Clinical outcomes of camrelizumab + rivoceranib vs sorafenib (CARES-310) as first-line treatment for patients with unresectable hepatocellular carcinoma (uHCC) of viral and non-viral etiology

Rachna T. Shroff,[1] Wei Shi,[2] Xiuzhi Wu,[2] Xianzhang Meng,[3] Kristin Ryan,[3] Christopher Galloway,[3] Laura Alexander[3]

[1]University of Arizona Cancer Center, Tucson, AZ, USA; [2]Jiangsu Hengrui Pharmaceuticals, Shanghai, China; [3]Elevar Therapeutics, Fort Lee, NJ, USA

Presented at ASCO GI 2025

Abstract #578

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® or the author of this poster.

BACKGROUND

- CARES-310 (NCT03764293) compared the combination of the anti-programmed death-1 (PD-1) antibody camrelizumab plus the vascular endothelial growth factor receptor 2 (VEGFR2)-targeted tyrosine kinase inhibitor (TKI) rivoceranib versus sorafenib for the treatment of unresectable HCC.[1]
- Camrelizumab plus rivoceranib showed significantly improved median overall survival (mOS) and median progression-free survival (mPFS) compared with sorafenib (mOS 23.8 months vs 15.2 months; HR 0.64 [95% CI, 0.52-0.79; P<0.0001]; mPFS 5.6 months vs 3.7 months; HR 0.54 [95% CI, 0.44-0.67; P<0.0001]).[2]
- The most common ≥5% grade 3-4 treatment-related adverse events for camrelizumab plus rivoceranib were hypertension (38%) and increased aspartate aminotransferase (AST; 17%) vs palmar-plantar erythrodysesthesia syndrome (16%) for sorafenib.[2]
- We performed a post-hoc analysis of the CARES-310[2] study evaluating the impact of viral, hepatitis B virus infection or hepatitis C virus infection, and non-viral HCC etiology on survival and safety outcomes.

METHODS

CARES-310 Study Design and Endpoints[1]

Key Eligibility Criteria

- HCC
- BCLC Stage B (unsuitable for radical surgery and/or locoregional treatment) or C
- No prior systemic therapy
- ECOG PS 0 or 1
- Child-Pugh A
- At least one measurable lesion per RECIST v1.1

N=543

R 1:1

n=272: Camrelizumab (200 mg IV Q2W) + Rivoceranib (250 mg PO QD)

n=271: Sorafenib (400 mg PO BID)

Treatment until loss of clinical benefits* or intolerable toxicity

Stratification Factors

- MVI and/or EHS (yes vs no)
- Geographical region (Asia vs non-Asia)
- Baseline serum AFP (<400 vs ≥ 400 ng/mL)

Primary Endpoints

- PFS‡
- OS

Key Secondary Endpoint

- ORR‡

*Treatment beyond progression allowed if there was evidence of clinical benefits per investigator. ‡By BIRC per RECIST v1.1.
AFP, alpha-fetoprotein; BCLC, Barcelona Clinic Liver Cancer; BID, twice daily; BIRC, blinded independent review committee; ECOG PS, Eastern Cooperative Oncology Group performance status; EHS, extrahepatic spread; IV, intravenous; MVI, macrovascular invasion; ORR, overall response rate; OS, overall survival; PFS, progression-free survival; PO, by mouth; Q2W, every 2 weeks; QD, once daily; RECIST v1.1, Response Evaluation Criteria for Solid Tumors version 1.1.

Post-Hoc Analysis

- mOS and mPFS were estimated using the Kaplan-Meier method and compared across three HCC etiology groups of hepatitis B virus infection, hepatitis C virus infection, and non-viral using the log-rank test.

RESULTS

Baseline Characteristics by HCC Etiology

Category	Hepatitis B Virus		Hepatitis C Virus		Non-Viral	
	Rivo + Cam (N=208)	Sorafenib (N=197)	Rivo + Cam (N=22)	Sorafenib (N=29)	Rivo + Cam (N=42)	Sorafenib (N=45)
Median age, years (range)	54 (23-82)	53 (23-76)	66 (50-83)	61 (50-82)	67 (39-79)	67 (28-82)
Male sex, n (%)	174 (83.7)	175 (88.8)	19 (86.4)	23 (79.3)	34 (81.0)	32 (71.1)
Geographical region, n (%)						
Asia	200 (96.2)	191 (97.0)	11 (50.0)	10 (34.5)	14 (33.3)	23 (51.1)
Non-Asia	8 (3.8)	6 (3.0)	11 (50.0)	19 (65.5)	28 (66.7)	22 (48.9)
Race, n (%)						
Asian	201 (96.6)	191 (97.0)	11 (50.0)	10 (34.5)	14 (33.3)	23 (51.1)
Caucasian (non-Asian)	7 (3.4)	6 (3.0)	11 (50.0)	19 (65.5)	28 (66.7)	22 (48.9)
BCLC stage, n (%)						
B (Middle Stage)	26 (12.5)	26 (13.2)	5 (22.7)	7 (24.1)	7 (16.7)	7 (15.6)
C (Advanced Stage)	182 (87.5)	171 (86.8)	17 (77.3)	22 (75.9)	35 (83.3)	38 (84.4)
Child-Pugh Score, n (%)						
A5	184 (88.5)	167 (84.8)	18 (81.8)	24 (82.8)	34 (81.0)	39 (86.7)
A6	24 (11.5)	30 (15.2)	4 (18.2)	5 (17.2)	8 (19.0)	6 (13.3)
ECOG PS, n (%)						
0	90 (43.3)	79 (40.1)	11 (50.0)	17 (58.6)	19 (45.2)	20 (44.4)
1	118 (56.7)	118 (59.9)	11 (50.0)	12 (41.4)	23 (54.8)	25 (55.6)
AFP, n (%)						
<400 ng/mL	129 (62.0)	120 (60.9)	15 (68.2)	20 (69.0)	32 (76.2)	31 (68.9)
≥400 ng/mL	79 (38.0)	77 (39.1)	7 (31.8)	9 (31.0)	10 (23.8)	14 (31.1)

AFP, alpha-fetoprotein; BCLC, Barcelona Clinic Liver Cancer; Cam, camrelizumab; ECOG PS, Eastern Cooperative Oncology Group Performance Status; HCC, hepatocellular carcinoma; Rivo, rivoceranib.
Percentages are based on number of subjects in each treatment group per HCC Etiology, HBV, HCV and non-viral classification. Non-viral included alcohol cirrhosis, genetic, metabolic disorders (such as NASH/AFLD) and others. ITT patient population

Overall Survival: Hepatitis B Virus Infection HCC

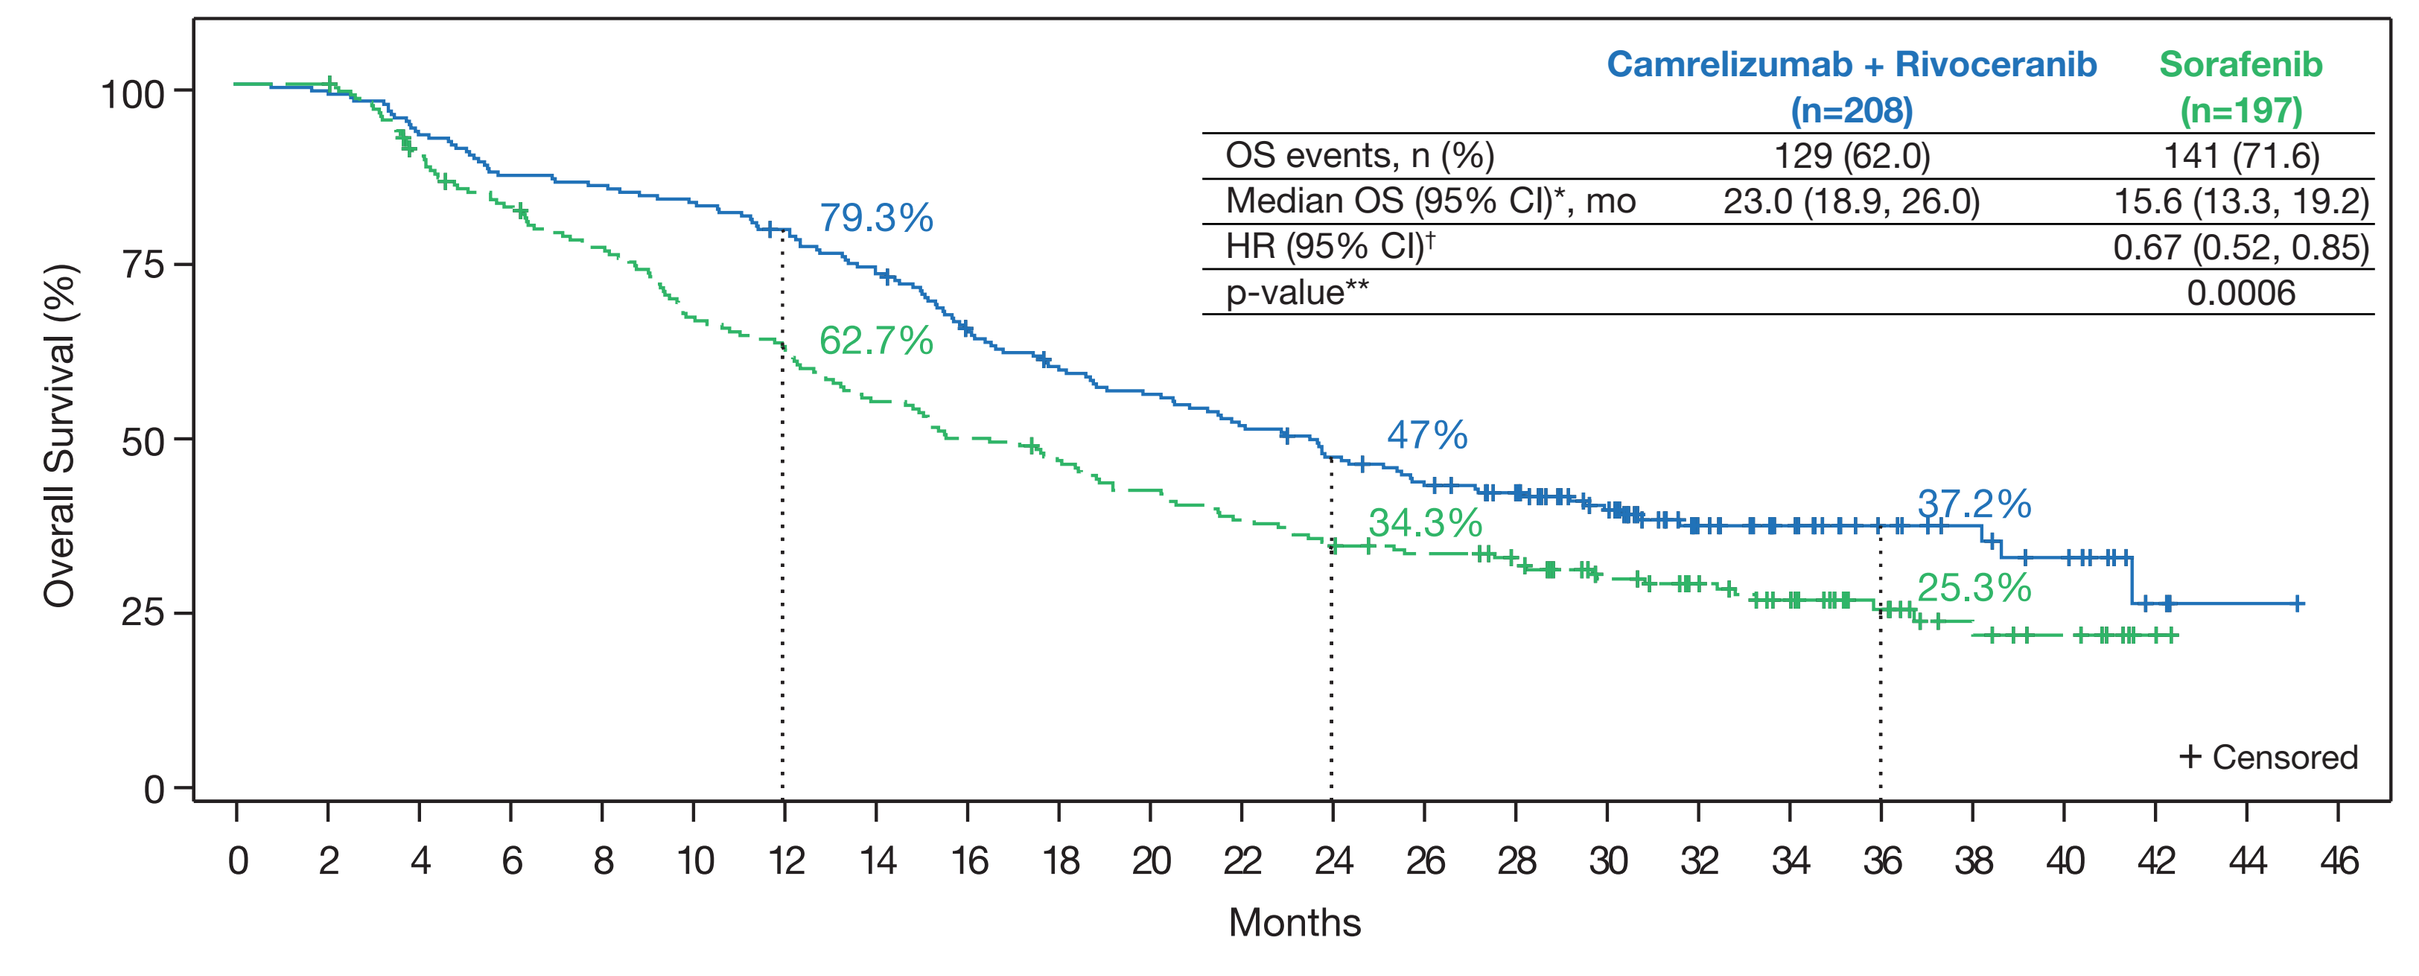

Overall Survival (%)
Months
79.3%
62.7%
47%
34.3%
37.2%
25.3%
+ Censored

	Camrelizumab + Rivoceranib (n=208)	Sorafenib (n=197)
OS events, n (%)	129 (62.0)	141 (71.6)
Median OS (95% CI)*, mo	23.0 (18.9, 26.0)	15.6 (13.3, 19.2)
HR (95% CI)†		0.67 (0.52, 0.85)
p-value**		0.0006

Number of patients at risk:
Cam + Rivo 208 206 194 181 178 173 164 153 134 121 113 104 94 86 78 62 42 31 22 17 13 3 1 0
Sorafenib 197 197 174 159 147 128 120 105 95 89 80 72 65 61 56 45 39 31 19 12 8 2 0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence), geographical region (Asia vs countries outside of Asia) and baseline AFP (AFP < 400 ng/mL vs AFP ≥ 400 ng/mL) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Overall Survival: Hepatitis C Virus Infection HCC

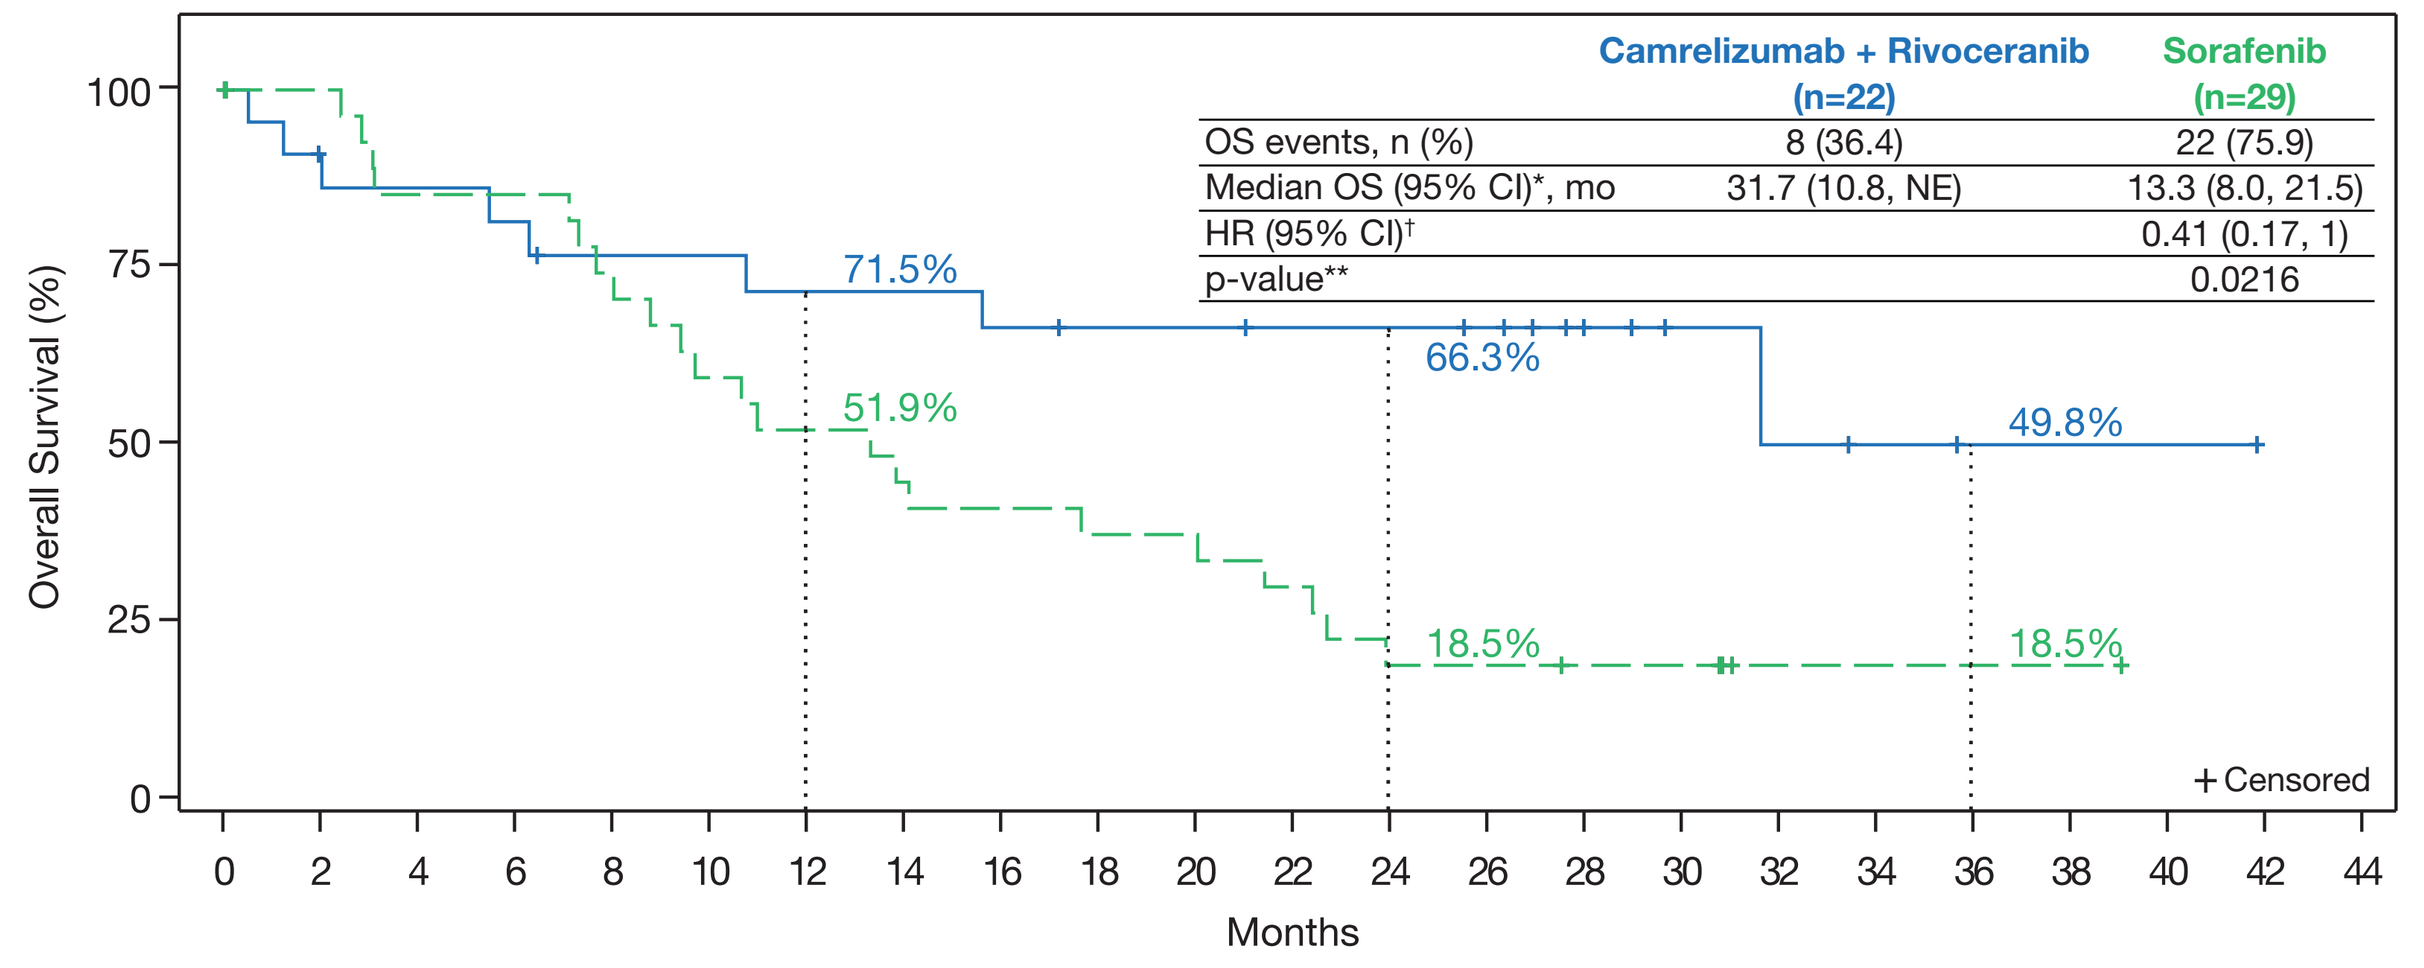

Overall Survival (%)
Months
71.5%
51.9%
66.3%
18.5%
49.8%
18.5%
+ Censored

	Camrelizumab + Rivoceranib (n=22)	Sorafenib (n=29)
OS events, n (%)	8 (36.4)	22 (75.9)
Median OS (95% CI)*, mo	31.7 (10.8, NE)	13.3 (8.0, 21.5)
HR (95% CI)†		0.41 (0.17, 1)
p-value**		0.0216

Number of patients at risk:
Cam + Rivo 22 19 18 17 15 15 14 14 13 12 12 11 11 10 7 4 3 2 1 1 1 0
Sorafenib 29 27 23 23 20 16 14 12 11 10 10 8 5 5 4 4 1 1 1 1 0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence), geographical region (Asia vs countries outside of Asia) and baseline AFP (AFP < 400 ng/mL vs AFP ≥ 400 ng/mL) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Overall Survival: Non-Viral HCC

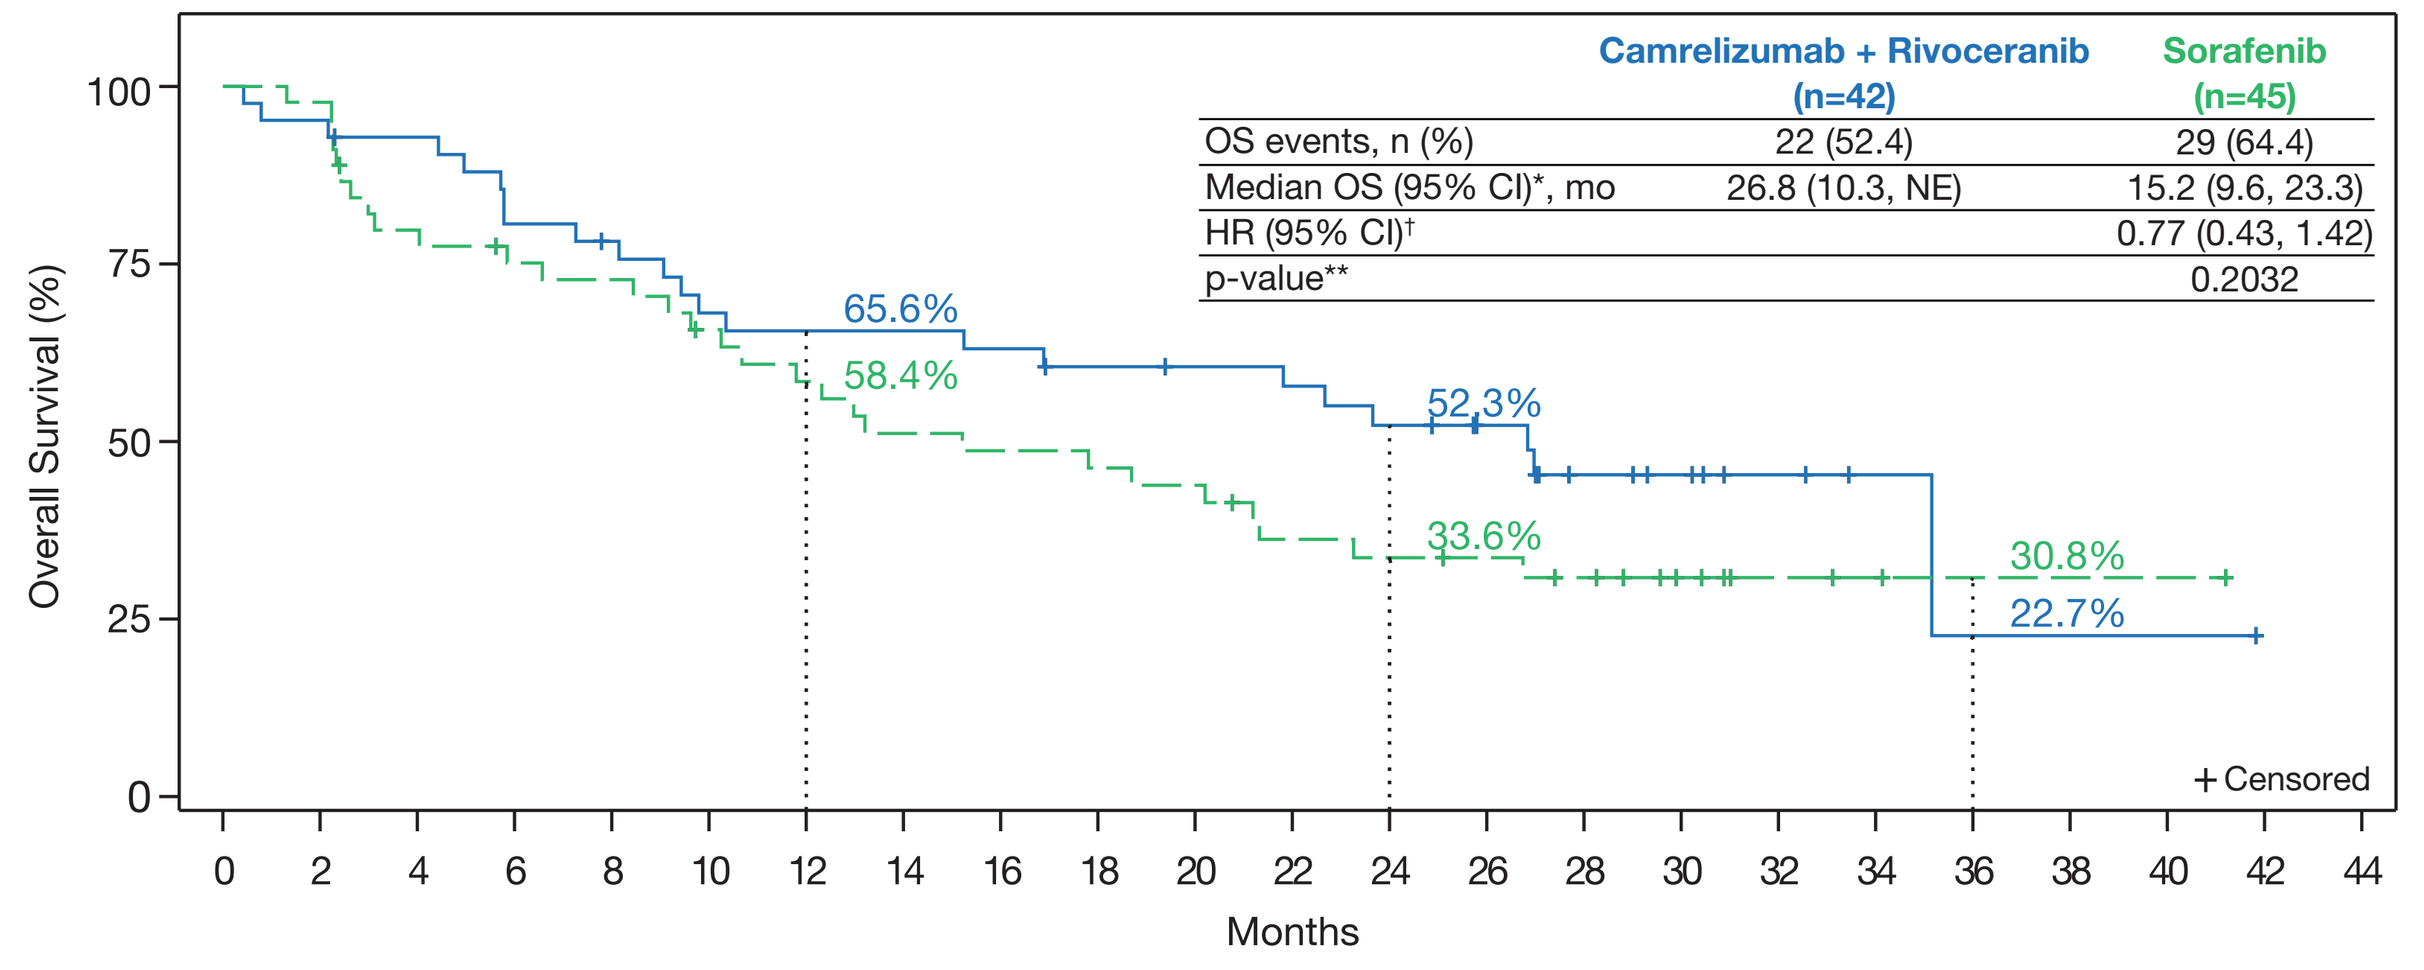

Overall Survival (%)
Months
65.6%
58.4%
52.3%
33.6%
30.8%
22.7%
+ Censored

	Camrelizumab + Rivoceranib (n=42)	Sorafenib (n=45)
OS events, n (%)	22 (52.4)	29 (64.4)
Median OS (95% CI)*, mo	26.8 (10.3, NE)	15.2 (9.6, 23.3)
HR (95% CI)†		0.77 (0.43, 1.42)
p-value**		0.2032

Number of patients at risk:
Cam + Rivo 42 40 38 33 31 27 26 26 25 23 22 21 19 15 9 7 4 2 1 1 1 0
Sorafenib 45 44 35 32 31 27 24 21 20 19 18 14 13 12 10 6 3 2 1 1 1 0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence), geographical region (Asia vs countries outside of Asia) and baseline AFP (AFP < 400 ng/mL vs AFP ≥ 400 ng/mL) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Progression-Free Survival: Hepatitis B Virus Infection HCC

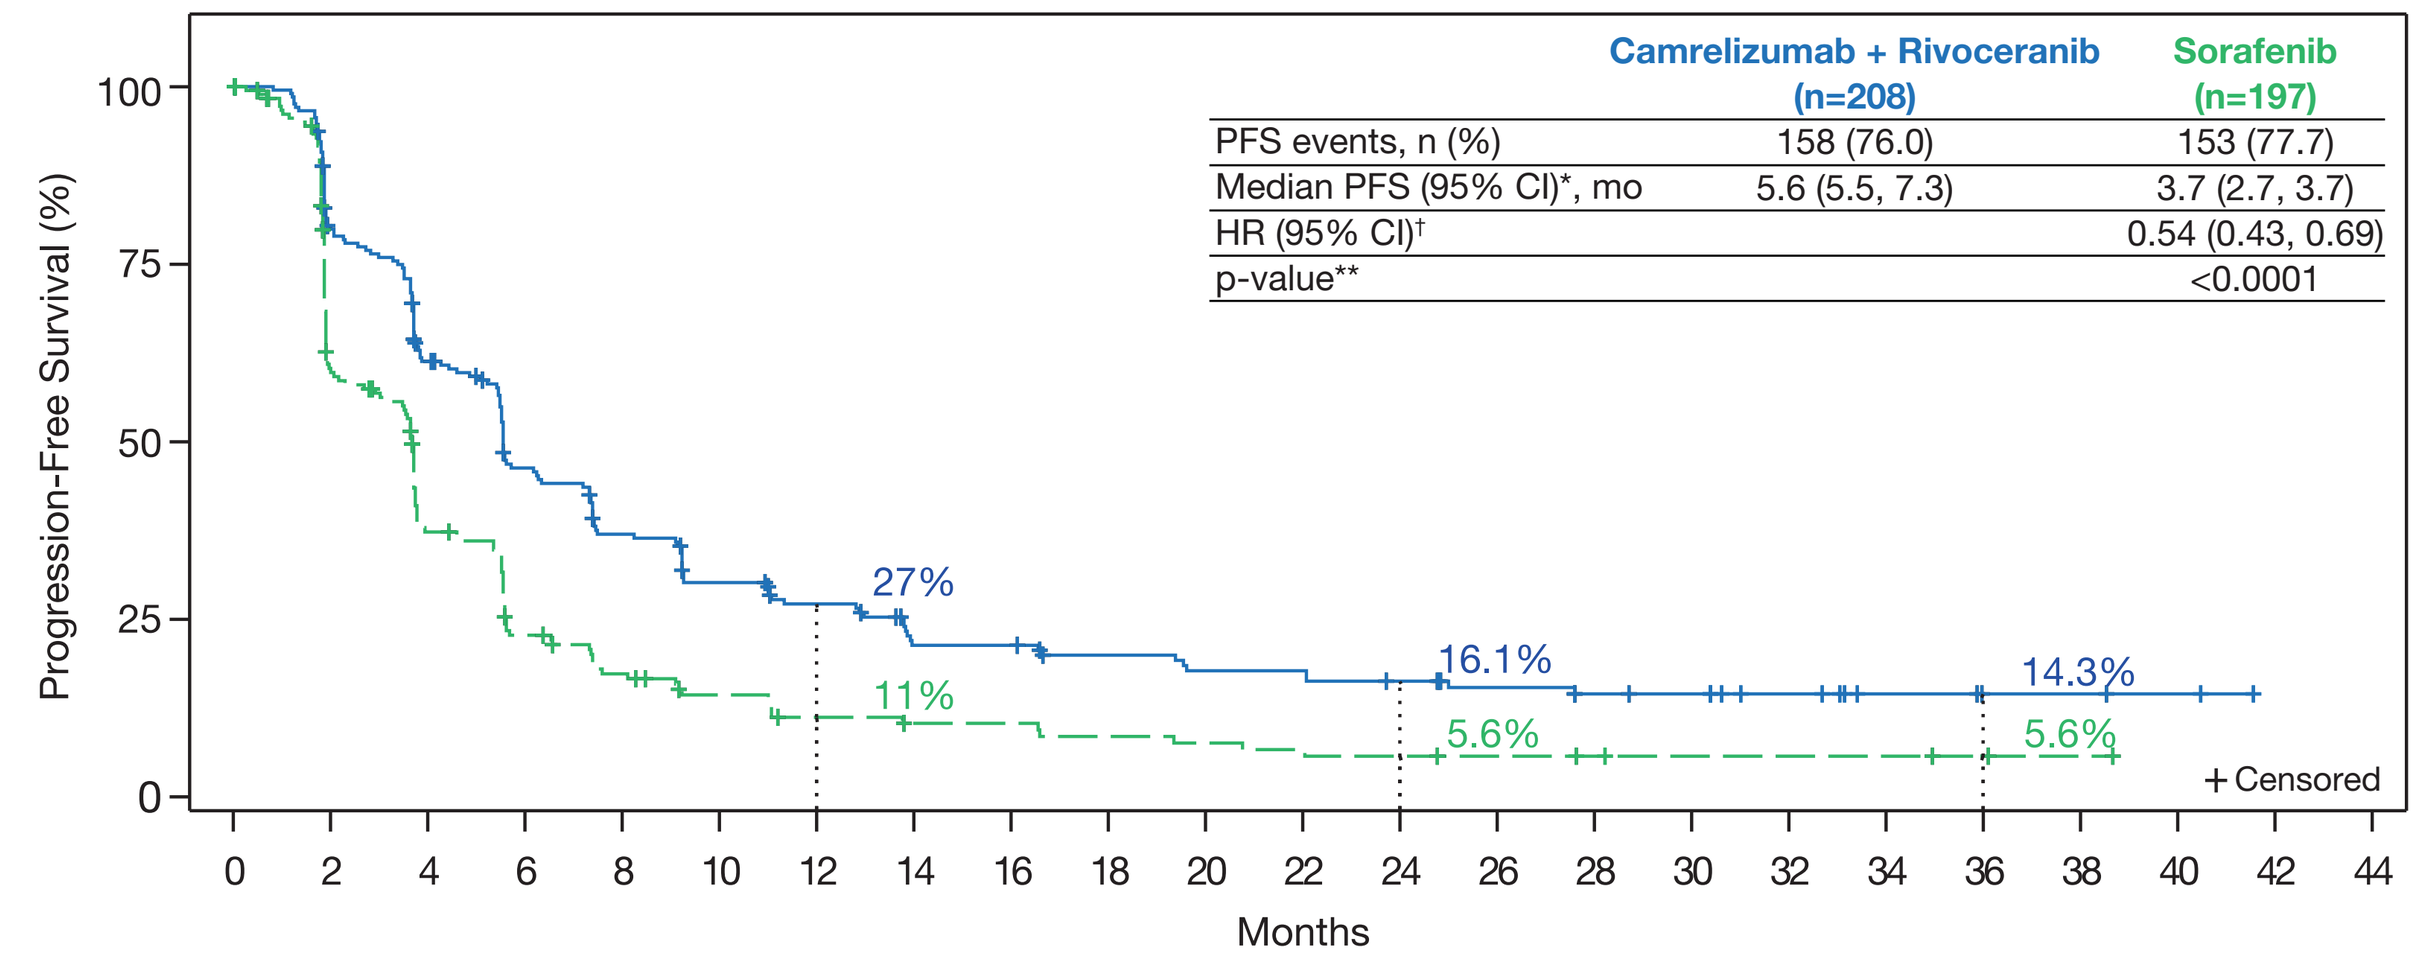

Progression-Free Survival (%)
Months
27%
11%
16.1%
5.6%
14.3%
5.6%
+ Censored

	Camrelizumab + Rivoceranib (n=208)	Sorafenib (n=197)
PFS events, n (%)	158 (76.0)	153 (77.7)
Median PFS (95% CI)*, mo	5.6 (5.5, 7.3)	3.7 (2.7, 3.7)
HR (95% CI)†		0.54 (0.43, 0.69)
p-value**		<0.0001

Number of patients at risk:
Cam + Rivo 208 161 118 85 66 52 44 32 32 27 24 24 21 17 14 13 9 5 3 3 2 0
Sorafenib 197 104 60 35 25 18 13 11 11 9 8 7 6 5 4 3 3 3 2 1 0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence), geographical region (Asia vs countries outside of Asia) and baseline AFP (AFP < 400 ng/mL vs AFP ≥ 400 ng/mL) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Progression-Free Survival: Hepatitis C Virus Infection HCC

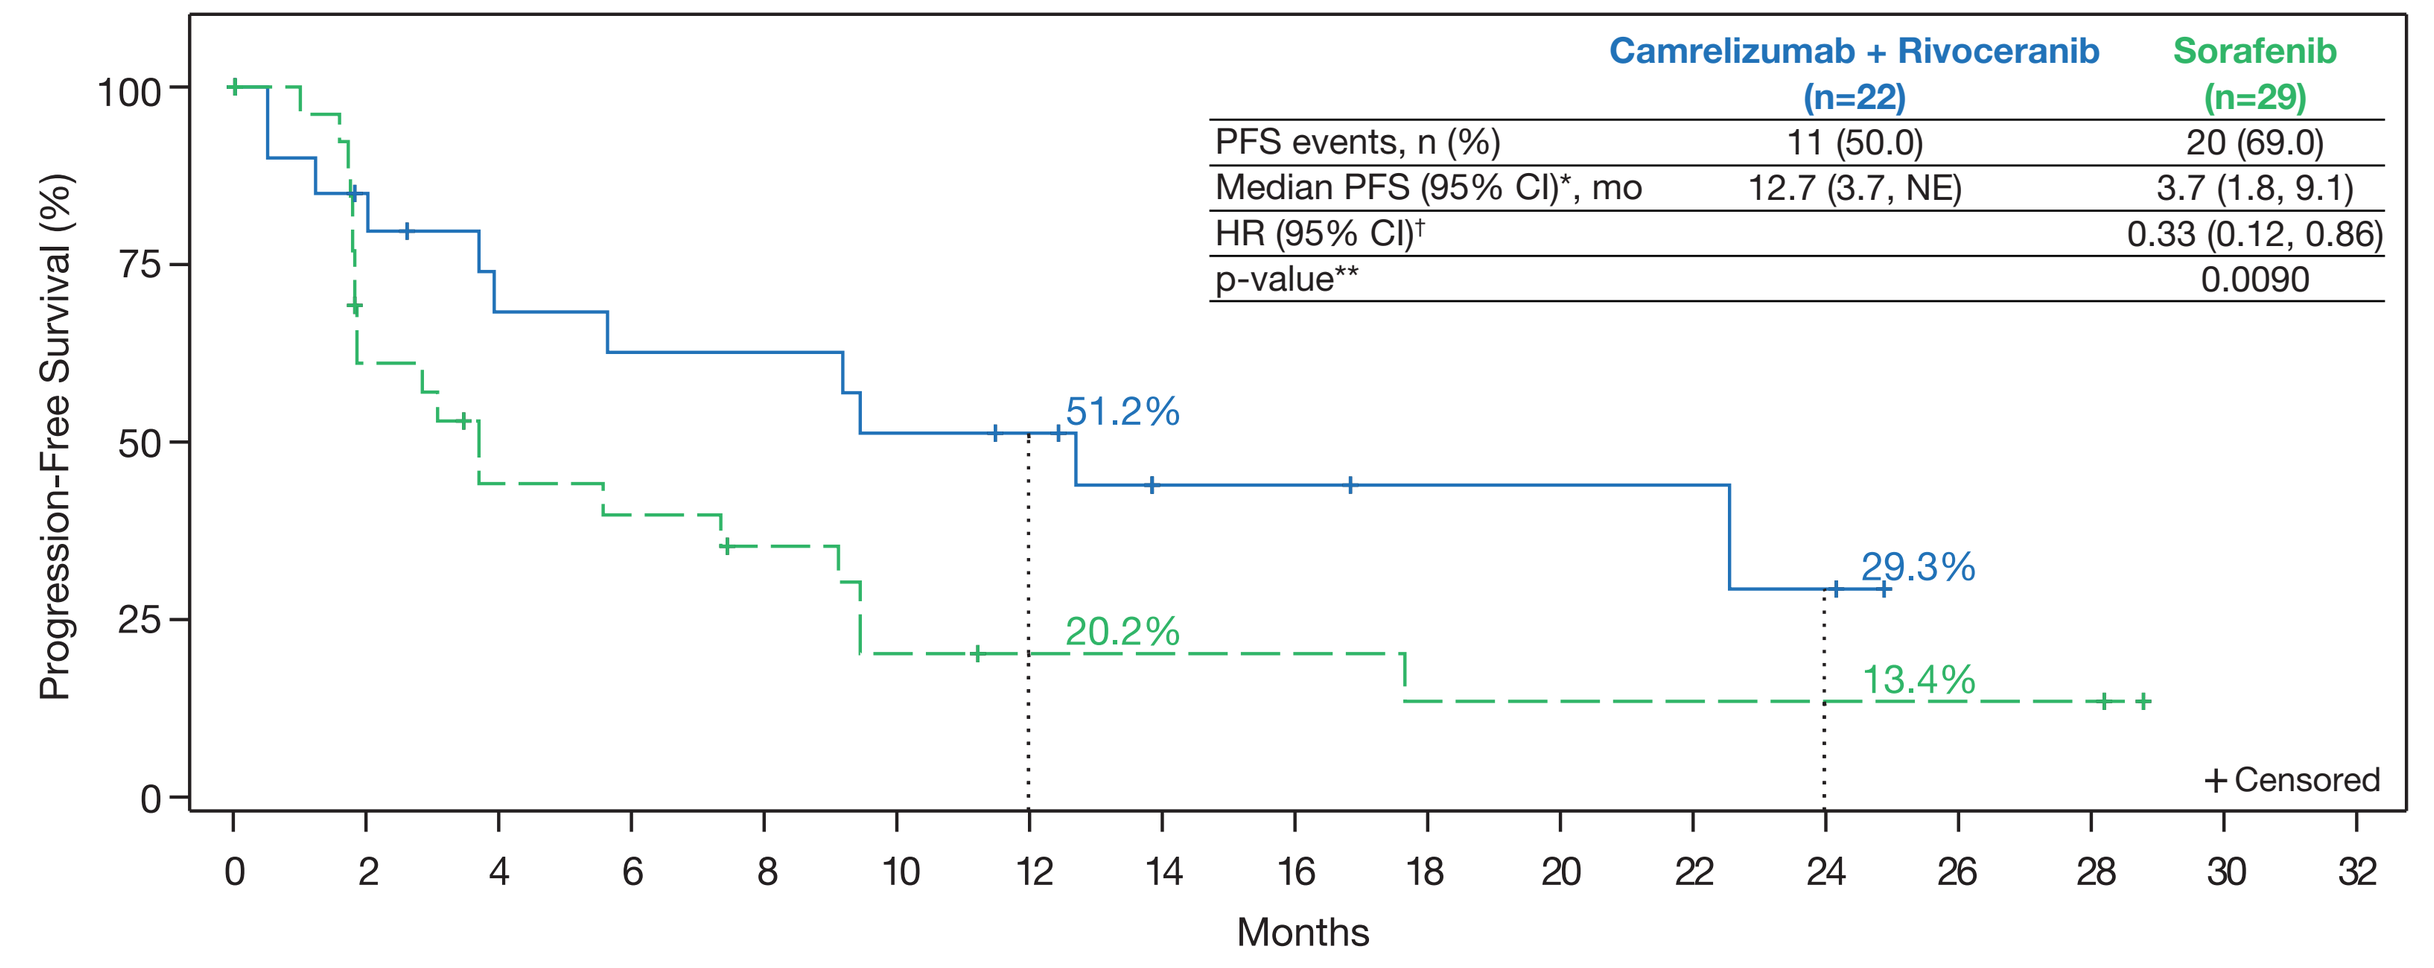

Progression-Free Survival (%)
Months
51.2%
20.2%
29.3%
13.4%
+ Censored

	Camrelizumab + Rivoceranib (n=22)	Sorafenib (n=29)
PFS events, n (%)	11 (50.0)	20 (69.0)
Median PFS (95% CI)*, mo	12.7 (3.7, NE)	3.7 (1.8, 9.1)
HR (95% CI)†		0.33 (0.12, 0.86)
p-value**		0.0090

Number of patients at risk:
Cam + Rivo 22 16 12 11 11 9 8 4 4 3 3 3 2 0
Sorafenib 29 15 10 9 7 4 3 3 3 2 2 2 2 2 2 0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence), geographical region (Asia vs countries outside of Asia) and baseline AFP (AFP < 400 ng/mL vs AFP ≥ 400 ng/mL) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Progression-Free Survival: Non-Viral HCC

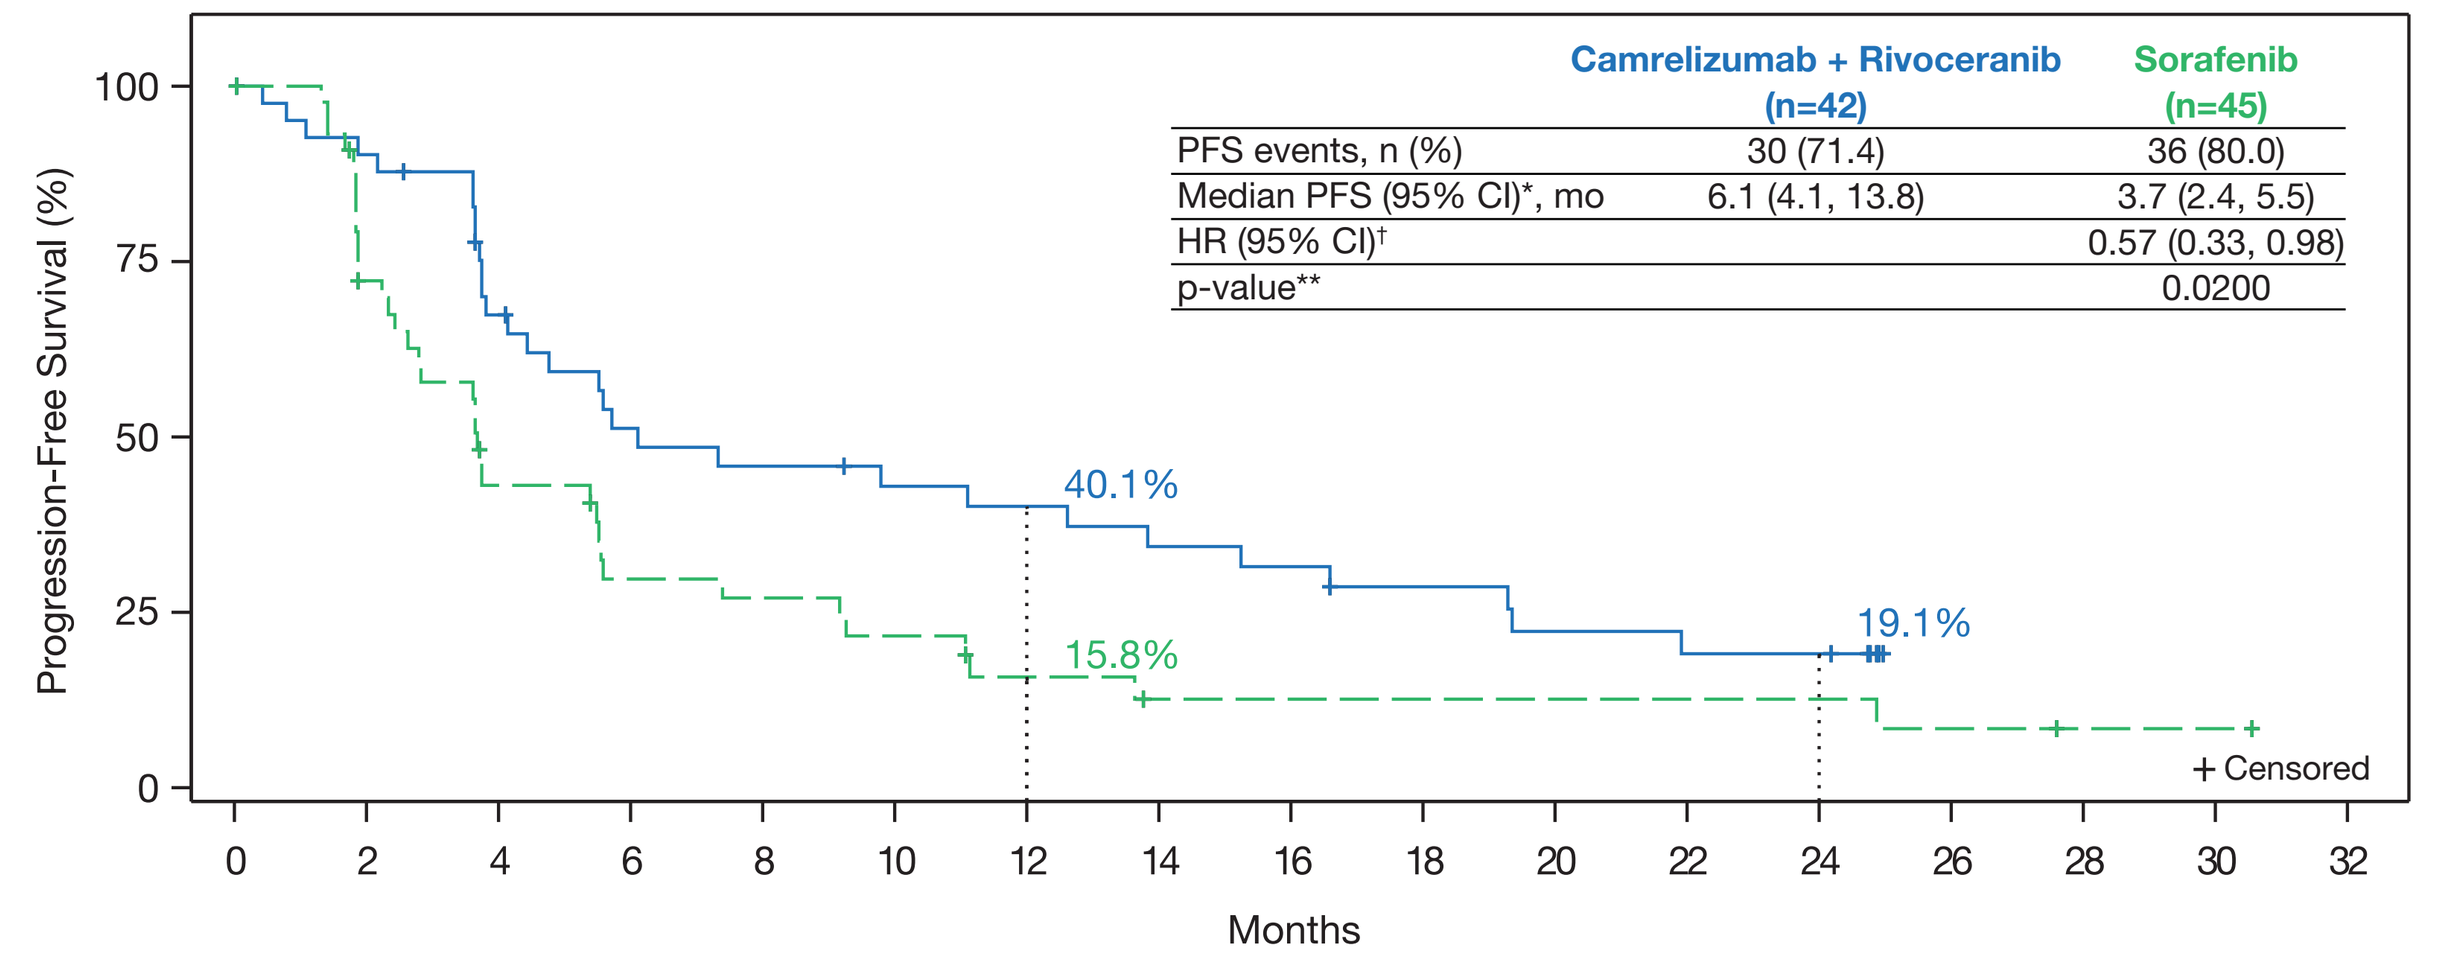

Progression-Free Survival (%)
Months
40.1%
15.8%
19.1%
+ Censored

	Camrelizumab + Rivoceranib (n=42)	Sorafenib (n=45)
PFS events, n (%)	30 (71.4)	36 (80.0)
Median PFS (95% CI)*, mo	6.1 (4.1, 13.8)	3.7 (2.4, 5.5)
HR (95% CI)†		0.57 (0.33, 0.98)
p-value**		0.0200

Number of patients at risk:
Cam + Rivo 42 37 26 19 17 15 14 12 11 9 7 6 6 0
Sorafenib 45 30 17 11 10 8 5 3 3 3 3 3 3 2 1 1 0

*Medians were estimated using the Kaplan-Meier methods with CIs calculated using Brookmeyer and Crowley method.
†Hazard ratios and the corresponding 95% CIs were stratified by macrovascular invasion and/or extrahepatic metastasis (presence vs absence), geographical region (Asia vs countries outside of Asia) and baseline AFP (AFP < 400 ng/mL vs AFP ≥ 400 ng/mL) in the interactive response technology (IRT) system.
**p-value (one-sided) is calculated based on log-rank test.

Response and Disease Control Rate by BIRC Assessment (RECIST v1.1)

	Hepatitis B Virus		Hepatitis C Virus		Non-Viral	
	Rivo + Cam (N=208)	Sorafenib (N=197)	Rivo + Cam (N=22)	Sorafenib (N=29)	Rivo + Cam (N=42)	Sorafenib (N=45)
Best overall response, n (%)						
Complete response (CR)	3 (1.4)	0	1 (4.5)	2 (6.9)	1 (2.4)	0
Partial response (PR)	52 (25.0)	10 (5.1)	5 (22.7)	2 (6.9)	11 (26.2)	2 (4.4)
Stable disease (SD)	103 (49.5)	91 (46.2)	11 (50.0)	11 (37.9)	25 (59.5)	27 (60.0)
Progressive disease	42 (20.2)	78 (39.6)	1 (4.5)	10 (34.5)	2 (4.8)	12 (26.7)
Not evaluable	8 (3.8)	18 (9.1)	4 (18.2)	4 (13.8)	3 (7.1)	4 (8.9)
Objective response rate [ORR (CR+PR)], n (%) [95% CI][1]	55 (26.4) [20.6, 33.0]	10 (5.1) [2.5, 9.1]	6 (27.3) [10.7, 50.2]	4 (13.8) [3.9, 31.7]	12 (28.6) [15.7, 44.6]	2 (4.4) [0.5, 15.1]
Difference (95% CI) in ORR (vs sorafenib) p-value	21.4 (14.6, 28.1) p<0.0001		13.5 (-9.0, 35.9) p=0.1152		24.1 (9.2, 39.1) p=0.0026	
Disease control rate [(DCR (CR+PR+SD)], n (%) [95% CI][1]	158 (76.0) [69.6, 81.6]	101 (51.3) [44.1, 58.4]	17 (77.3) [54.6, 92.2]	15 (51.7) [32.5, 70.6]	37 (88.1) [74.4, 96.0]	29 (64.4) [48.8, 78.1]
Difference (95% CI) in DCR (vs sorafenib)[2] p-value[3]	24.7 (15.6, 33.8) p<0.0001		25.5 (0.3, 50.8) p=0.0317		23.7 (6.6, 40.7) p=0.0079	

BIRC, blinded independent review committee; Cam, camrelizumab; CI, confidence interval; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; Rivo, rivoceranib.
The denominator of the percentages is the number of patients in the ITT Set within each treatment group.
[1]95% CI is calculated using Clopper-Pearson method.
[2]95% CI is calculated using normal approximation in binomial proportions.
[3]p-value (one sided) is calculated using Cochran-Mantel-Haenszel (CMH) test stratified by randomization stratification factors.

Time to Response and Duration of Response by BIRC Assessment (RECIST v1.1) in Patients with Confirmed Complete Response or Partial Response

	Hepatitis B Virus		Hepatitis C Virus		Non-Viral	
	Rivo + Cam (N=208)	Sorafenib (N=197)	Rivo + Cam (N=22)	Sorafenib (N=29)	Rivo + Cam (N=42)	Sorafenib (N=45)
Patients with response (CR or PR), n (%)	55 (26.4)	10 (5.1)	6 (27.3)	4 (13.8)	12 (28.6)	2 (4.4)
Median time to response, months	1.94	2.83	1.91	3.79	2.89	2.79
Patients with events, n (%)	27 (49.1)	8 (80.0)	1 (16.7)	1 (25.0)	5 (41.7)	1 (50.0)
PD	26 (47.3)	8 (80.0)	1 (16.7)	1 (25.0)	4 (33.3)	1 (50.0)
Death without progression	1 (1.8)	0	0	0	1 (8.3)	0
Kaplan-Meier estimates of median duration of response, months (95% CI)[1]	14.8 (8.3, NR)	9.2 (3.7, 14.8)	NR (7.7, NR)	NR (5.3, NR)	NR (4.3, NR)	NR (7.4, NR)
Range of duration of response[2]	1.8–39.7+	1.1–23.9+	7.7–23.0+	1.9–26.2+	3.4–23.1+	7.4–25.8+

BIRC, blinded independent review committee; Cam, camrelizumab; CR, complete response; PD, progressive disease; PR, partial response; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; Rivo, rivoceranib.
The denominator to calculate percentages of N, the number of subjects with confirmed CR or PR in ITT Set within each treatment group. Estimates are calculated using the Kaplan-Meier method.
[1]Confidence intervals are calculated using Brookmeyer and Crowley method.
[2]Symbol '+' means censoring

Any Grade TRAEs (≥20%) and Grade 3-4 TRAEs (≥5%): Hepatitis B Virus Infection HCC

Preferred term, n (%)	Rivo + Cam (N=208)	
	All Grades	Grade 3-4
Hypertension	157 (75.5)	85 (40.9)
Proteinuria	120 (57.7)	–
AST increased	116 (55.8)	29 (13.9)
Platelet count decreased	112 (53.8)	29 (13.9)
ALT increased	99 (47.6)	23 (11.1)
Blood bilirubin increased	100 (48.1)	19 (9.1)
Palmar-plantar erythrodysesthesia syndrome	92 (44.2)	28 (13.5)
Diarrhea	66 (31.7)	–
White blood cell count decreased	68 (32.7)	–
Reactive capillary endothelial proliferation	66 (31.7)	–
Neutrophil count decreased	67 (32.2)	15 (7.2)
GGT increased	59 (28.4)	23 (11.1)
Conjugated bilirubin increased	44 (21.2)	12 (5.8)
Lipase increased	–	13 (6.3)
Hypothyroidism	42 (20.2)	–

ALT, alanine aminotransferase; AST, aspartate aminotransferase increased; Cam, camrelizumab; GGT, gamma-glutamyltransferase; Rivo, rivoceranib; TRAE, treatment-related adverse event.

Any Grade TRAEs (≥20%) and Grade 3-4 TRAEs (≥5%): Hepatitis C Virus Infection HCC

Preferred term, n (%)	Rivo + Cam (N=22)	
	All Grades	Grade 3-4
Hypertension	8 (36.4)	6 (27.3)
Proteinuria	6 (27.3)	-
AST increased	10 (45.5)	6 (27.3)
Platelet count decreased	7 (31.8)	2 (9.1)
ALT increased	9 (40.9)	6 (27.3)
Blood bilirubin increased	9 (40.9)	3 (13.6)
Diarrhea	7 (31.8)	-

ALT, alanine aminotransferase; AST, aspartate aminotransferase increased; Cam, camrelizumab; GGT, gamma-glutamyltransferase; Rivo, rivoceranib; TRAE, treatment-related adverse event.

Any Grade TRAEs (≥20%) and Grade 3-4 TRAEs (≥5%): Non-Viral HCC

Preferred term, n (%)	Rivo + Cam (N=42)	
	All Grades	Grade 3-4
Hypertension	24 (57.1)	13 (31.0)
ALT increased	21 (50.0)	9 (21.4)
AST increased	23 (54.8)	12 (28.6)
Reactive capillary endothelial proliferation	13 (31.0)	3 (7.1)
Hypothyroidism	13 (31.0)	-
Diarrhea	11 (26.2)	3 (7.1)
Fatigue	11 (26.2)	3 (7.1)
Asthenia	10 (23.8)	-
Proteinuria	9 (21.4)	-
Rash	9 (21.4)	-
Palma-plantar erythrodysesthesia syndromer	-	4 (9.5)
GGT increased	-	3 (7.1)
Hyponatremia	-	3 (7.1)

ALT, alanine aminotransferase; AST, aspartate aminotransferase increased; Cam, camrelizumab; GGT, gamma-glutamyltransferase; Rivo, rivoceranib; TRAE, treatment-related adverse event.

CONCLUSIONS

- The CARES-310 exploratory post-hoc analysis of viral and non-viral HCC etiology suggests clinically meaningful mOS and mPFS benefit for patients in the camrelizumab plus rivoceranib arm with hepatitis B or C viral infection HCC, or non-viral HCC.
- Safety results showed that hypertension was the most common treatment-related adverse event for those with HCC due to hepatitis B virus infection and non-viral etiology.
- Although the sample size of HCC etiology groups is not equal, these results indicate the favorable clinical benefit of using camrelizumab plus rivoceranib as first-line treatment for patients with unresectable HCC, independent of viral or non-viral etiology.

REFERENCES

1. Qin S, et al. *Lancet*. 2023;402(10408):1133-1146.
2. Vogel A, et al. *J Clin Oncol*. 2024;42(suppl_16):abs 4110.

ACKNOWLEDGMENTS

This post-hoc analysis was conducted by Elevar Therapeutics. The Phillips Group Oncology Communications Inc. provided professional assistance with poster preparation. Financial support for writing and editorial services was provided by Elevar Therapeutics.