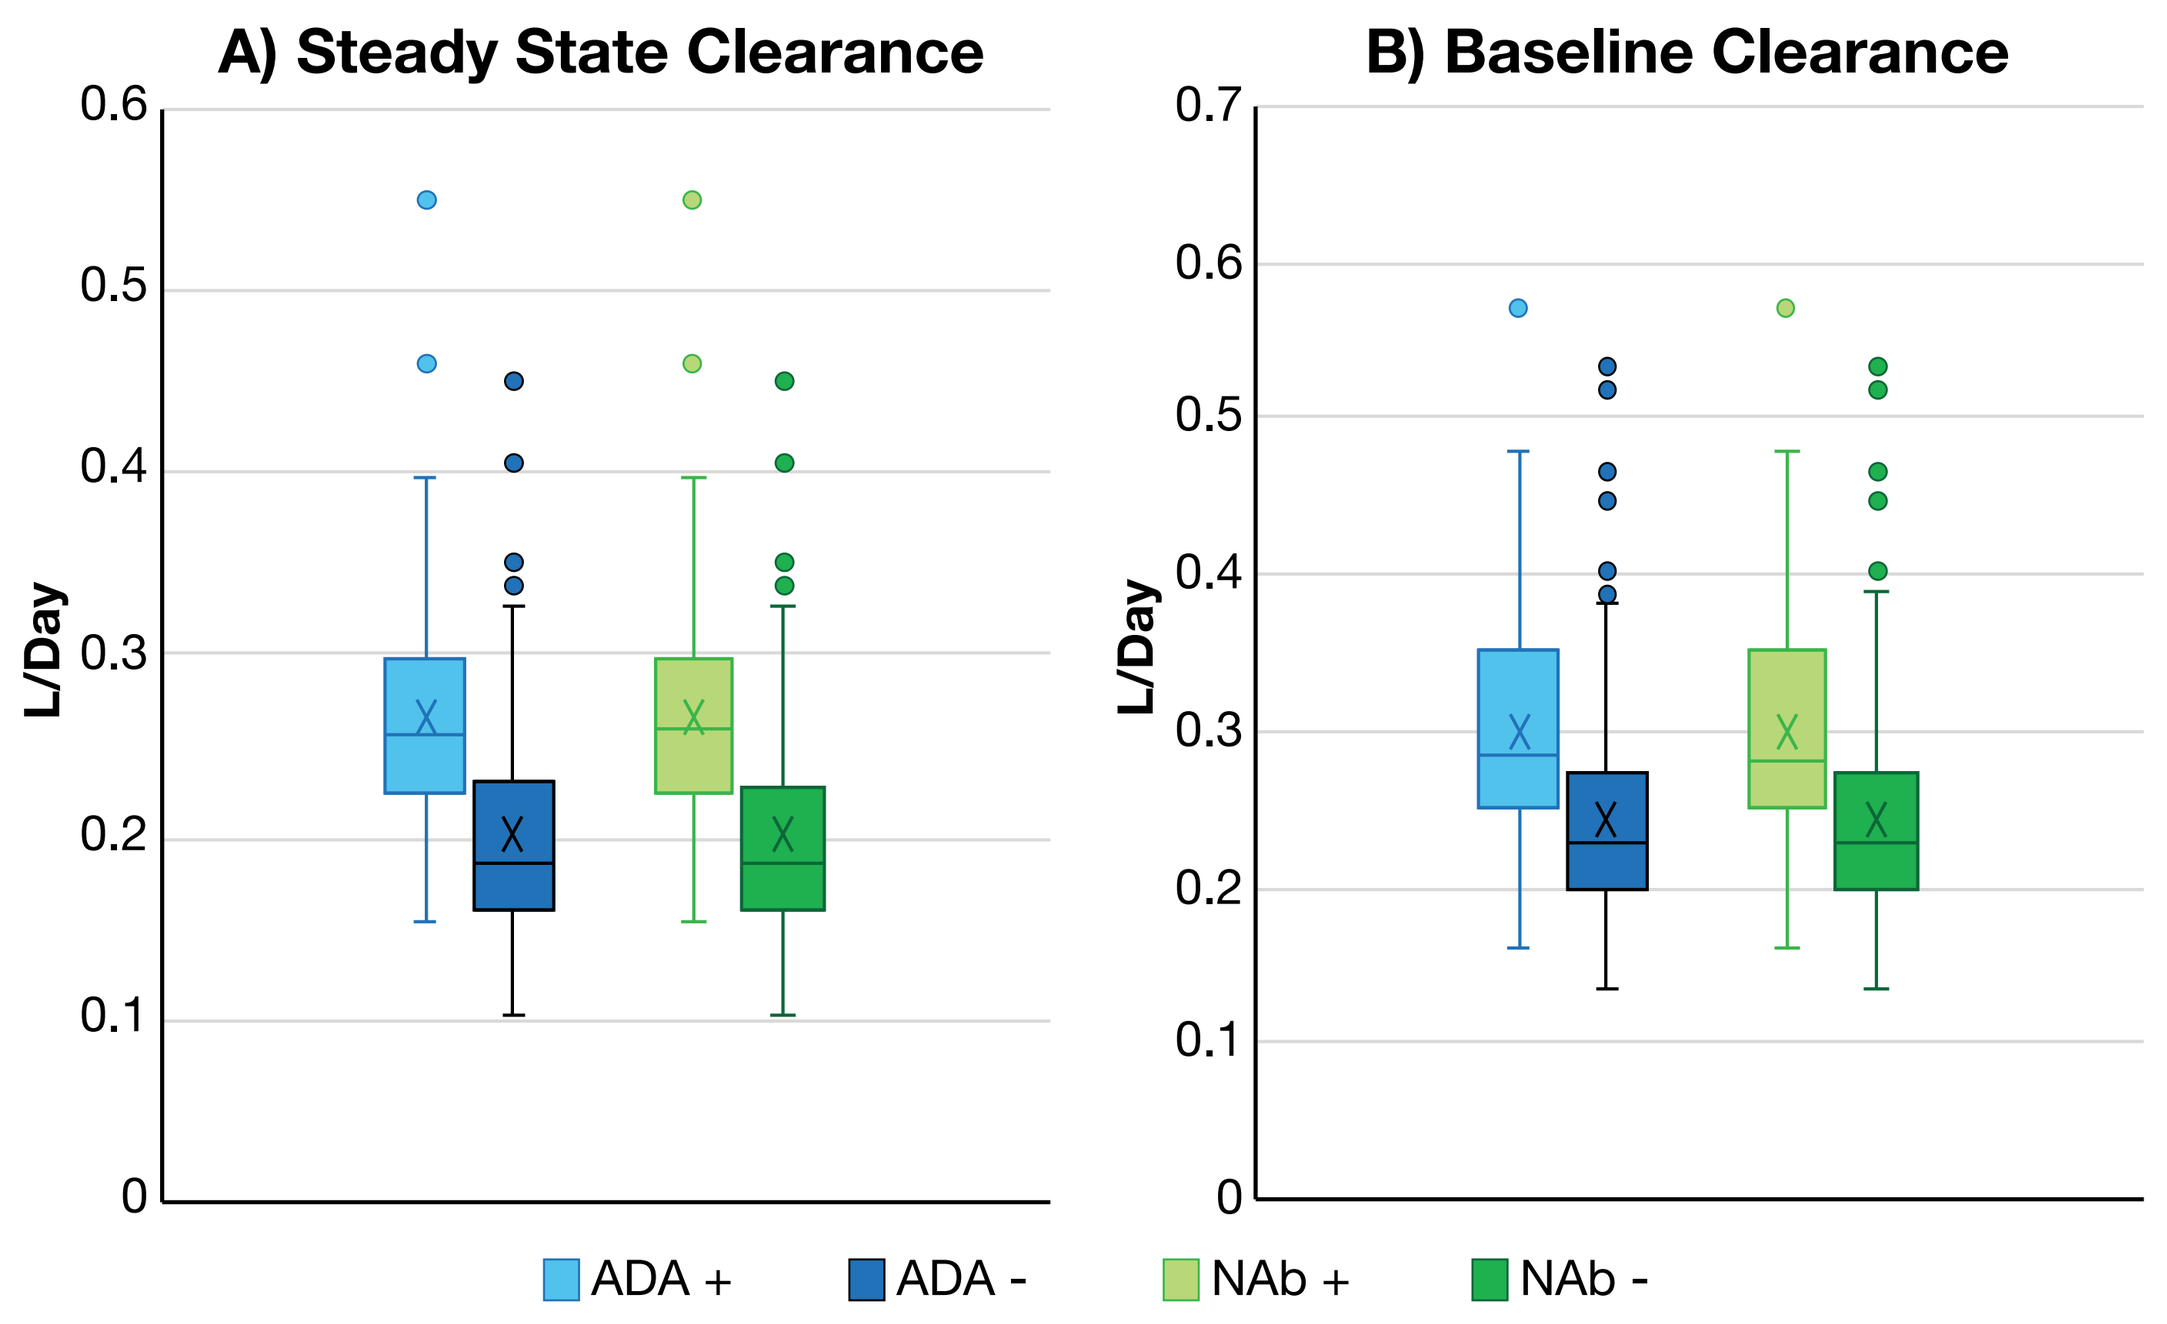
A) Steady State Clearance
B) Baseline Clearance
L/Day
ADA +
ADA -
NAb +
NAb -

Figure 6: Clearance of Camrelizumab by ADA Separated by Median Peak Titers

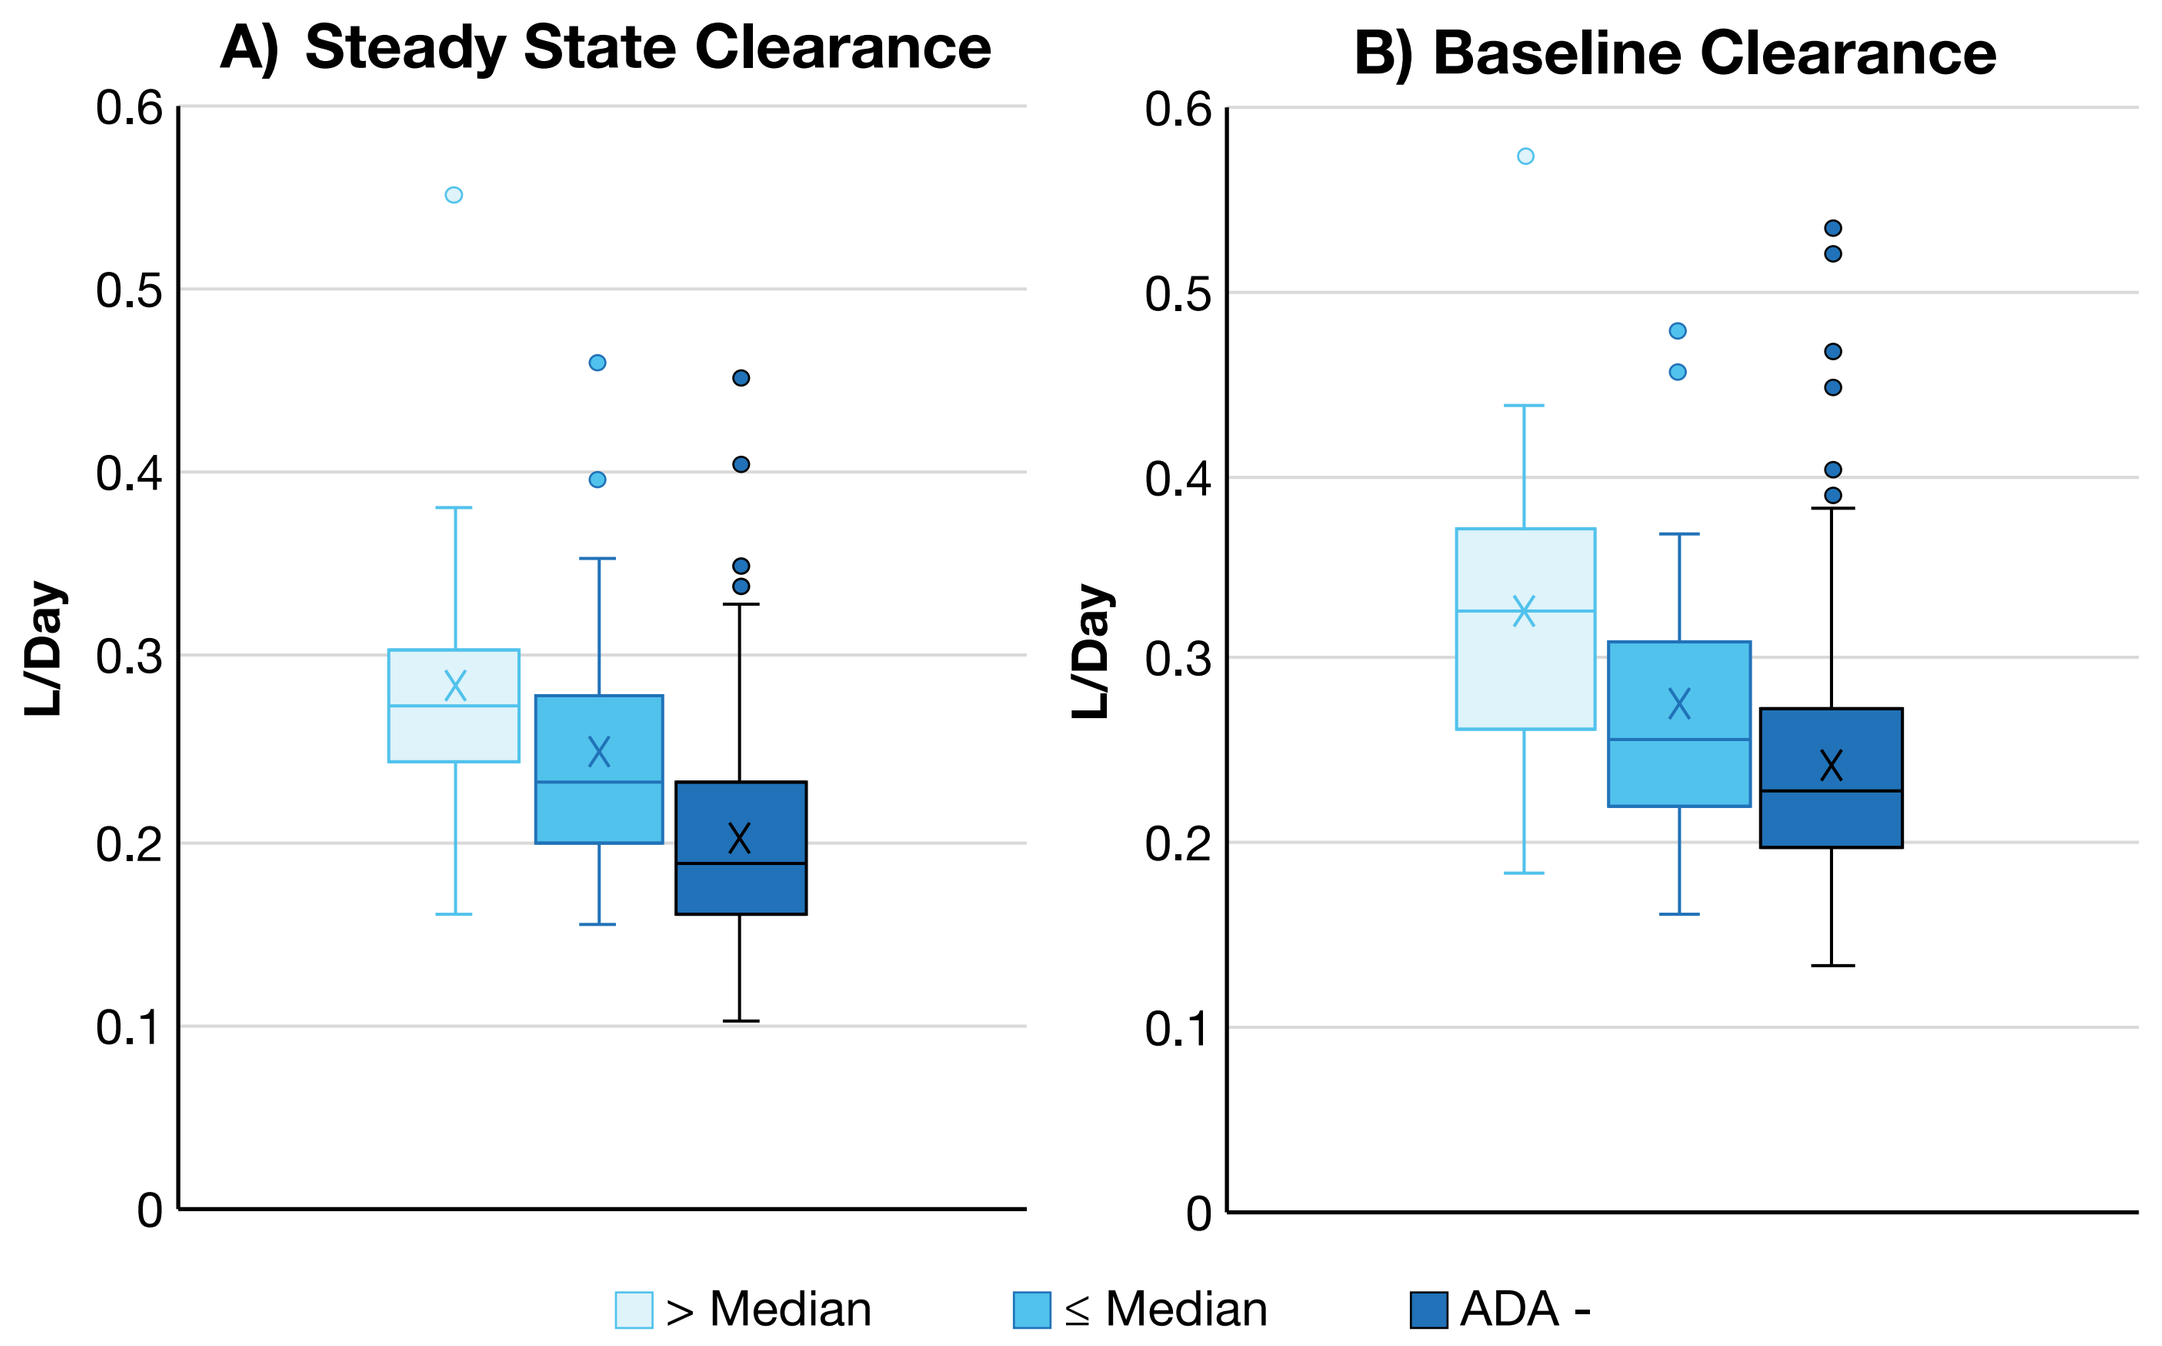
A) Steady State Clearance
B) Baseline Clearance
L/Day
> Median
≤ Median
ADA -

CONCLUSIONS

- ADAs had no clinically significant impact on efficacy and safety of camrelizumab plus rivoceranib.
- ADAs did not appear to have substantial impact on the PK of camrelizumab.
- Minor trends towards increased clearance of camrelizumab in ADA+ patients were observed in both steady state as well as baseline clearance models, where ADAs had not yet developed, possibly reflecting differences in baseline characteristics.
- Minor numeric differences observed between ADA+ and ADA- subgroups in clinical outcomes are possibly due to between-group imbalances in disease characteristics at baseline.
- Follow-up monitoring of ADAs to camrelizumab is not indicated.