The Mass Balance and Metabolic Profile of Lirafugratinib in Healthy Male Volunteers

Tracey Wei, Seong Jang, Grace Lee, Laura Alexander, Kristin Ryan

Elevar Therapeutics, Fort Lee, NJ, USA

Presented at
ASCPT 2026 Annual Meeting
March 4-6, 2026

BACKGROUND

- Lirafugratinib (RLY-4008; QSC206365), is the first highly selective, irreversible inhibitor of fibroblast growth factor receptor (FGFR)2 designed to target oncogenic FGFR2 driver alterations and resistance mutations.
- FGFR tyrosine kinase pathway plays a crucial role in regulating essential physiological processes, and its improper regulation has been linked to the development of cancer.[1,2]
- Lirafugratinib has been developed and submitted for the new drug application (NDA) for the treatment of adults with previously treated, unresectable, locally advanced or metastatic cholangiocarcinoma (CCA) and other solid tumors harboring FGFR2 fusion or other rearrangement (f/r), who have not received FGFR inhibitors.

PURPOSE

- To determine the mass balance and routes and rates of excretion for lirafugratinib after a single oral dose of [^{14}C]-lirafugratinib.
- To perform metabolite profiling and structural identification from plasma, urine, and fecal samples.

METHODS

- This was a single center, single period, non-randomized, single dose, open label study in healthy male participants to assess the mass balance recovery, pharmacokinetics (PK), metabolite profile, metabolite identification, safety, and tolerability of lirafugratinib.
- A single [^{14}C]-lirafugratinib 70 mg oral dose (not more than 4.8 MBq ^{14}C) was administered in a fasted condition to 6 healthy, non-smoker, Caucasian male volunteers [median age of 39 (range: 32-50) years old; median BMI 29.1 kg/m^2]. Participants received 240 mL water immediately after dosing and remained fasted until 4 h post-dose.
- Blood, urine, and fecal samples were collected at multiple timepoints for PK evaluation until 312 hours post-dose. Total radioactivity (TR) concentrations in plasma, whole blood, urine, and feces were determined using liquid scintillation counting. Plasma concentration of lirafugratinib and its metabolite profiling were analyzed using liquid chromatography-radio-detection with tandem mass spectrometry. Adverse events were monitored.

REFERENCES

1. Haugsten EM, Wiedlocha A, Olsnes S, Wesche J. Roles of fibroblast growth factor receptors in carcinogenesis. *Mol Cancer Res*. 2010;8(11):1439-1452.
2. Chae YK, Ranganath K, Hammerman PS, et al. Inhibition of the fibroblast growth factor receptor (FGFR) pathway: the current landscape and barriers to clinical application. *Oncotarget*. 2017;8(9):16052-16074.

RESULTS

Figure 1. Lirafugratinib Concentration in Plasma and Total Radioactivity in Plasma and Whole Blood Versus Time Following a Single Oral Dose of 70 mg [^{14}C]-Lirafugratinib

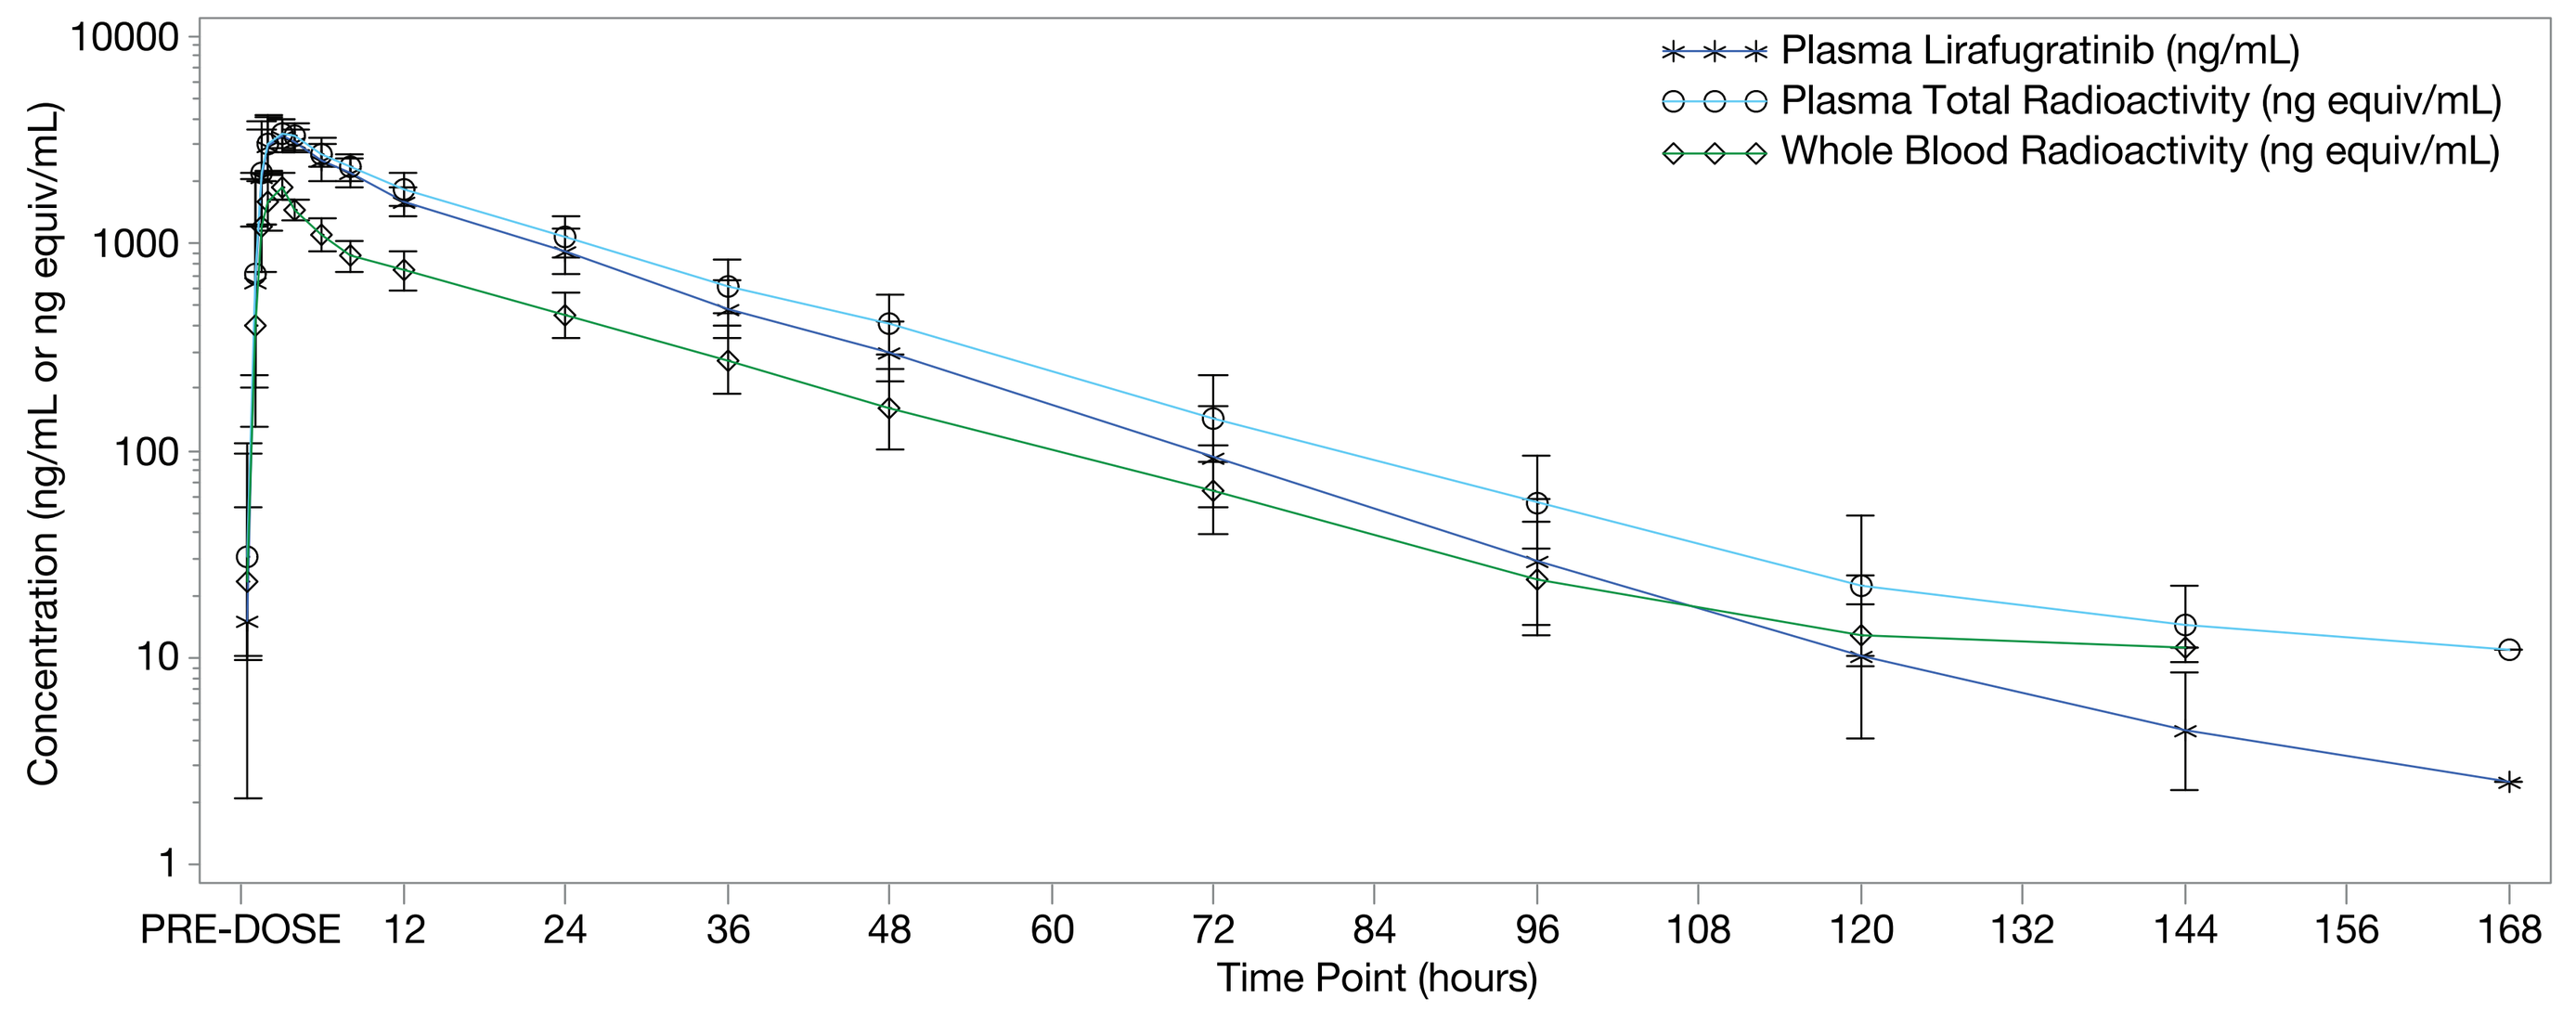

Plasma Lirafugratinib (ng/mL)
Plasma Total Radioactivity (ng equiv/mL)
Whole Blood Radioactivity (ng equiv/mL)
Concentration (ng/mL or ng equiv/mL)
10000
1000
100
10
1
PRE-DOSE 12 24 36 48 60 72 84 96 108 120 132 144 156 168
Time Point (hours)

Data presented as geometric mean (±geometric SD) in Log_{10}/Linear scale.

Table 1. Summary of Geometric Mean (Geometric CV%) PK Parameters for Lirafugratinib in Plasma and Total Radioactivity in Plasma and Whole Blood Following a Single Oral Dose of 70 mg [^{14}C]-Lirafugratinib

Parameter (units)	Lirafugratinib Plasma N=6	Total Radioactivity Plasma N=6	Total Radioactivity Whole Blood N=6
T_{lag}[a] (h)	0.25 (0.00-0.50)	0.25 (0.00-0.50)	0.25 (0.00-0.50)
T_{max}[a] (h)	3.00 (1.50-4.00)	3.00 (1.50-4.00)	3.00 (1.50-4.00)
C_{max} (ng/mL)[b]	3470 (21.4%)	3520 (20.3%)	1970 (17.3%)
$AUC_{(0-last)}$ (ng·h/mL)[b]	60400 (24.8%)	70800 (24.7%)	30000 (26.3%)
$AUC_{(0-inf)}$ (ng·h/mL)[b]	60600 (24.8%)	71700 (24.5%)	30900 (25.7%)
$t_{½}$ (h)	14.9 (15.1%)	17.4 (14.9%)	17.3 (14.3%)
CL/F (mL/min)	19.2 (24.8%)	NA	NA
V_z/F (L)	24.9 (12.1%)	NA	NA
B:P AUC	NA	NA	0.431 (7.7%)

AUC, area under the curve; AUC_{0-inf}, AUC from time 0 extrapolated to infinity; $AUC_{0-tlast}$, AUC from time 0 to time of the last quantifiable concentration; B:P AUC, ratio of whole blood: plasma TR based on AUC; CL/F, apparent total body clearance; C_{max}, maximum observed concentration; NA, not applicable; PK, pharmacokinetics; $t_{½}$, terminal elimination half-life; T_{lag}, time prior to the first measurable concentration; T_{max}, time of maximum observed concentration; TR, total radioactivity; V_z/F, Apparent volume of distribution.
[a]Median (range).
[b]ng equivalents for TR. TR concentrations in plasma and whole blood were determined using liquid scintillation counting with ng equivalents/mL for plasma/whole blood.

Table 2. Mean Cumulative Amount of Total Radioactivity Excreted and Recovered (± SD) in Urine, Feces and Total (Urine and Feces Combined) Following Single Oral Administration of 70 mg [^{14}C]-Lirafugratinib

Collection Period (h)	Cum%Ae(urine) (%) N=6	Cum%Ae(faeces) (%) N=6	Cum%Ae(total) (%) N=6
0 - 12[a]	2.064 (0.303)	NA	NA
0 - 24	3.895 (0.486)	0.132 (0.289)	4.029 (0.607)
0 - 48	6.016 (0.491)	4.308 (4.853)	10.327 (4.735)
0 - 72	6.989 (0.502)	29.306 (10.821)	36.298 (10.617)
0 - 96	7.442 (0.535)	56.165 (18.759)	63.610 (18.417)
0 - 120	7.641 (0.580)	60.011 (20.777)	67.655 (20.339)
0 - 144	7.722 (0.598)	73.200 (7.803)	80.925 (7.369)
0 - 168	7.764 (0.611)	76.078 (5.346)	83.844 (4.970)
0 - 192	7.788 (0.618)	78.507 (5.314)	86.298 (5.115)
0 - 216	7.803 (0.622)	79.016 (4.957)	86.821 (4.760)
0 - 240	7.812 (0.624)	79.178 (4.861)	86.993 (4.662)
0 - 264	7.818 (0.625)	79.385 (4.882)	87.206 (4.691)
0 - 288	7.822 (0.626)	79.465 (4.872)	87.290 (4.680)
0 - 312	7.826 (0.626)	79.540 (4.887)	87.369 (4.695)
CumAe(Overall) (mg equivalents)	5.39 (0.431)	54.7 (3.360)	60.1 (3.230)

NA, not applicable.
[a]urine only

Table 3. Lirafugratinib Metabolite Abundance in Human Plasma and Excreta in Healthy Male Subjects as Compared to Metabolite Identification in Rats and Dogs

Species			Human[a]			Rat[b]			Dog[c]	
Component	Molecular weight[d] (Da)	Molecular formula	Plasma (mean %ROI)	Urine (mean %Dose)	Feces (mean %Dose)	Plasma (AUC)	Feces (mean %Dose)	Bile (mean %Dose)	Plasma (AUC)	Feces
M417: RTX-1262245	417	$C_{23}H_{20}FN_5O_2$	✓	x	F7 (3.12)	P5 (✓)	F6 (5)	B15 (1)	✓	F10 (1)
M617: Amide hydrolysis and glucuronidation of lirafugratinib	617	$C_{30}H_{28}FN_7O_7$	NA	U10 (2.84)	NA	NA	NA	NA	NA	NA
M543: Lirafugratinib +2O +2H	543	$C_{28}H_{26}FN_7O_4$	NA	NA	**F8 (10.45)**	P7 (1)	F8 (10)[e]	B18 (1)	NA	NA
M483: Amide hydrolysis and N-acetylation of lirafugratinib	483	$C_{26}H_{22}FN_7O_2$	NA	NA	**F10 (7.67)**	P8 (9)	F10 (7)	B20 (1)	NA	NA
M525: Monohydroxy lirafugratinib or hydroxylation of lirafugratinib	525	$C_{28}H_{24}FN_7O_3$	NA	NA	F14 (6.72)	P10 (11)	F14 (7)	B25 (2)	NA	NA
M509: Lirafugratinib	509	$C_{28}H_{24}FN_7O_2$	**P3 (88.81)**	✓	**F16 (23.16)**	✓ (53)	✓ (16)	✓ (<1)	✓ (47)	✓ (12)

ROI, region of interest.
[a]Following a single oral dose of 70 mg [^{14}C]-lirafugratinib in healthy male subjects
[b]Following a single oral dose of 3 mg/kg [^{14}C]-lirafugratinib in male rats
[c]Following a single oral dose of 3 mg/kg [^{14}C]-lirafugratinib in male dogs
[d]Molecular weights quoted are mono-isotopic and refer to unlabeled components
[e]F8 (10) in rats is combined of M792 and M543
✓: Component confirmed as present by liquid chromatography-mass spectrometry but no region identifier assigned
x: Component not observed
NA: Component below threshold required for metabolite identification and does not correspond to supplied reference standard

Figure 2. Proposed Metabolic Pathway of Lirafugratinib in Humans

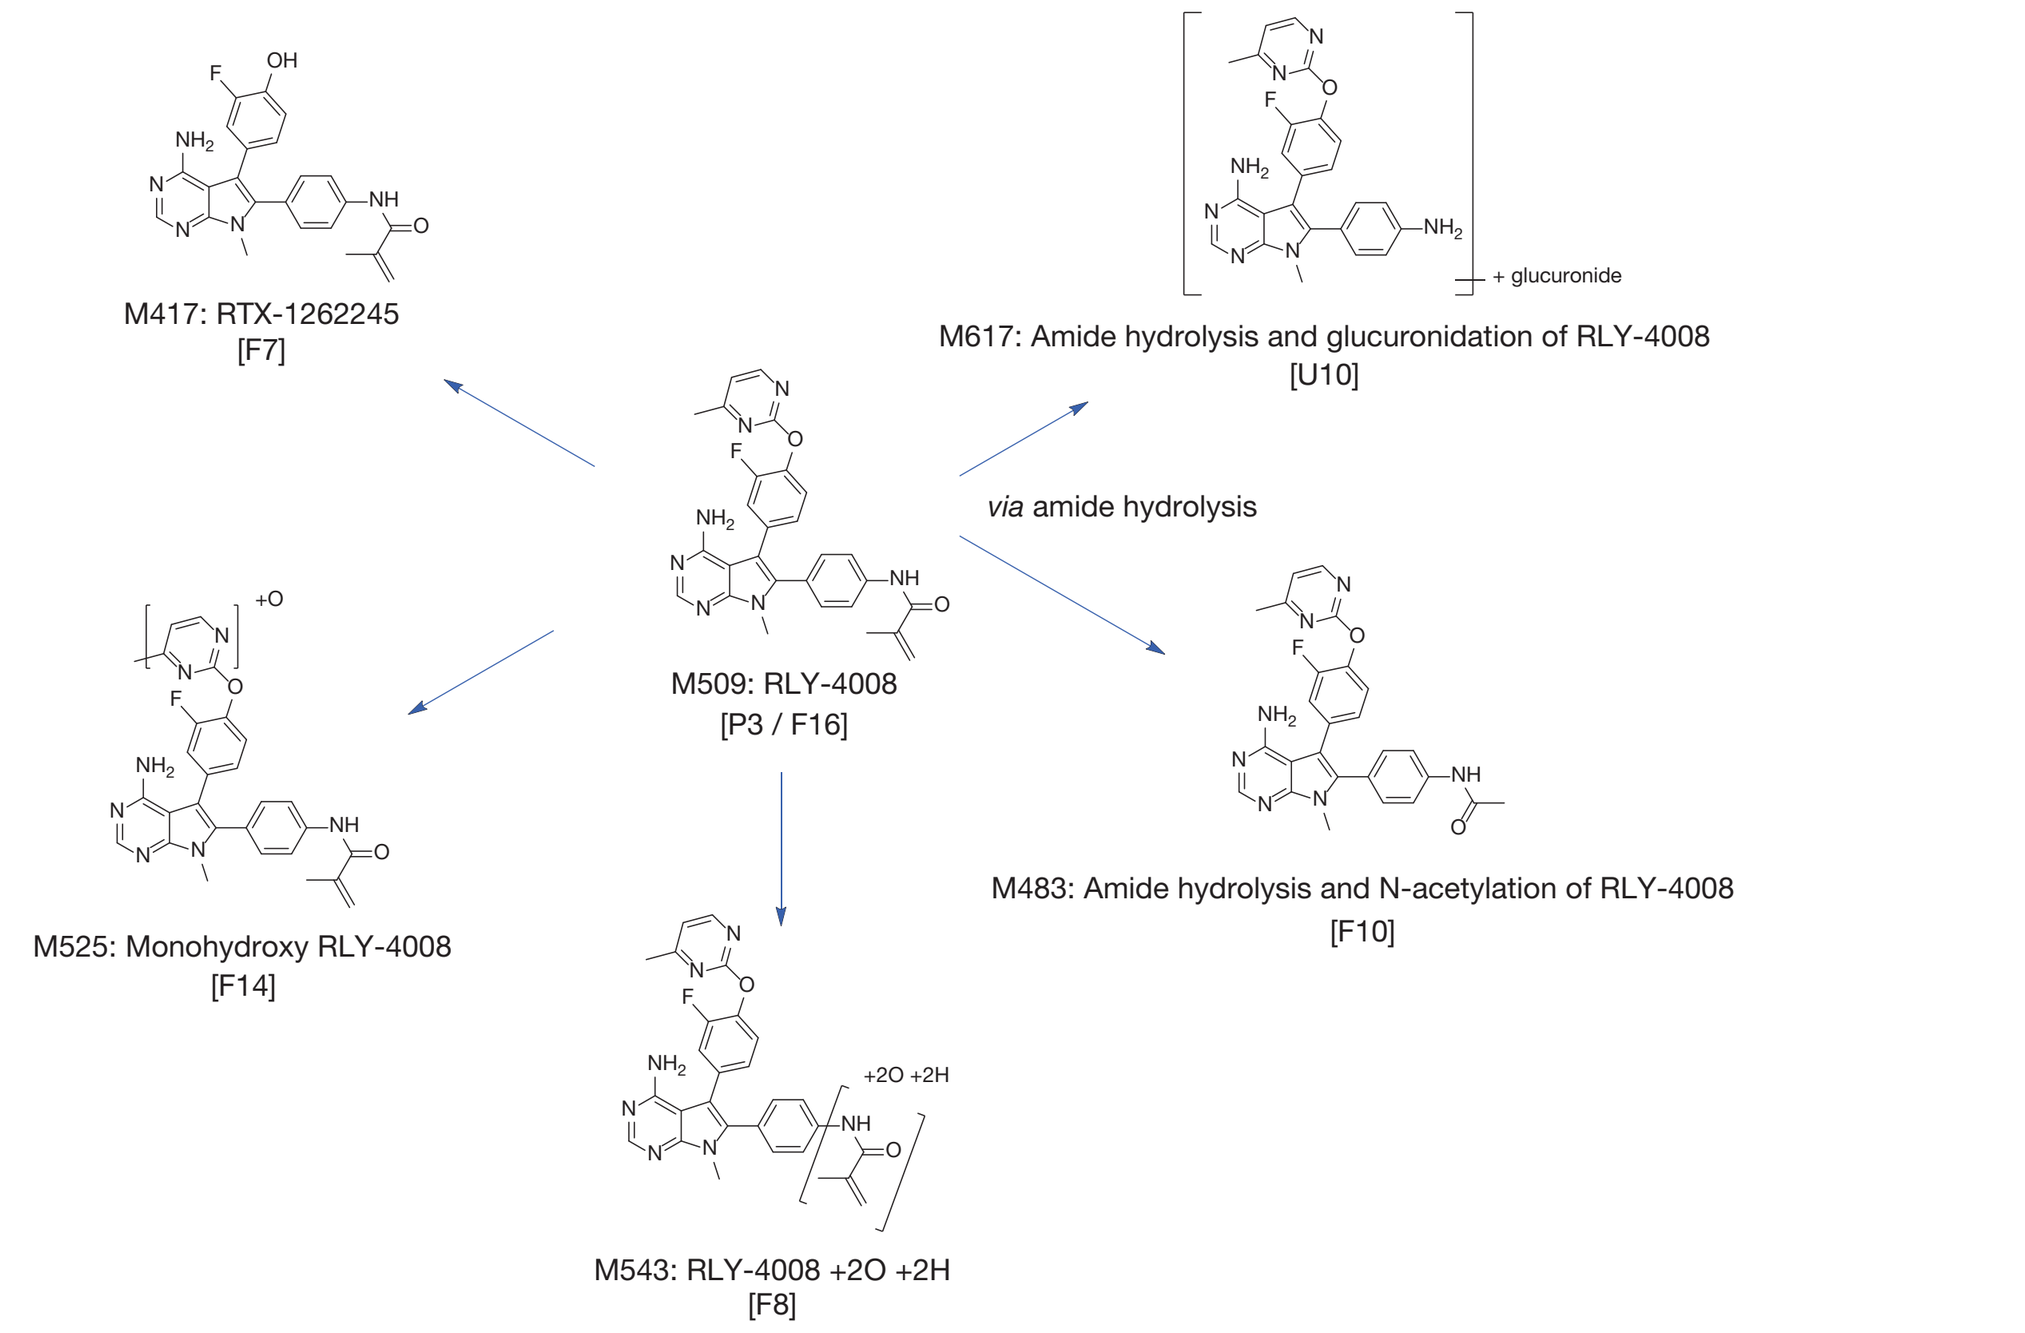

M417: RTX-1262245
[F7]
M617: Amide hydrolysis and glucuronidation of RLY-4008
[U10]
+ glucuronide
via amide hydrolysis
M509: RLY-4008
[P3 / F16]
M525: Monohydroxy RLY-4008
[F14]
M483: Amide hydrolysis and N-acetylation of RLY-4008
[F10]
M543: RLY-4008 +2O +2H
[F8]

Figure 3. Lirafugratinib Metabolite Identification *in Vivo* in Mice, Rats, and Dogs

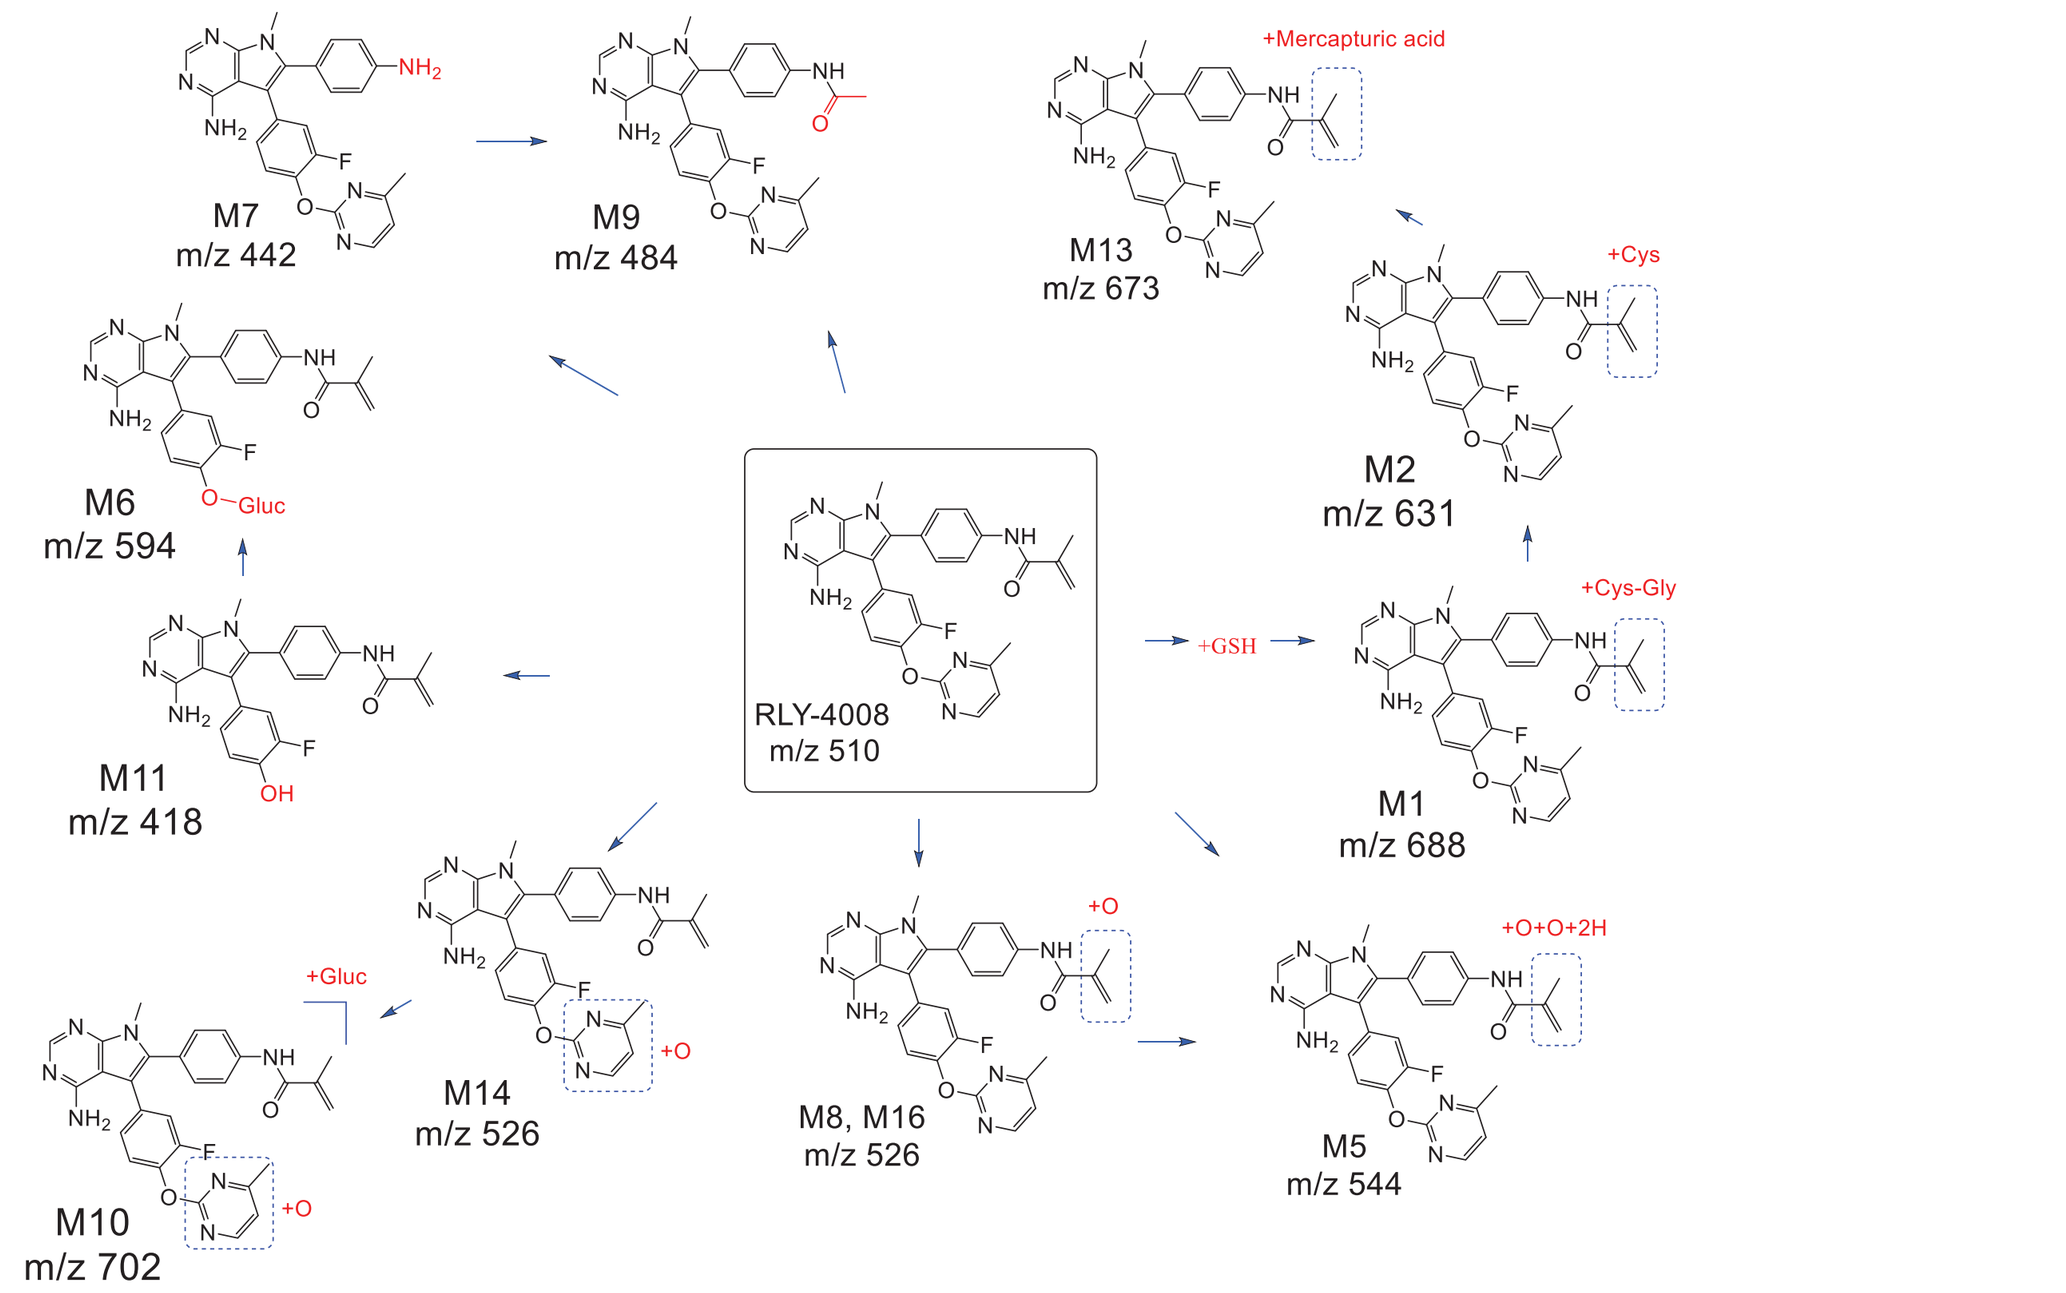

M7
m/z 442
M9
m/z 484
M13
m/z 673
+Mercapturic acid
M2
m/z 631
+Cys
M6
m/z 594
+Gluc
RLY-4008
m/z 510
+GSH
M1
m/z 688
+Cys-Gly
M11
m/z 418
M14
m/z 526
M8, M16
m/z 526
M5
m/z 544
+O+O+2H
M10
m/z 702
+O

+2H, hydrogenation; +Cys, cysteine conjugate; +Cys-Gly, cysteinylglycine conjugate; Gluc, glucuronide conjugate; +GSH, glutathione conjugate; +O, oxidation; M, metabolite; m/z, mass-to-charge ratio.

- The mean plasma concentration vs time profiles of lirafugratinib and TR are presented in **Figure 1**. Plasma and whole blood PK parameters of lirafugratinib are presented in **Table 1**.
- Lirafugratinib was absorbed with a median time to peak drug concentration of 3.0 hours. The blood:plasma TR ratios were consistently below 1.0, with a geometric mean of 0.431 (range: 0.372 to 0.568) based on AUC_{0-inf}, indicating a non-preferential distribution to cellular components of whole blood.
- The geometric mean apparent terminal half-life for total radioactivity (17.4 hours) was similar to that estimated for lirafugratinib in plasma (14.9 hours), indicating that lirafugratinib was the most abundant plasma component.
- The mean cumulative amounts of TR excreted and recovered over the entire collection period are summarized in **Table 2**. Approximately 87% of [^{14}C]-lirafugratinib was recovered in excreta over the 312-hour sampling period, of which, 79.5% was excreted via feces and 7.8% via urine. Only 4% of the fecal recovery occurred within the first 48 hours post-dose, with the remainder excreted between 48 hours and 312 hours.
- Unchanged lirafugratinib was identified as the predominant component in plasma, accounting for 89% of circulating TR based on AUC_{0-inf}. No metabolites represented ≥ 10% of circulating TR (**Table 3**).
- In feces, 23.2% of the administered dose was recovered as unchanged lirafugratinib. The structures of 4 metabolites in fecal excretion (accounting for 28% of the administered dose) and 1 metabolite in urine excretion (accounting for 2.84% of the dose) were identified (**Table 3**).
- The main metabolites in feces were dihydroxy lirafugratinib, amide hydrolyzed and N-acetylated lirafugratinib, and monohydroxy lirafugratinib. In urine, the main component was identified as amide hydrolyzed and glucuronidated lirafugratinib (**Figure 2**).
- The metabolites identified in humans' fecal excretion were also found from *in vivo* studies in rats and dogs (**Table 3, Figure 3**).
- Four participants (66.7%) experienced one treatment emergent adverse event (TEAE) each; all were mild and assessed as unrelated to lirafugratinib. There were no severe, serious or life-threatening TEAEs, or TEAEs leading to subject withdrawal.

CONCLUSIONS

- Lirafugratinib and its metabolites were mainly excreted in feces. Unchanged lirafugratinib represented the predominant circulating component in plasma. No metabolites accounted for ≥ 10% of the circulating TR.
- No substantial difference in lirafugratinib metabolic profiles was identified between humans and rats/dogs.
- Lirafugratinib was well tolerated by healthy male participants when administered as a single oral dose of 70 mg capsule with NMT 4.8 MBq ^{14}C in the fasted state.