Quality-adjusted Time Without Symptoms or Toxicity (Q-TWiST) Analysis to Assess the Impact of Treatment With Camrelizumab + Rivoceranib (cam+rivo) on Quality of Life vs Sorafenib in Patients With Unresectable Hepatocellular Carcinoma: Study CARES-310

Andrew Moon,[1] Natalia Raphael,[2] Stephen Chan,[3] Ahmed Kaseb,[4] Seong Jang,[2] Xianzhang Meng,[2] Ann-Lii Cheng,[5] Shukui Qin,[6] Arndt Vogel[7,8,9]

[1]UNC School of Medicine, Chapel Hill, NC, USA; [2]Elevar Therapeutics, Fort Lee, NJ, USA; [3]State Key Laboratory of Translational Oncology, Department of Clinical Oncology, Hong Kong Cancer Institute, The Chinese University of Hong Kong, Hong Kong Special Administrative Region, China; [4]Department of Gastrointestinal Medical Oncology, MD Anderson Cancer Center, Houston, TX, USA; [5]Department of Oncology National Taiwan University Hospital, Taipei, Taiwan; [6]Cancer Centre of Jinling Hospital, Nanjing University of Chinese Medicine and Nanjing Medical University, Nanjing, China; [7]Department of Gastroenterology, Hepatology and Endocrinology, Hannover Medical School, Hannover, Germany; [8]Division of Gastroenterology and Hepatology, Toronto General Hospital, Toronto, ON, Canada; [9]Department of Medical Oncology, Princess Margaret Cancer Centre, Toronto, ON, Canada

Presented at ESMO 2024

FPN# 963P

Copies of this poster obtained through QR (Quick Response) codes are for personal use only and may not be reproduced without written permission of the authors.

BACKGROUND

- Study CARES-310 (NCT03764293) compared the combination of the anti-programmed death-1 (PD-1) antibody camrelizumab plus the vascular endothelial growth factor receptor 2 (VEGFR2)-targeted tyrosine kinase inhibitor (TKI) rivoceranib (also known as apatinib) with sorafenib for the treatment of unresectable hepatocellular carcinoma.[1]
- At the protocol-specified final analysis (median follow-up: 22.1 months), camrelizumab plus rivoceranib continued to show significantly improved overall survival (OS) and progression-free survival (PFS) compared with sorafenib (median OS 23.8 months vs 15.2 months; HR 0.64 [95% CI, 0.52-0.79; P<0.0001]; mPFS 5.6 months vs 3.7 months; HR 0.54 [95% CI, 0.44-0.67; P<0.0001]).[2]
- The most common (≥10%) grade ≥3 treatment-related adverse events at the protocol-specified final analysis for camrelizumab plus rivoceranib were hypertension (38.2%), increased aspartate aminotransferase (AST) (17.3%), increased alanine aminotransferase (ALT) (14.0%), palmar-plantar erythrodysesthesia (PPE) syndrome (12.1%), and platelet count decreased (11.8%) vs PPE syndrome (15.6%) and hypertension (14.9%) for sorafenib.[2]
- We performed a Quality-adjusted Time Without Symptoms of disease progression or Toxicity of treatment (Q-TWiST) analysis based on safety data from the interim analysis to assess the impact of treatment with camrelizumab plus rivoceranib on quality of life as compared with sorafenib.

METHODS

CARES-310 Study Design and Endpoints[1]

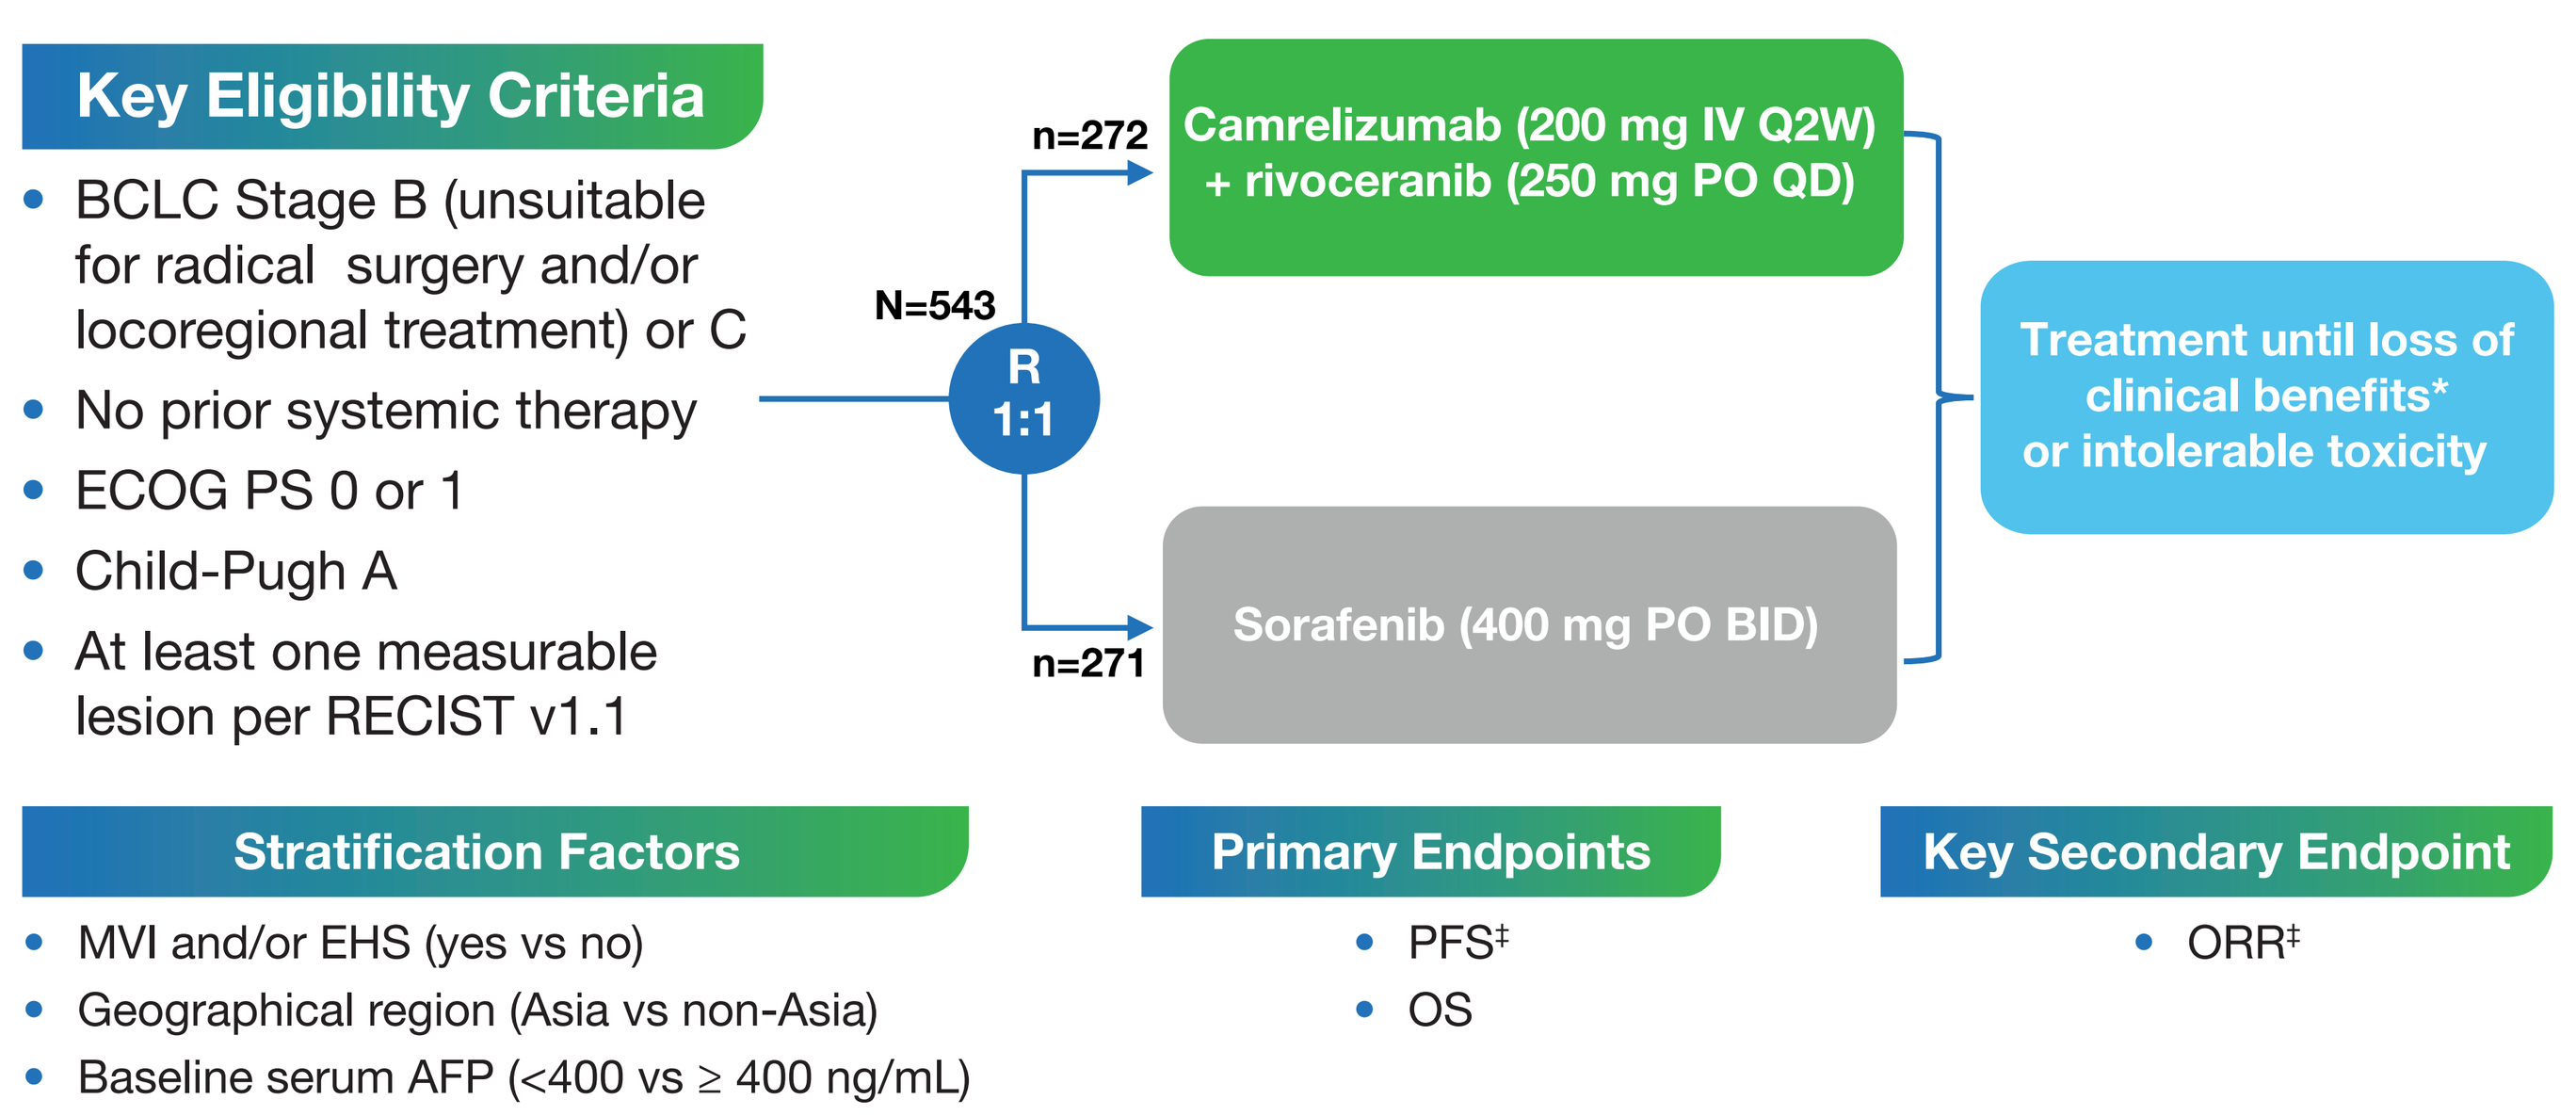
Key Eligibility Criteria
BCLC Stage B (unsuitable for radical surgery and/or locoregional treatment) or C
No prior systemic therapy
ECOG PS 0 or 1
Child-Pugh A
At least one measurable lesion per RECIST v1.1
N=543
R 1:1
n=272
Camrelizumab (200 mg IV Q2W) + rivoceranib (250 mg PO QD)
n=271
Sorafenib (400 mg PO BID)
Treatment until loss of clinical benefits* or intolerable toxicity
Stratification Factors
MVI and/or EHS (yes vs no)
Geographical region (Asia vs non-Asia)
Baseline serum AFP (<400 vs ≥ 400 ng/mL)
Primary Endpoints
PFS‡
OS
Key Secondary Endpoint
ORR‡

*Treatment beyond progression allowed if there was evidence of clinical benefits per investigator.
‡By BIRC per RECIST v1.1.
AFP, alpha-fetoprotein; BCLC, Barcelona Clinic Liver Cancer; BID, twice daily; BIRC, blinded independent review committee; ECOG PS, Eastern Cooperative Oncology Group performance status; EHS, extrahepatic spread; IV, intravenous; MVI, macrovascular invasion; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PO, by mouth; Q2W, every 2 weeks; QD, once daily; RECIST v1.1, Response Evaluation Criteria for Solid Tumors version 1.1.

Q-TWiST Analysis

- Q-TWiST is a weighted analysis that evaluates how much of a patient's survival time is spent with toxicities, after disease progression or relapse, or time without symptoms of disease or toxicity (TWiST).[3,4]

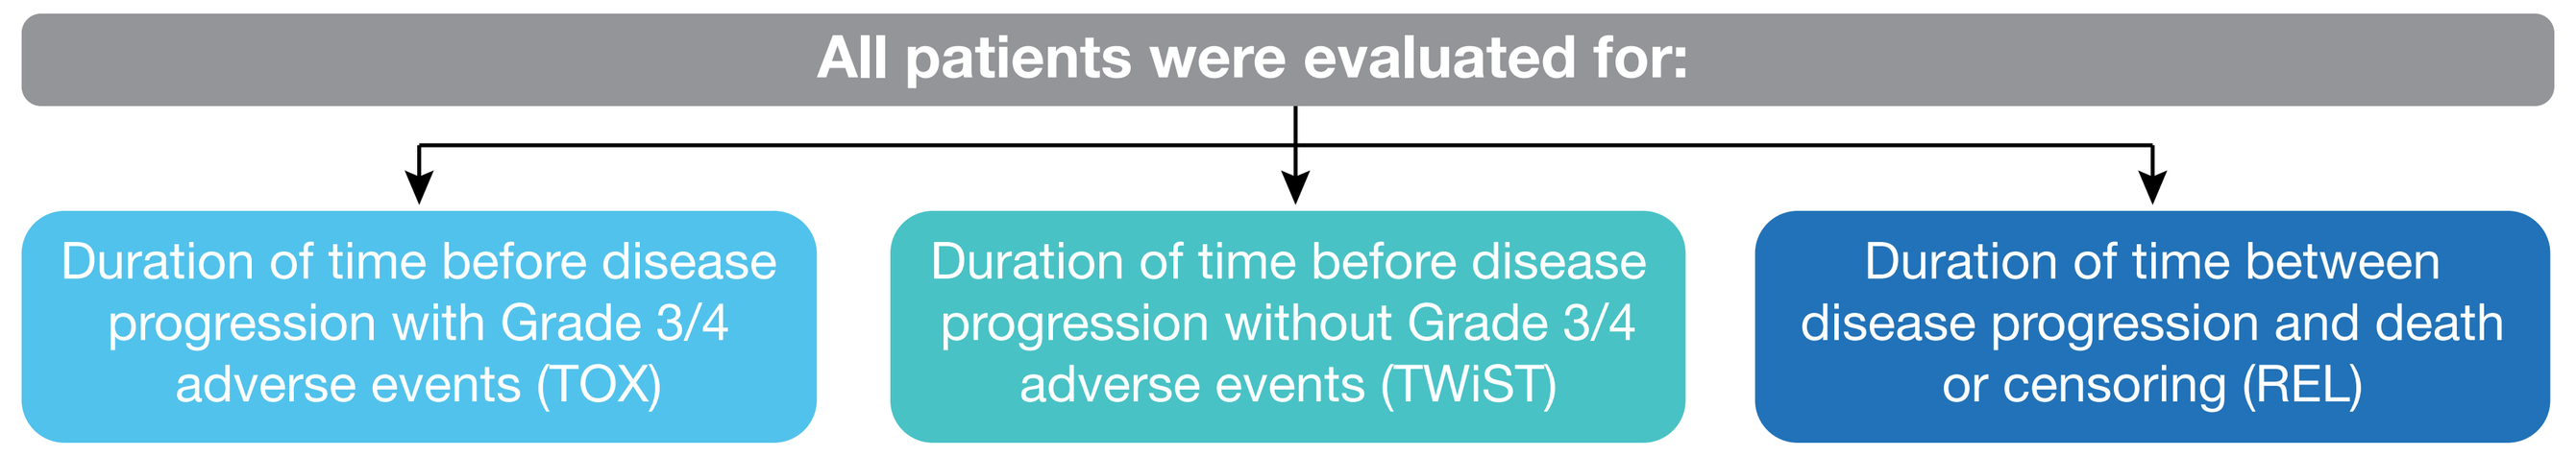
All patients were evaluated for:
Duration of time before disease progression with Grade 3/4 adverse events (TOX)
Duration of time before disease progression without Grade 3/4 adverse events (TWiST)
Duration of time between disease progression and death or censoring (REL)

- Utility coefficients (u) of 1 for TWiST, and 0.5 for TOX and REL were used to calculate Q-TWiST:

$$\text{Q-TWiST} = (u\text{TOX} * \text{TOX}) + (u\text{TWiST} * \text{TWiST}) + (u\text{REL} * \text{REL})$$

- Mean duration in each state was calculated using the method of Restricted Mean Survival Time (RMST)

REFERENCES

1. Qin S, et al. *Lancet*. 2023;402(10408):1133-1146. 2. Vogel A, et al. *J Clin Oncol*. 2024;42(suppl_16):abs 4110. 3. Solem CT, et al. *Exp Rev Pharmacoecon Outcomes Res*. 2018;18:245–53. 4. Revicki DA, et al. *Qual Life Res*. 2006;15:411–23.

ACKNOWLEDGMENTS

- This study was funded by Jiangsu Hengrui Pharmaceuticals Co., Ltd. and Elevar Therapeutics. The Phillips Group Oncology Communications Inc. provided professional assistance with poster preparation. Financial support for writing and editorial services was provided by Elevar Therapeutics.

AUTHOR INFO

DOI for presenting author, Andrew Moon: Consultant to TARGET RWE and Intercept Pharmaceuticals. Research grant (to institution) from DCN Diagnostics.
For questions or additional information, please contact medicalinformation@elevartherapeutics.com.

RESULTS

- At the interim analysis for overall survival, the most common grade 3 or 4 treatment-related adverse events were hypertension (102 [38%] of 272 patients in the camrelizumab plus rivoceranib arm vs 40 [15%] of 269 patients in the sorafenib arm), palmar-plantar erythrodysaesthesia syndrome (33 [12%] vs 41 [15%]), increased aspartate aminotransferase (45 [17%] vs 14 [5%]), and increased alanine aminotransferase (35 [13%] vs 8 [3%]) (**Table 1**).

Table 1: TRAEs in The Safety Analysis Set at the Interim Analysis for Overall Survival[1]

	Camrelizumab + rivoceranib (n=272)				Sorafenib (n=269)			
TRAE, n (%)	Grade 1-2	Grade 3	Grade 4	Grade 5	Grade 1-2	Grade 3	Grade 4	Grade 5
Any TRAE	45 (17)	193 (71)	26 (10)	1 (<1)	128 (48)	128 (48)	12 (4)	1 (<1)
Hypertension	87 (32)	100 (37)	2 (1)	0	76 (28)	40 (15)	0	0
AST increased	102 (38)	42 (15)	3 (1)	0	85 (32)	14 (5)	0	0
Proteinuria	118 (43)	16 (6)	0	0	67 (25)	5 (2)	0	0
ALT increased	92 (43)	34 (13)	1 (<1)	0	72 (27)	8 (3)	0	0
Platelet count decreased	94 (35)	28 (10)	4 (1)	0	85 (32)	4 (1)	0	0
Blood bilirubin increased	92 (34)	24 (9)	0	0	71 (26)	4 (1)	0	0
Palmar-plantar erythrodysaesthesia syndrome	69 (25)	33 (12)	0	0	122 (45)	41 (15)	0	0
Diarrhea	77 (28)	6 (2)	0	0	91 (34)	14 (5)	0	0
Reactive cutaneous capillary endothelial proliferation	72 (26)	7 (3)	0	0	0	0	0	0
Neutrophil count decreased	57 (21)	14 (5)	2 (1)	0	24 (9)	1 (<1)	2 (1)	0
White blood cell count decreased	66 (24)	7 (3)	0	0	35 (13)	3 (1)	0	0
Gamma-glutamyl transferase increased	39 (14)	25 (9)	2 (1)	0	29 (11)	15 (6)	5 (2)	0
Hypothyroidism	58 (21)	0	0	0	16 (6)	0	0	0
Fatigue	46 (17)	7 (3)	0	0	20 (7)	1 (<1)	0	0
Blood alkaline phosphatase increased	44 (16)	3 (1)	0	0	30 (11)	3 (1)	0	0
Conjugated blood bilirubin increased	34 (13)	10 (4)	2 (1)	0	28 (10)	6 (2)	2 (1)	0
Rash	40 (15)	5 (2)	0	0	47 (17)	3 (1)	0	0
Anemia	41 (15)	4 (1)	0	0	19 (7)	2 (1)	0	0
Decreased appetite	39 (14)	3 (1)	0	0	31 (12)	3 (1)	0	0
Unconjugated blood bilirubin increased	33 (12)	2 (1)	0	0	20 (7)	1 (<1)	0	0
Hypoalbuminemia	34 (13)	0	0	0	21 (8)	0	0	0
Weight decreased	28 (10)	4 (1)	0	0	33 (12)	6 (2)	0	0
Asthenia	29 (11)	3 (1)	0	0	15 (6)	0	0	0
Hematuria	31 (11)	0	0	0	12 (4)	0	0	0
Nausea	31 (11)	0	0	0	14 (5)	0	0	0
Headache	28 (10)	2 (1)	0	0	4 (1)	1 (<1)	0	0
Blood lactate dehydrogenase increased	26 (10)	1 (<1)	0	0	29 (11)	0	0	0
Lymphocyte count decreased	18 (7)	8 (3)	0	0	14 (5)	3 (1)	0	0
Amylase increased	15 (6)	9 (3)	1 (<1)	0	6 (2)	0	1 (<1)	0
Hyponatremia	13 (5)	8 (3)	0	0	8 (3)	1 (<1)	0	0
Lipase increased	7 (3)	7 (3)	6 (2)	0	6 (2)	4 (1)	1 (<1)	0
Hypophosphatemia	17 (6)	2 (1)	0	0	27 (10)	12 (4)	0	0
Upper GI hemorrhage	2 (1)	6 (2)	0	0	0	0	0	0
Alopecia	4 (1)	0	0	0	52 (19)	0	0	0

ALT, alanine aminotransferase; AST, aspartate aminotransferase; GI, gastrointestinal; TRAE, treatment related adverse event.

Q-TWiST Analysis Results

- Duration of TOX and TWiST was longer for patients in the camrelizumab plus rivoceranib arm vs sorafenib arm (**Table 2**).
- Duration of REL was not significantly different between arms (**Table 2**).
- Clinically important relative Q-TWiST gains favored camrelizumab plus rivoceranib vs sorafenib by 10.6% (= difference 1.61/median overall survival of sora 15.2) (**Table 2**, **Figure 1**, and **Figure 2**).

Table 2: Mean Duration of TOX, TWiST, REL, and Q-TWiST

Mean duration, months (SE)	Camrelizumab + rivoceranib (n=272)	Sorafenib (n=271)	Difference, Camrelizumab + rivoceranib vs Sorafenib	P value
TOX	2.18 (0.2)	1.21 (0.14)	0.97 (0.25)	<0.0001
TWiST	8.92 (0.67)	6.01 (0.55)	2.91 (0.87)	0.0008
REL	13.71 (0.67)	12.64 (0.61)	1.07 (0.91)	0.2375
Q-TWiST	10.11 (0.31)	8.51 (0.29)	1.61 (0.42)	0.0002

REL, duration of time between disease progression and death or censoring; SE, standard of error; TOX, duration of time before disease progression with Grade 3/4 adverse events; TWiST, duration of time before disease progression without Grade 3/4 adverse events.

Figure 1: Kaplan-Meier Plot for Partitioned Overall Survival: Camrelizumab + Rivoceranib

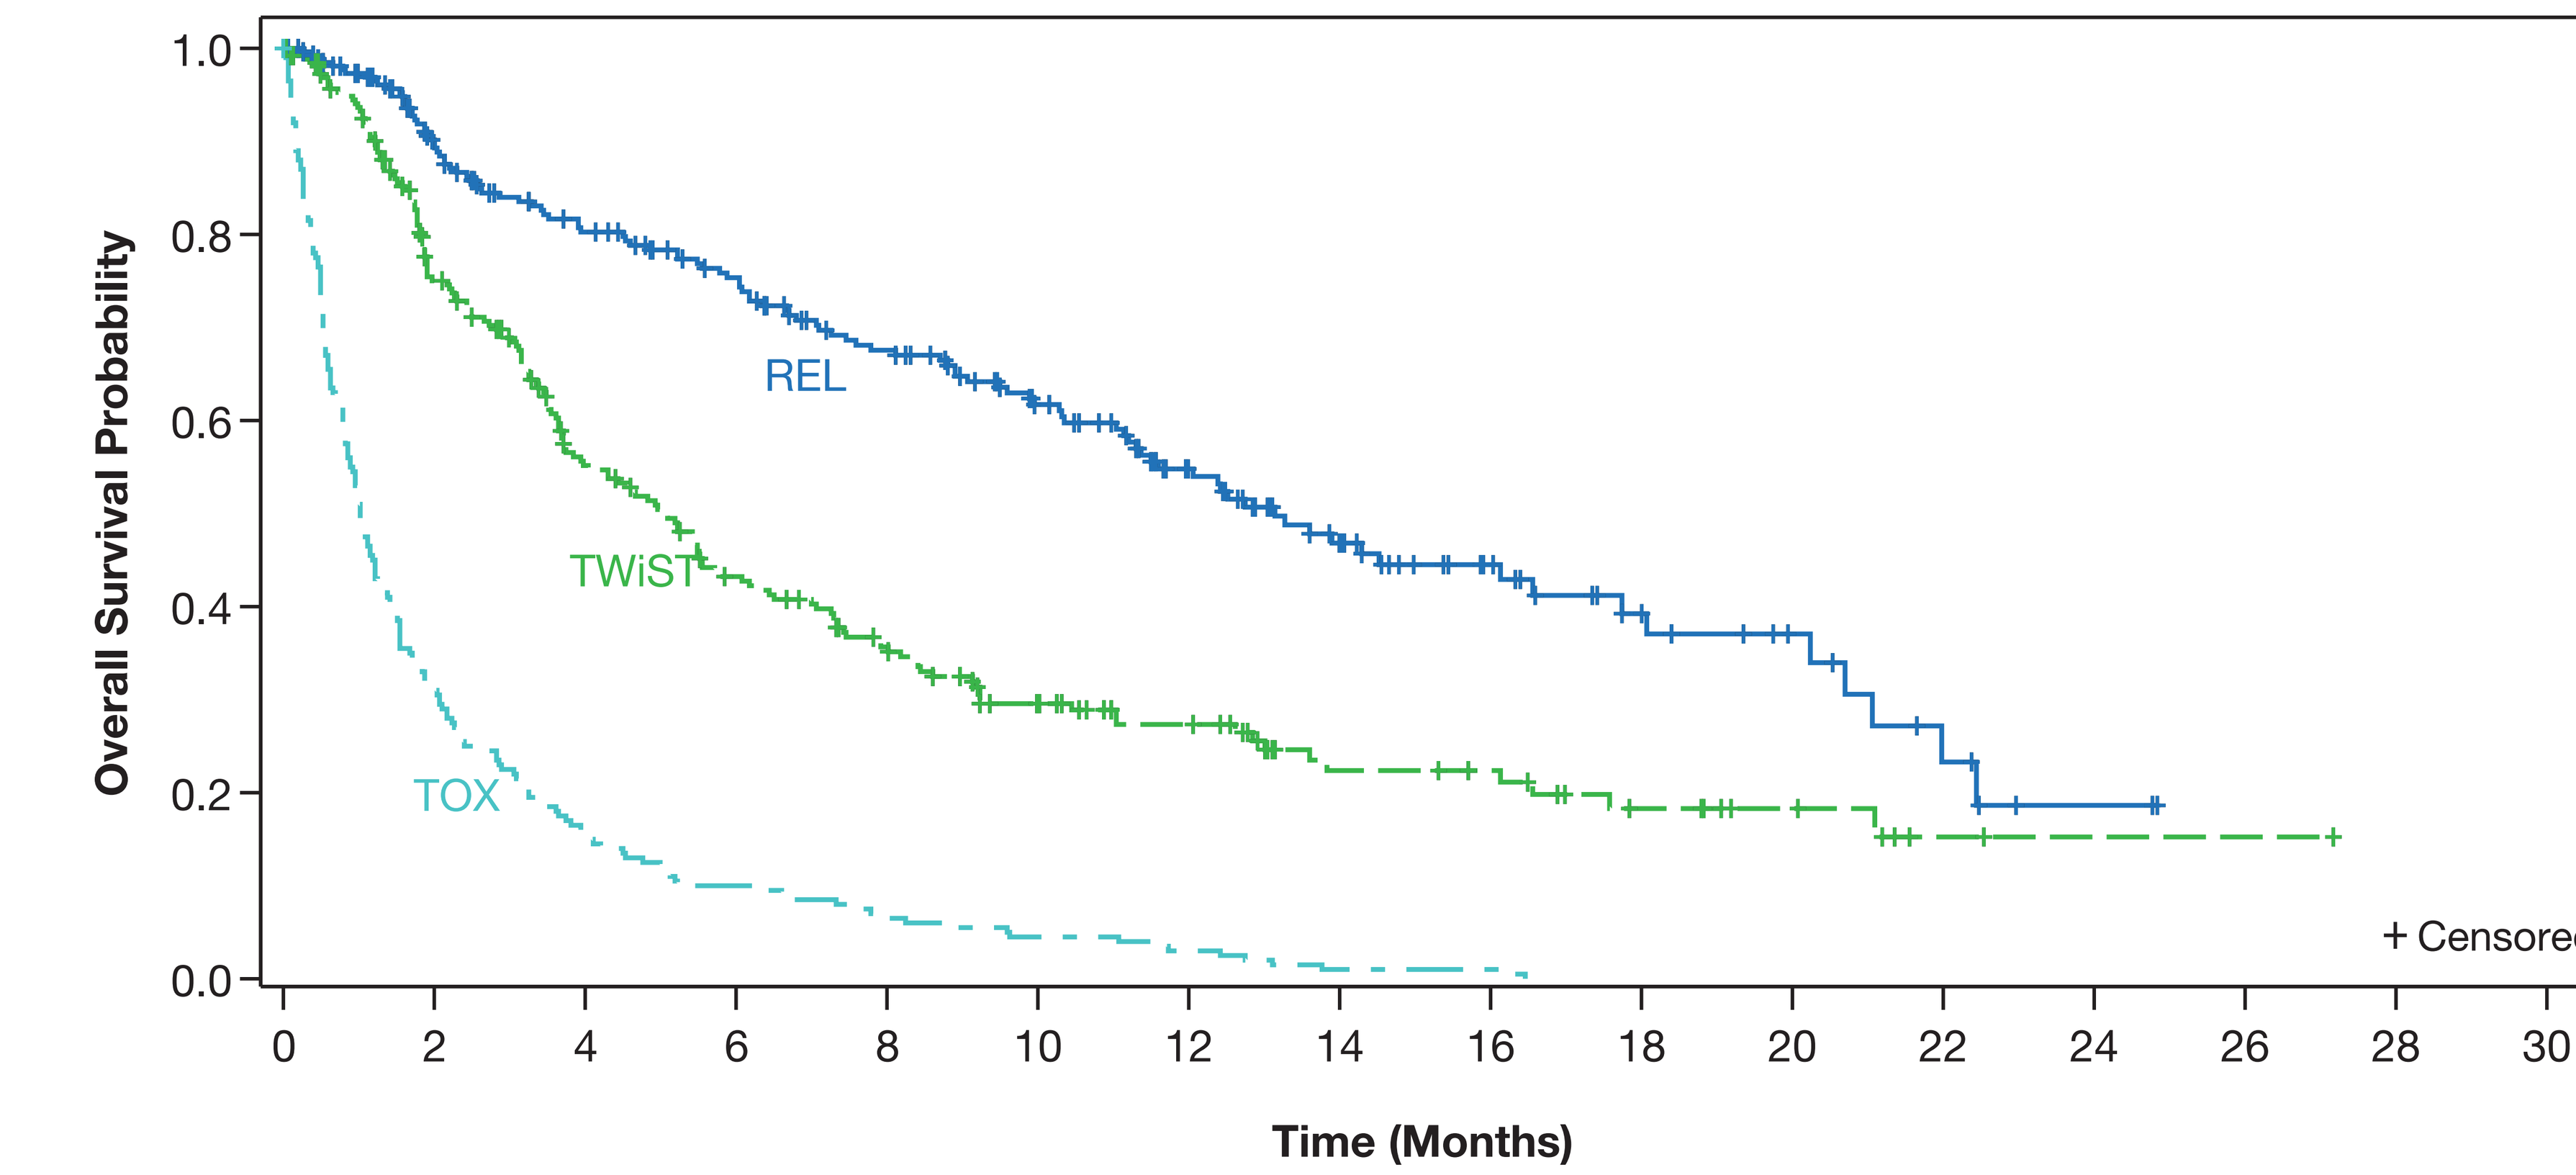
Overall Survival Probability
1.0
0.8
0.6
0.4
0.2
0.0
REL
TWiST
TOX
+ Censored
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Time (Months)

Number of patients at risk

	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28
REL	272	207	171	150	126	96	68	46	29	19	12	6	2	0	
TWiST	272	174	118	88	68	47	35	20	18	11	7	2	1	1	0
TOX	272	62	31	20	13	9	6	2	2	0					

REL, time from disease progression until death or the last recorded date on which patients were known to be alive; TOX, toxicity time for grade 3/4 adverse events; TWiST, time without toxicity or disease progression.

Figure 2: Kaplan-Meier Plot for Partitioned Overall Survival: Sorafenib

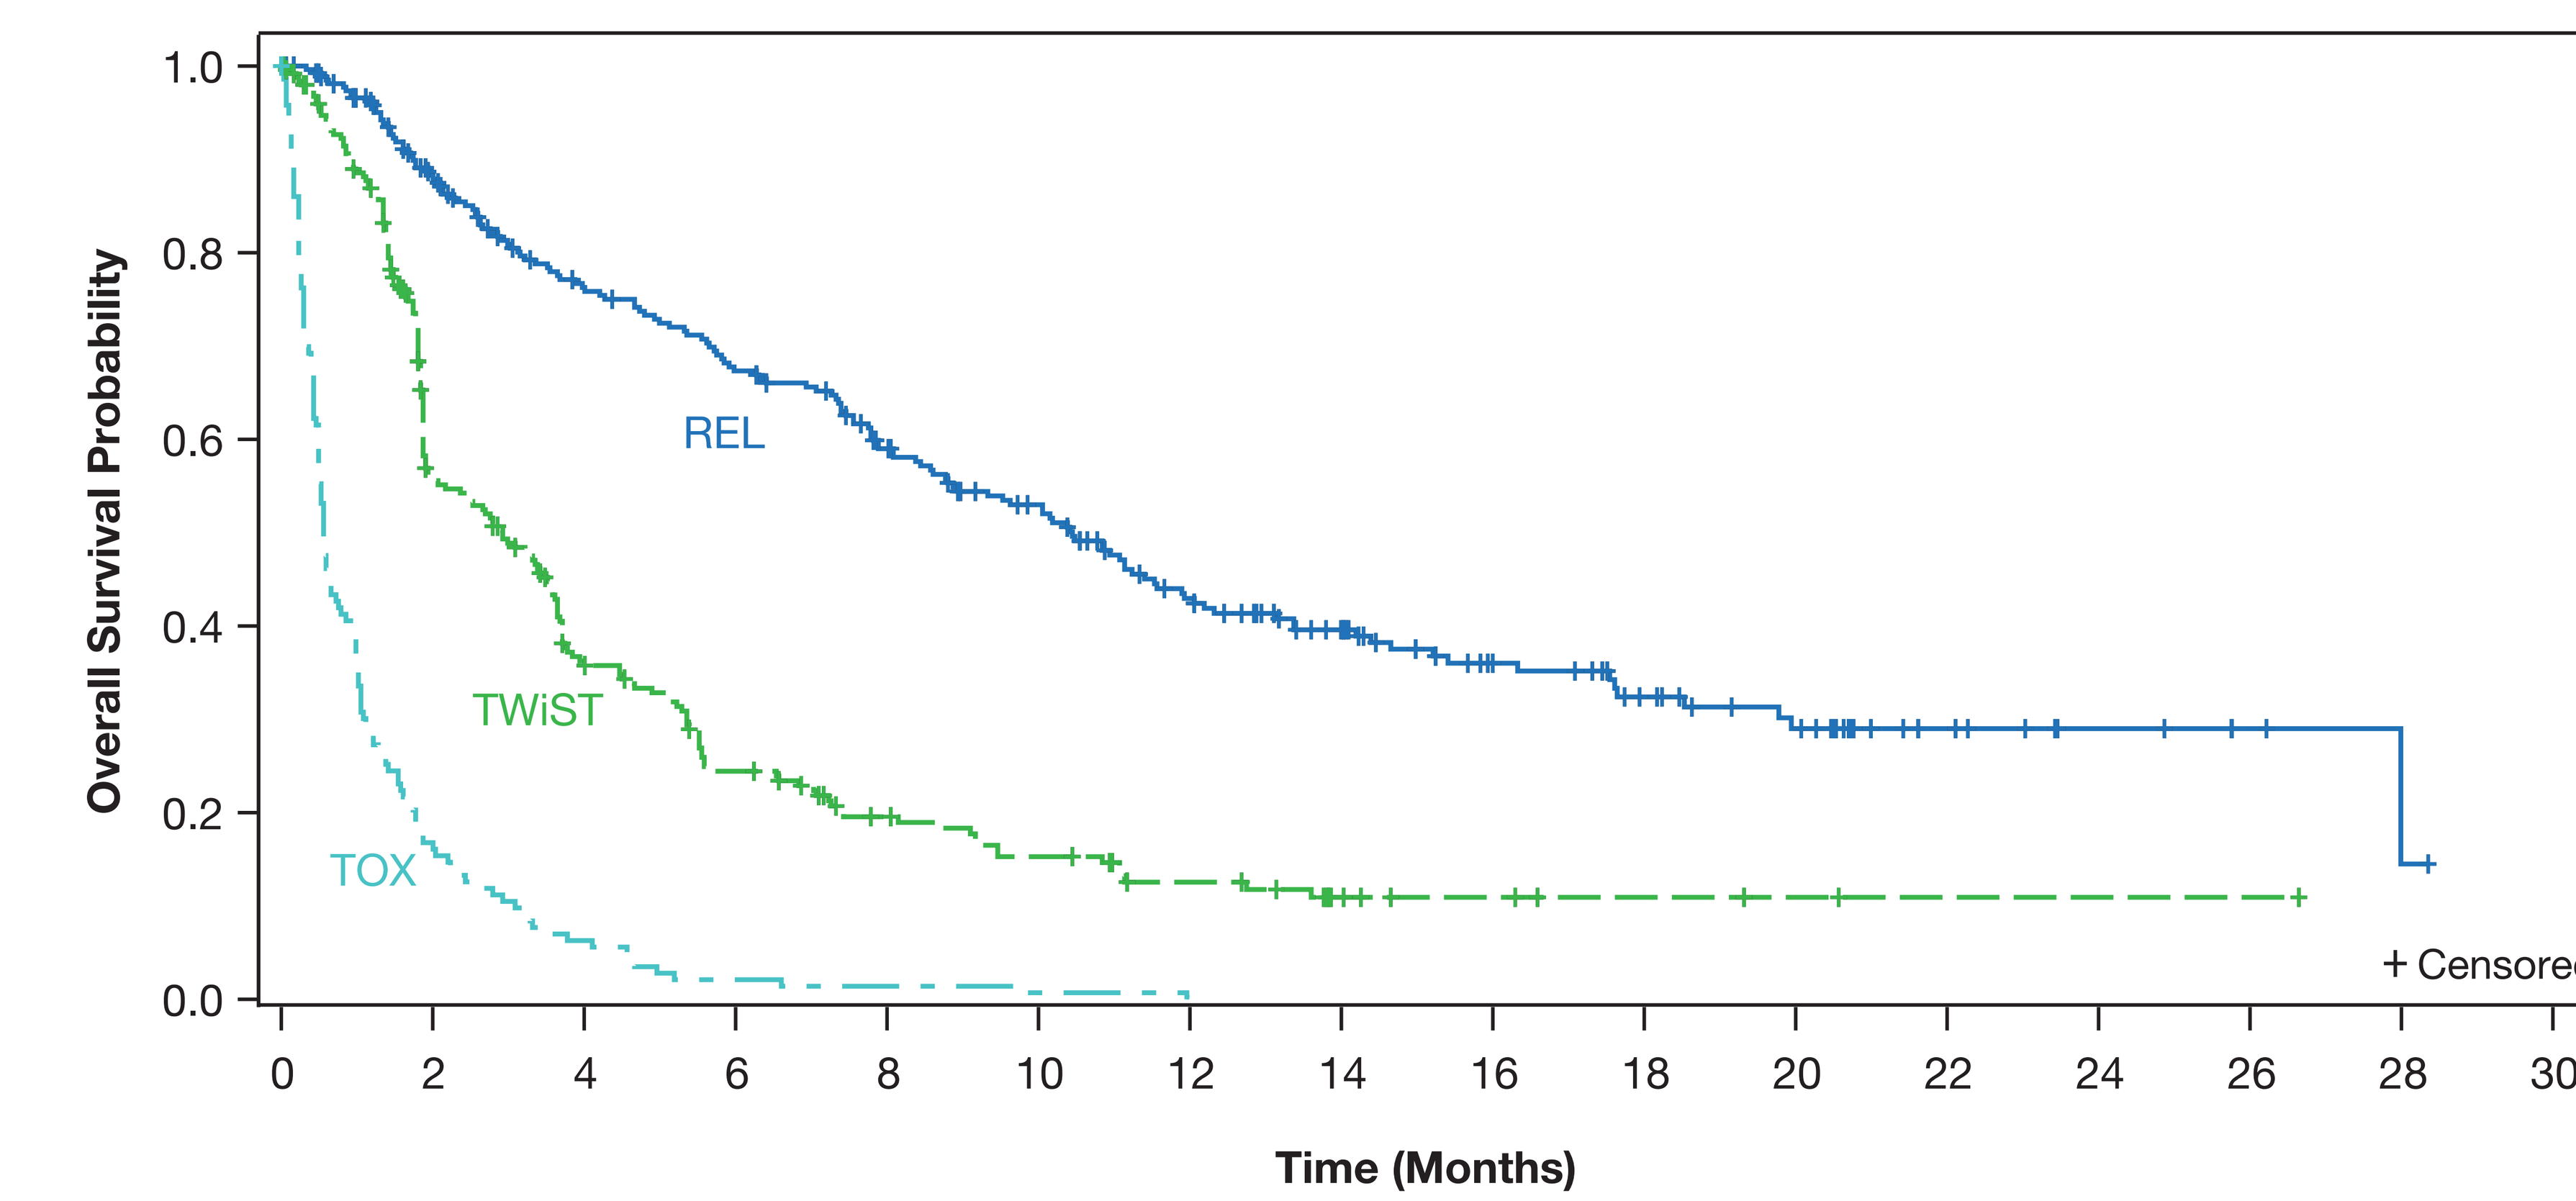
Overall Survival Probability
1.0
0.8
0.6
0.4
0.2
0.0
REL
TWiST
TOX
+ Censored
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30
Time (Months)

Number of patients at risk

	0	2	4	6	8	10	12	14	16	18	20	22	24	26	28	30
REL	269	219	180	158	131	110	82	63	44	33	25	12	7	3	1	0
TWiST	269	126	76	49	33	25	17	8	5	3	2	1	1	1	0	
TOX	269	24	9	3	2	1	0									

REL, time from disease progression until death or the last recorded date on which patients were known to be alive; TOX, toxicity time for grade 3/4 adverse events; TWiST, time without toxicity or disease progression.

CONCLUSIONS

- Patients with unresectable hepatocellular carcinoma treated with camrelizumab plus rivoceranib had clinically meaningful quality-adjusted survival benefits compared with patients treated with sorafenib.
- This benefit was driven primarily by time before disease progression without toxicity, supporting that the higher incidence of adverse events in patients treated with camrelizumab + rivoceranib vs sorafenib is likely due to longer treatment duration with camrelizumab